Pelareorep + atezolizumab and chemotherapy in third-line (3L) metastatic colorectal cancer (mCRC) patients – Interim results from the GOBLET study

ONCOLYTICS BIOTECH INC.

AIO-Studien-gGmbH

ESMO 2023, FPN: 619P

Guy Ungerechts [1], Dirk Arnold [2], Eray Goekkurt [3], Alexander Stein [3], Uwe M. Martens [4], Jack Chater [5], Houra Loghmani [6], Matt Coffey [6], Richard Trauger [7], Thomas Heineman [7]

Department of Medical Oncology, National Center for Tumor Diseases (NCT) Heidelberg and University Hospital Heidelberg (UKHD), Germany[1], Asklepios Tumorzentrum Hamburg, AK Altona, Hamburg, Germany[2], Hematology-Oncology Practice Eppendorf (HOPE), Hamburg, Germany[3], SLK-Kliniken Heilbronn GmbH, Heilbronn, Germany[4], Klinikum Chemnitz GmbH, Germany[5], Oncolytics Biotech Inc., Calgary, Canada[6], Oncolytics Biotech Inc, San Diego, USA[7]

Background and Study Design

Pelareorep (pela) is an intravenously administered, naturally occurring, non-genetically modified reovirus. Pela selectively kills tumor cells and promotes tumor-directed innate and adaptive immune responses resulting in increased T cell infiltration and PD-L1 expression in tumors, thereby priming the tumor for checkpoint blockade therapy (Samson et al., 2018)(Fig 1).

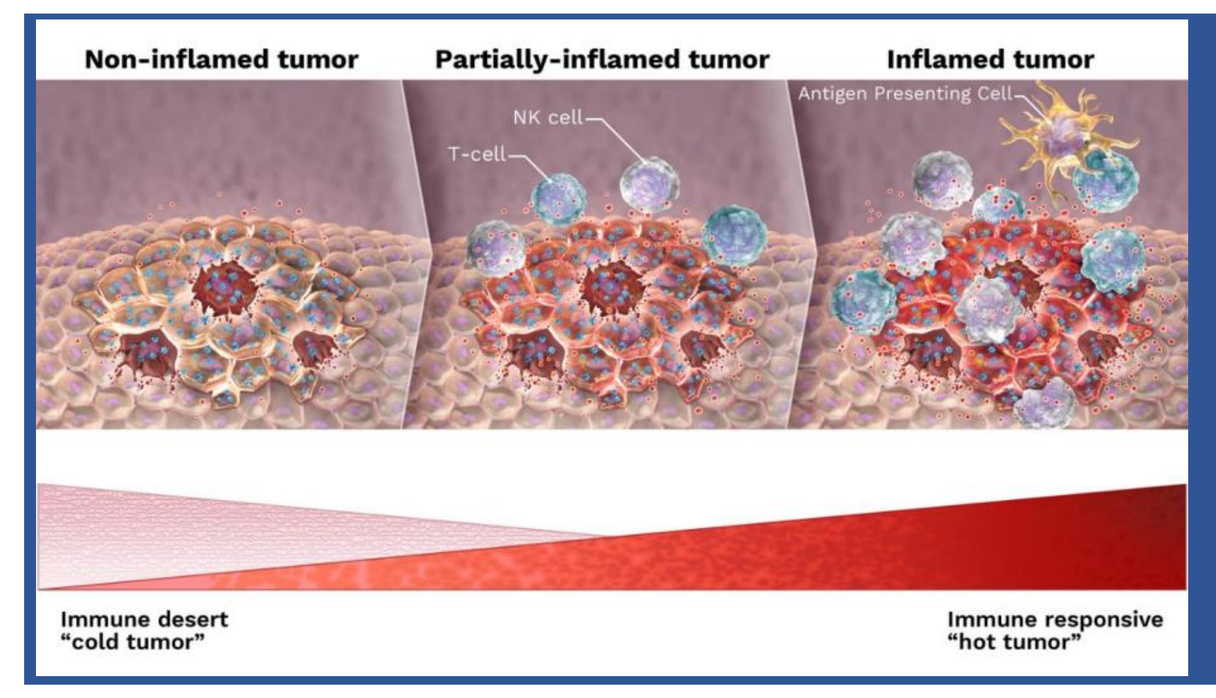
Non-inflamed tumor
Partially-inflamed tumor
Inflamed tumor
NK cell
T-cell
Antigen Presenting Cell
Immune desert "cold tumor"
Immune responsive "hot tumor"

Figure 1: Pelareorep's mechanism of action. Pelareorep selectively infects cancer cells leading to tumor cell lysis. In addition, its dsRNA genome is identified by pattern recognition receptors leading to the expression of interferons and inflammatory cytokines. This, in turn, results in immune cell recruitment and promotes the development anti-tumor innate and adaptive immune response.

GOBLET is an open-label, multiple-cohort, phase 1/2, Simon 2-stage study to assess the safety and efficacy of pela in combination with atezolizumab (atezo) +/- chemotherapy in different gastrointestinal indications. We hypothesize that treatment with pela primes the tumor microenvironment for checkpoint blockade therapy by increasing PD-L1 expression, stimulating the generation of new T cell clones, and facilitating immune cell infiltration into the tumor. Here we report the interim results for patients with third-line (3L) metastatic colorectal cancer (mCRC), independent of microsatellite instability status.

Study Design & Methods:

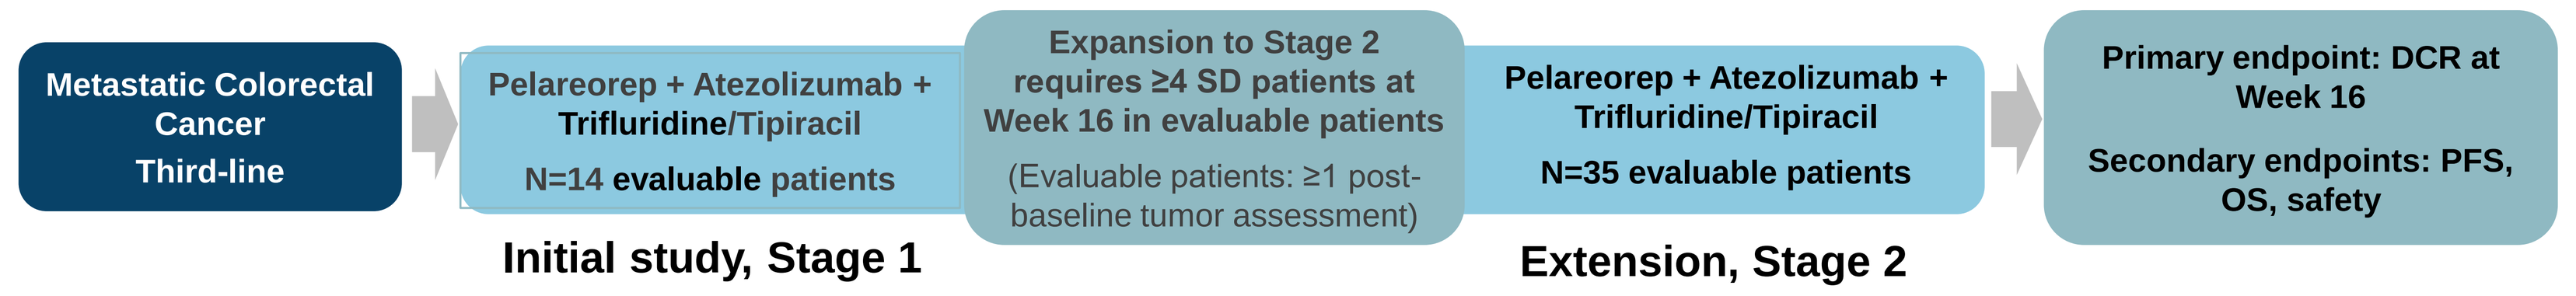
Metastatic Colorectal Cancer
Third-line
Pelareorep + Atezolizumab + Trifluridine/Tipiracil
N=14 evaluable patients
Expansion to Stage 2 requires ≥4 SD patients at Week 16 in evaluable patients
(Evaluable patients: ≥1 post-baseline tumor assessment)
Pelareorep + Atezolizumab + Trifluridine/Tipiracil
N=35 evaluable patients
Primary endpoint: DCR at Week 16
Secondary endpoints: PFS, OS, safety
Initial study, Stage 1
Extension, Stage 2

Figure 2: GOBLET, mCRC 3L study design (other GOBLET study cohorts not shown)

Primary Objectives:
- To evaluate disease control rate (DCR) at Week 16
- To evaluate the tolerability of the pela-based combination therapy

Secondary Objectives:
- To evaluate progression-free survival (PFS) and overall survival (OS)

Exploratory Objectives:
- To evaluate the immunologic effects of treatment and to explore potential biomarkers of treatment response

Patients with histologically or cytologically confirmed mCRC, independent of MSI/dMMR status, who failed 2 prior lines of treatment including oxaliplatin, irinotecan, 5-FU, ± targeted agents such as bevacizumab and/or an anti-EGFR antibody who are eligible for 3L chemotherapy with trifluridine/tipiracil were enrolled. Patients were treated with pela (4.5×10^{10} TCID50, days 1,8, 5 and 22, IV), atezo (840 mg IV, days 2 and 16), and trifluridine/tipiracil 35 mg/m^2 orally twice daily on days 1-5 and days 8-12 of each 28-day cycle. Patients must have locally advanced or metastatic unresectable disease evaluable by RECIST v1.1, be ≥18 years old and have an ECOG score ≤1. No previous treatment with a checkpoint inhibitor is permitted. The protocol-specified Stage 1 success criterion is ≥4 responses of stable disease or better at week 16.

Results – Patient Characteristics and Safety

The patient characteristics are listed in Table 1.
To date, the treatment combination has been well tolerated with no safety concerns. The most frequently (≥4 patients) reported treatment emergent adverse events (TEAEs) and Grade 3 & 4 TEAEs are listed in Table 2. There were no fatal events associated with the Investigational Medicinal Products (IMPs).

Table 2: Most frequent (>=4 patients) AEs

Adverse Event (MedDRA Preferred Term)	All TEAEs N=20 n (%)	Grade 3/4 TEAEs N=20 n (%)
Pyrexia	14 (70.0%)	2 (10.0%)
Fatigue	13 (65.0%)	1 (5.0%)
Anaemia	10 (50.0%)	6 (30.0%)
Chills	10 (50.0%)	0 (0.0%)
Nausea	8 (40.0%)	0 (0.0%)
Vomiting	7 (35.0%)	0 (0.0%)
Diarrhoea	6 (30.0%)	1 (5.0%)
Neutropenia	6 (30.0%)	6 (30.0%)
Urinary tract infection	5 (25.0%)	1 (5.0%)
Leukopenia	4 (20.0%)	0 (0.0%)
Lymphocyte count decreased	4 (20.0%)	2 (10.0%)
Neutrophil count decreased	4 (20.0%)	4 (20.0%)

Table 1: Patient Characteristics

Characteristic (N=15)	Treatment no. (%)
Age	
Median (range) — yr	54 (29-78)
<65 yr — no. (%)	11 (73%)
≥65 yr — no. (%)	4 (27%)
Male sex — no. (%)	10 (67%)
Female sex — no. (%)	5 (33%)
ECOG performance-status score — no. (%)	
0	8 (53%)
1	7 (47%)
Race or ethnic group — no. (%)	
Asian	1 (7%)
White	13 (87%)
Other	1 (7%)
Primary diagnosis — no. (%)	
Colon cancer	10 (67%)
Rectal cancer	5 (33%)
Location of primary tumor — no. (%)	
Right side	5 (33%)
Left side	10 (67%)
No. of previous regimens of treatments — no. (%)	
2	9 (60%)
≥3	6 (40%)
Median duration of disease (range) — yr	3.5 (1.25 ys to 7 ys)
Time from initial diagnosis to screen — no. (%)	
<18 mo	4 (27%)
≥18 mo	11 (73%)
No. of sites of metastasis — no. (%)	
1 or 2	9 (60%)
≥3	6 (40%)
MMR and MSI status — no. (%)	
MMR deficient and high MSI	1 (7%)
MMR proficient and stable or low MSI	14 (93%)
RAS status — no. (%)	
Mutated	8 (53%)
Wild type	7 (47%)
BRAF status — no. (%)	
Mutated	2 (13%)
Wild type	13 (87%)
Neutrophil–lymphocyte ratio — no./total no. (%)	15 (100%)
<3	8 (53%)
≥3	7 (47%)

Results – Tumor response over time

Six of 15 evaluable patients had SD as their best response, including 4 with SD lasting from 16 to 40 weeks; 9 patients had progressive disease (PD) as best response (Fig 3a). The overall DCR was 40%. The ORR, median PFS, and median OS were 0%, 2.8 months and 8.0 months, respectively (Fig 3b). The 12-month survival rate was 33%. Patients surviving greater than 12 months all had a baseline neutrophil to lymphocyte ratio ≤3.

Figure 3: Clinical course of evaluable patients

3a)

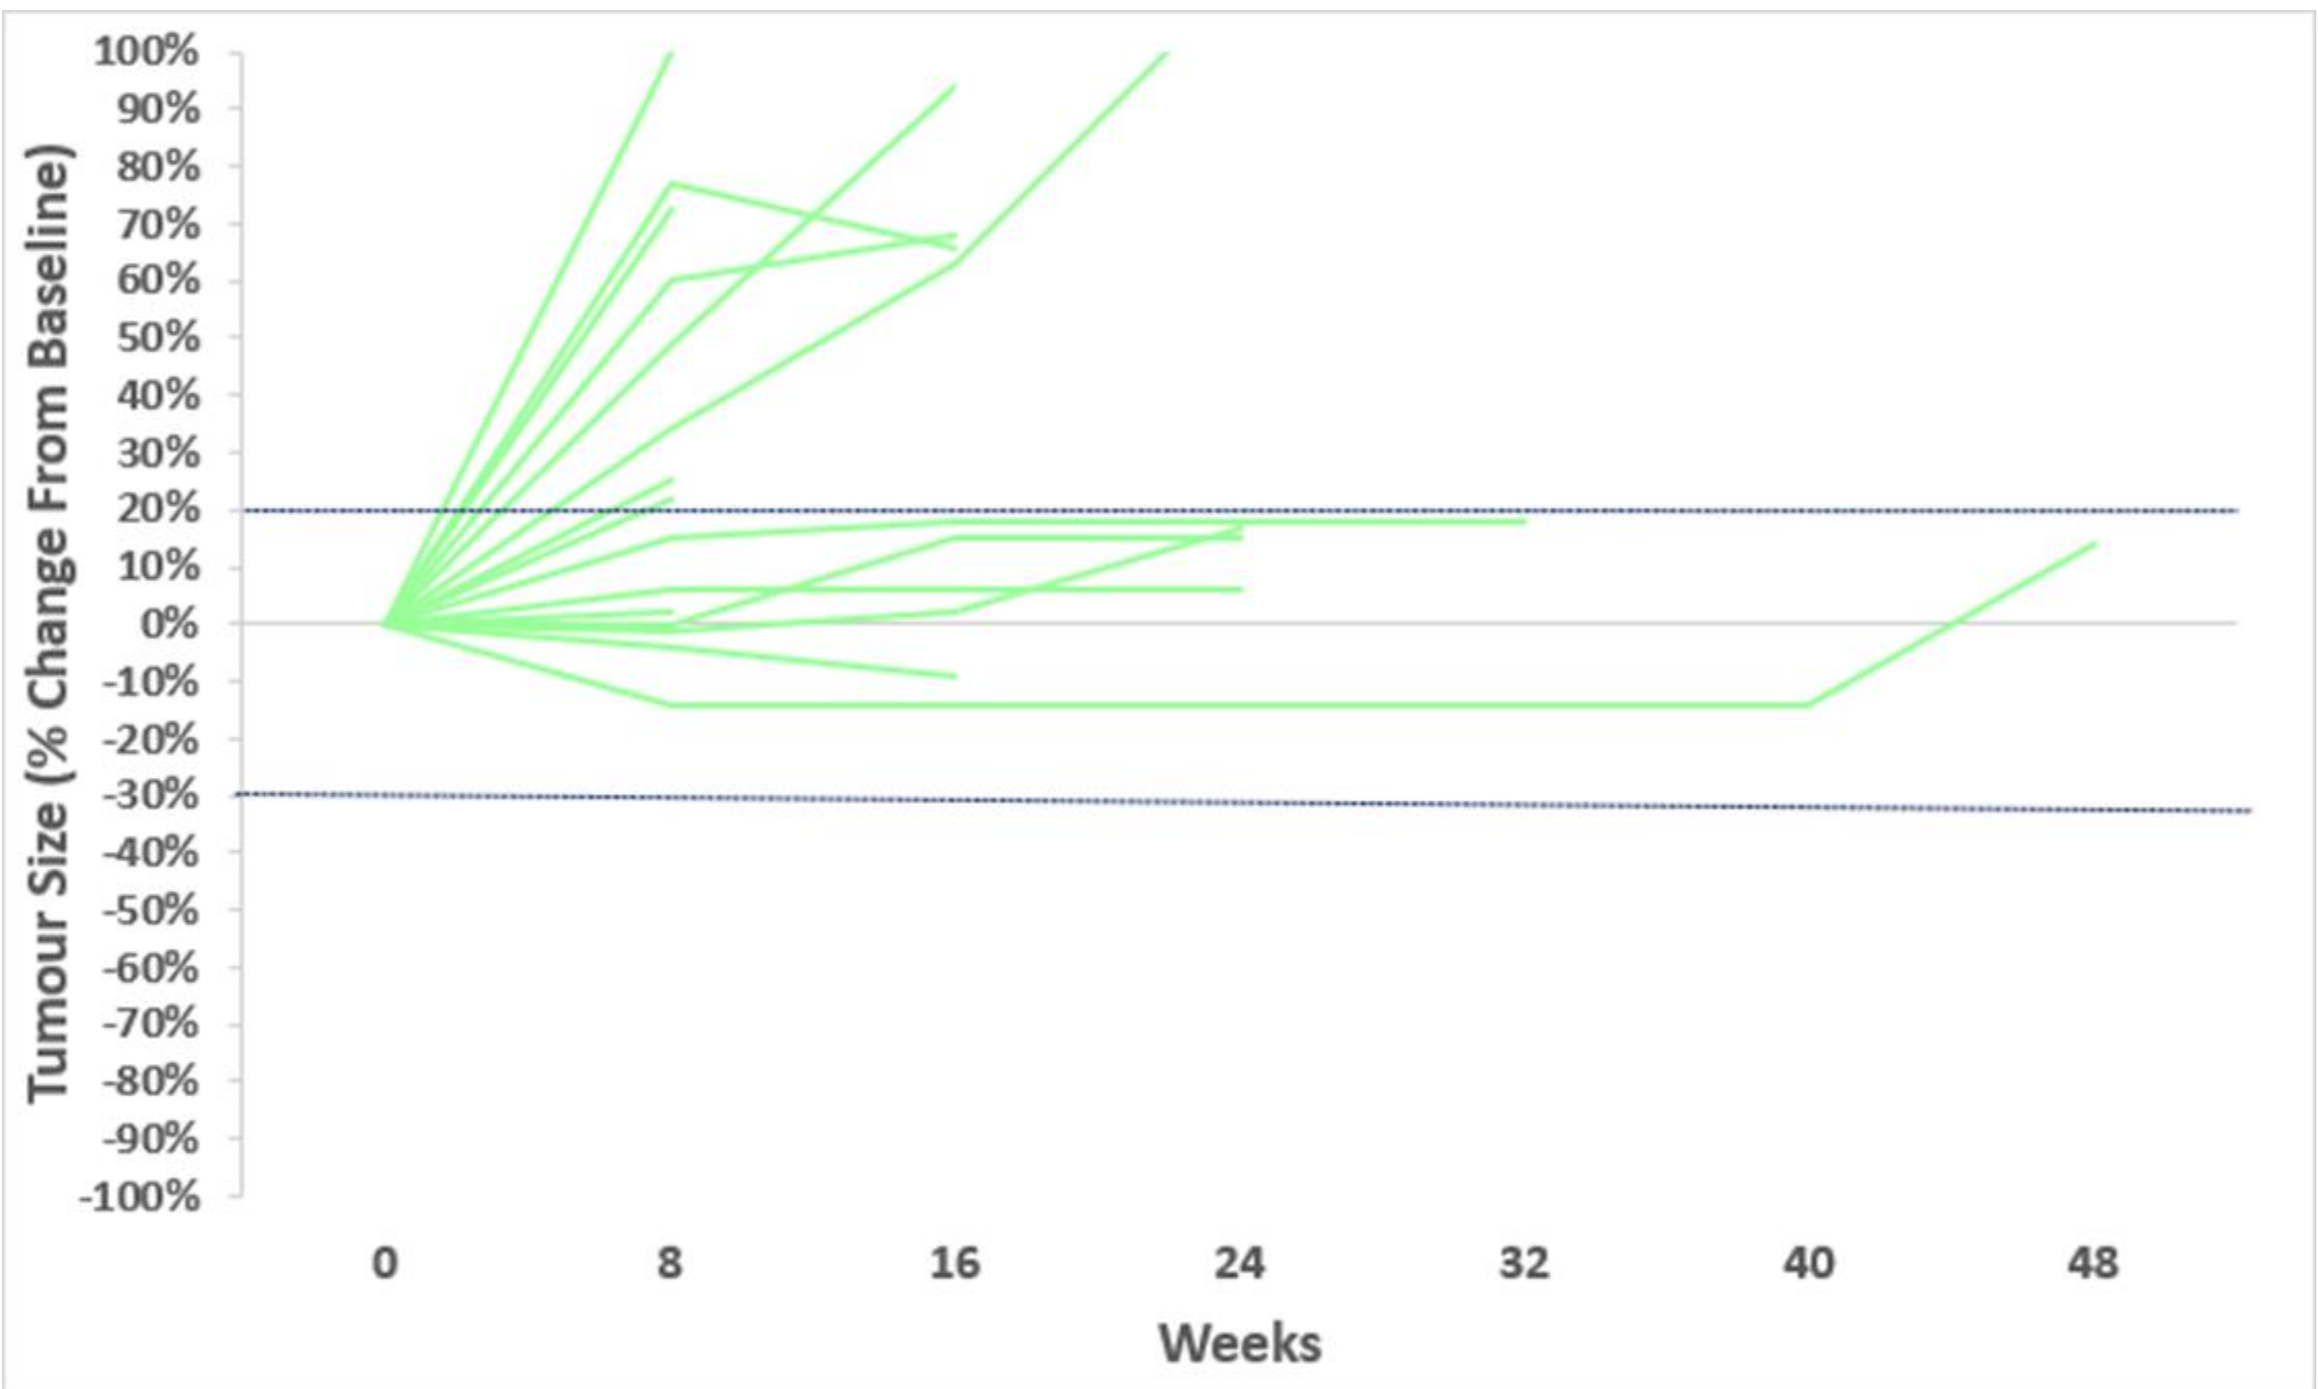
Tumour Size (% Change From Baseline)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
-10%
-20%
-30%
-40%
-50%
-60%
-70%
-80%
-90%
-100%
0
8
16
24
32
40
48
Weeks

3b)

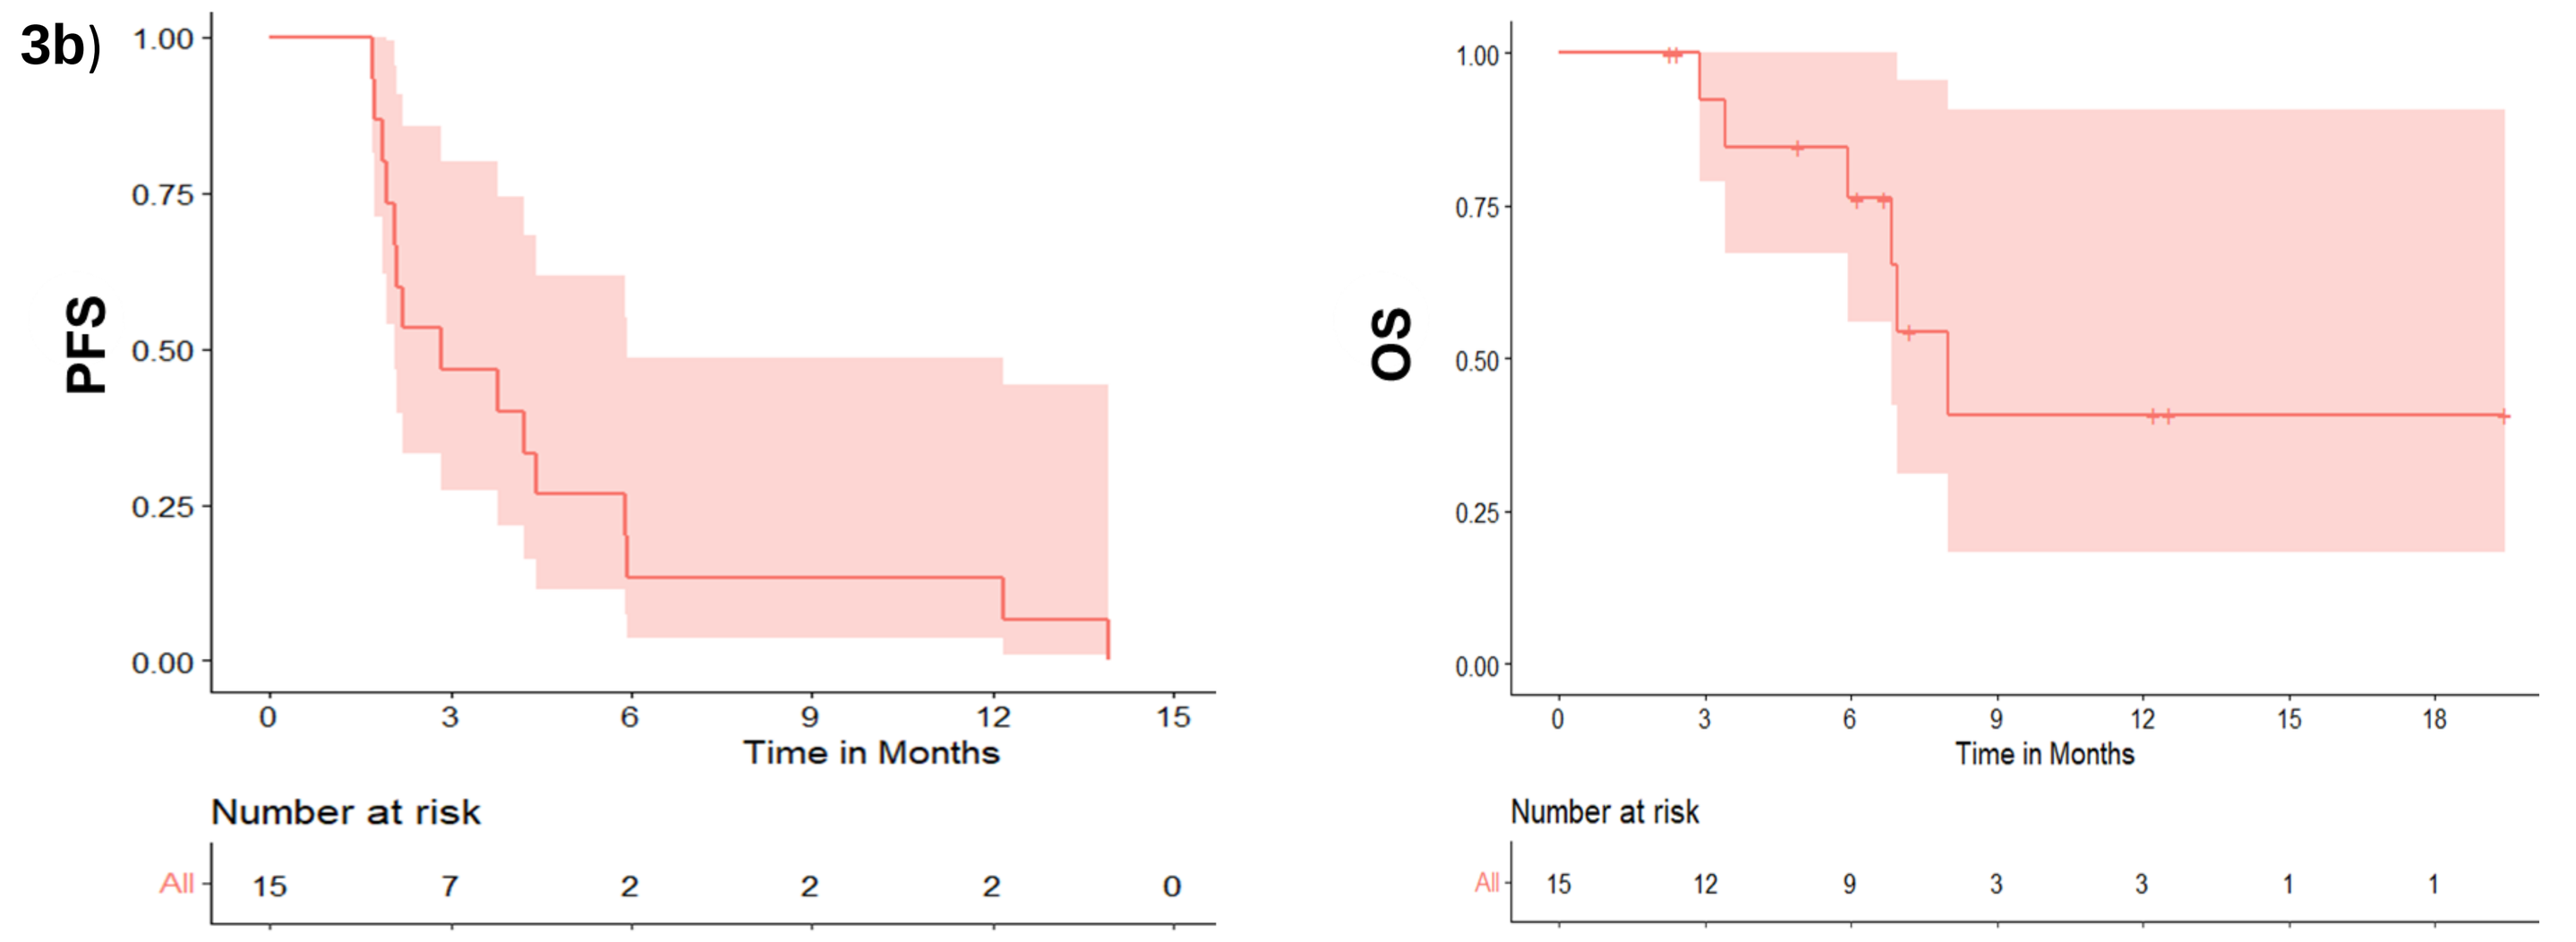
PFS
1.00
0.75
0.50
0.25
0.00
0
3
6
9
12
15
Time in Months
Number at risk
All
15
7
2
2
2
0
OS
1.00
0.75
0.50
0.25
0.00
0
3
6
9
12
15
18
Time in Months
Number at risk
All
15
12
9
3
3
1
1

PR	Partial response
SD	Stable disease
PD	Progressive disease
CR	Complete response
CBR	CR+PR+SD/total n
PFS	Progression-free survival
OS	Overall survival

Translational Results

Tumor samples collected at C1D23 (Cycle 1, Day 23) showed reoviral RNA and capsid protein staining within tumor cells but not in surrounding normal tissue (Fig 4a). Immunohistochemical staining revealed a numerical increase in PD-L1 positive cells in the tumors of patients tested (Fig 4b).

Analysis of changes in peripheral blood T cell populations post-treatment showed an expansion of new and pre-existing T cell clones (Fig 5a). At baseline, tumors had a mean of 19% tumor infiltrating lymphocytes (TILs) by T cell fraction and low clonal representation (Fig 5b). TCR-seq analysis of blood post-treatment revealed a limited expansion of pre-existing and new TIL-specific clones in the blood (Fig 5c). Clonality of baseline TILs did not predict pela-induced TIL expansion, and the expansion of new or pre-existing TILs in the blood did not appear to correlate with clinical response.

Figure 4: Tumor immunohistochemistry

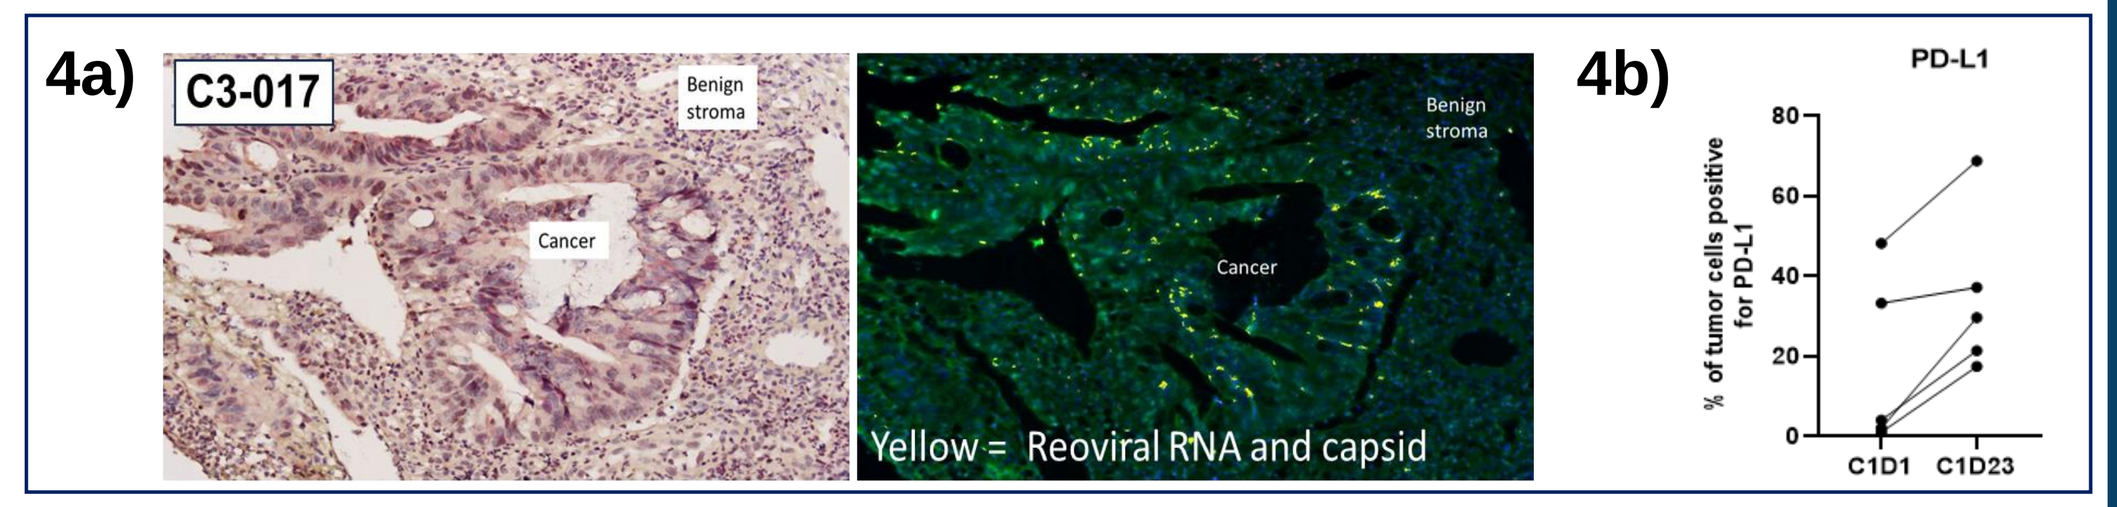
4a)
C3-017
Benign stroma
Cancer
Benign stroma
Cancer
Yellow = Reoviral RNA and capsid
4b)
PD-L1
% of tumor cells positive for PD-L1
80
60
40
20
0
C1D1
C1D23

Figure 5: TCR sequencing results

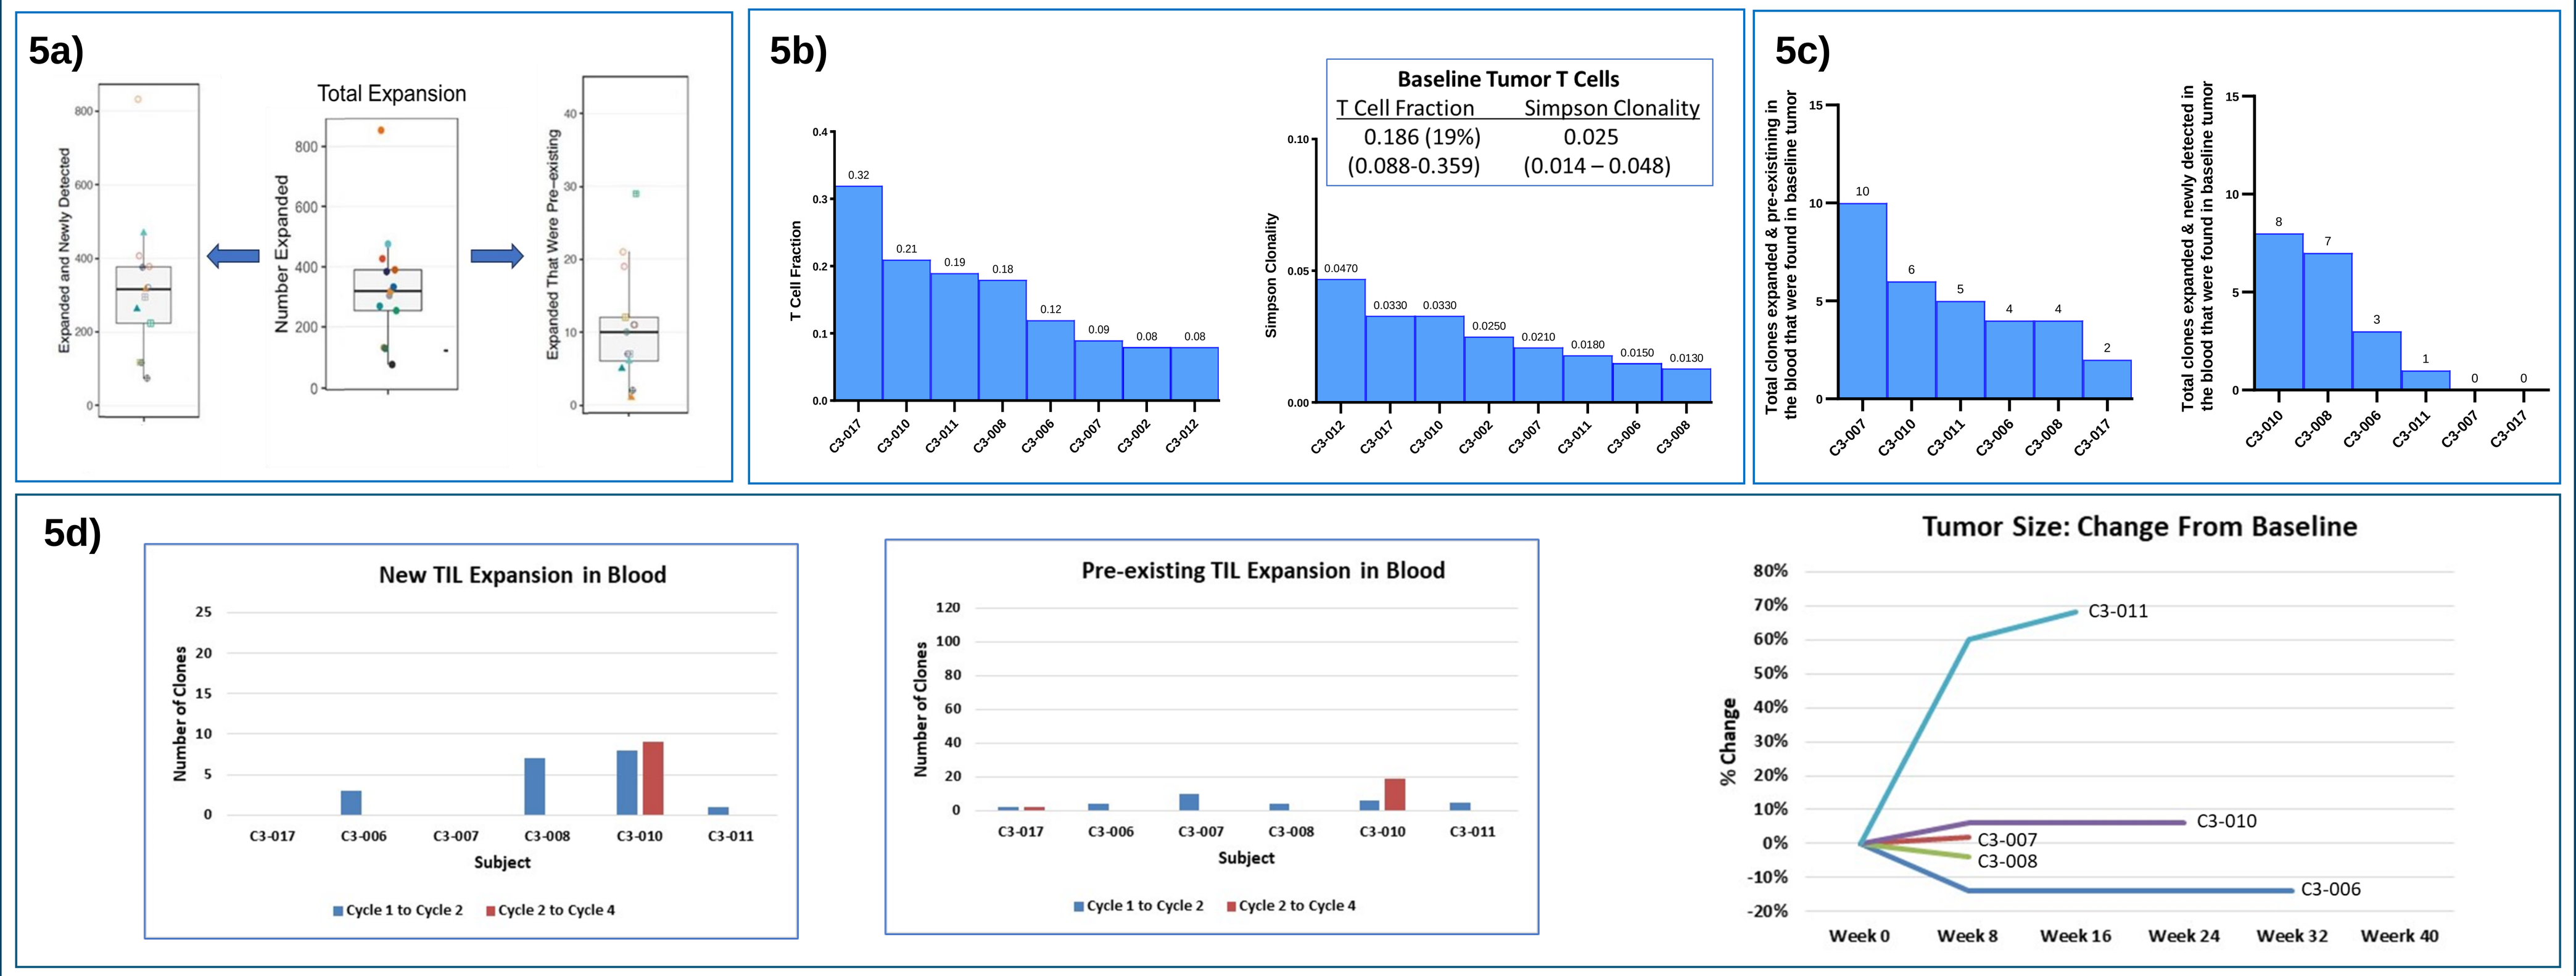
5a)
Expanded and Newly Detected
Total Expansion
Number Expanded
Expanded That Were Pre-existing
5b)
T Cell Fraction
0.32
0.21
0.19
0.18
0.12
0.09
0.08
0.08
C3-017
C3-010
C3-011
C3-008
C3-006
C3-007
C3-002
C3-012
Baseline Tumor T Cells
T Cell Fraction
Simpson Clonality
0.186 (19%)
0.025
(0.088-0.359)
(0.014 – 0.048)
Simpson Clonality
0.0470
0.0330
0.0330
0.0250
0.0210
0.0180
0.0150
0.0130
C3-012
C3-017
C3-010
C3-002
C3-007
C3-011
C3-006
C3-008
5c)
Total clones expanded & pre-existining in the blood that were found in baseline tumor
10
6
5
4
4
2
C3-007
C3-010
C3-011
C3-006
C3-008
C3-017
Total clones expanded & newly detected in the blood that were found in baseline tumor
8
7
3
1
0
0
C3-010
C3-008
C3-006
C3-011
C3-007
C3-017
5d)
New TIL Expansion in Blood
Number of Clones
Subject
C3-017
C3-006
C3-007
C3-008
C3-010
C3-011
Cycle 1 to Cycle 2
Cycle 2 to Cycle 4
Pre-existing TIL Expansion in Blood
Number of Clones
Subject
C3-017
C3-006
C3-007
C3-008
C3-010
C3-011
Cycle 1 to Cycle 2
Cycle 2 to Cycle 4
Tumor Size: Change From Baseline
% Change
C3-011
C3-010
C3-007
C3-008
C3-006
Week 0
Week 8
Week 16
Week 24
Week 32
Weerk 40

Conclusions

- The combination of pelareorep + atezolizumab + trifluridine/tipiracil in 3rd-line CRC met the protocol-specified success criterion of ≥4 patients with disease control for at least 16 weeks.
- The treatment combination was well tolerated, with no unexpected toxicity.
- Pela infection of the tumor was confirmed by immunohistochemistry. In addition, pela infection induced an increase in tumor expression of PD-L1 as well as the expansion of new T cell clones in the blood.
- Robust expansion of new T cell clones in the blood supports the ability of pela to infect colorectal tumors and induce an anti-viral T cell response. The degree of expansion of tumor specific T cell clones in the blood post-treatment is consistent with the observed level of tumor control. Given the evolving standard of care in later-line metastatic CRC patients, future studies may evaluate novel pela-based combination therapies in this disease.

Eudra-CT #:2020-003996-16 **Coordinating Investigator:** Dirk Arnold **Contact:** akupic@oncolytics.ca & Matthias.Lendner@aio-studien-ggmbh.de