Combination treatment with radiotherapy and oncolytic reovirus generates CD8+ T cell infiltration in primary and abscopal tumours in an organoid model of basal-like breast cancer

Ramsay Singer[1], Rebecca Marlow[1], Joan Kyula[1], Victoria Roulstone[1], Malin Pedersen[1], Kevin Harrington[1,2], Alan Melcher[1,2], Andrew Tutt[1, 3]
[1] The Institute of Cancer Research, London, UK. [2] The Royal Marsden, London, UK. [3] King's College London, UK

Introduction

Basal-like breast cancer (BLBC) is defined by its gene expression profile and usually does not express oestrogen, progesterone or HER2 receptors[1], so has limited treatment options. Immune checkpoint inhibition with atezolizumab has recently been approved for the treatment of advanced triple-negative breast cancer, however only a proportion of patients respond to treatment[2] and novel treatments are needed to enhance immunotherapy response rates. Oncolytic reovirus has been shown to mediate innate and adaptive immune responses, immunologically prime for checkpoint blockade[3], and enhance the cytotoxicity of radiotherapy[4].

Methods

- *In vitro:* Tumour organoids were generated from a spontaneous mammary tumour arising in a *BLG-Cre;Brca1$^{f/f}$;p53$^{+/-}$* genetically engineered mouse. Organoids were incubated with medium containing reovirus (Dearing type 3; Oncolytics Biotech) and cell survival assessed at 4 days. Irradiation was performed using an AGO 250 kV X-ray machine and cell survival assessed at 10 days. Cell survival was determined by CellTiter-Glo 3D Cell Viability Assay.
- *In vivo*: Organoids were orthotopically injected into the right 5th (10^5 cells; primary site) and left 4th (5×10^3 cells; abscopal site) mammary fat pads of C57BL/6 mice. Radiotherapy was delivered to the primary site using an Xstrahl Small Animal Radiotherapy Research Platform, under isoflurane anaesthesia. Reovirus and vehicle (PBS) were administered by intratumoural (IT) injection to the primary site, and αPD-1 (clone RMP1-14, BioXCell), by intraperitoneal injection. For survival cohorts (n=8 per group), mice were culled when either tumour reached 14mm in diameter. For IHC analysis (n=3-4 per group), 2 days after the final radiotherapy treatment, tumours were fixed for 24h in 10% neutral-buffered formalin Immunohistochemistry (IHC) was performed via standard techniques. CD8+ cell counts were calculated using QuPath v0.2.3.

BLBC organoids are sensitive to reovirus and radiotherapy *in vitro*

Organoids generated from a tumour arising in a *BLG-Cre;Brca1$^{f/f}$;p53$^{+/-}$* genetically engineered mouse, implanted into C57BL/6 mice, develop tumours (A), which display a basal-like phenotype by IHC analysis (negative for oestrogen receptor (B), Her2 (C), and positive for CK14 (D)).
In vitro, the BLBC organoids are sensitive to monotherapy with either reovirus (E) or radiotherapy (F), in a dose-dependent manner.

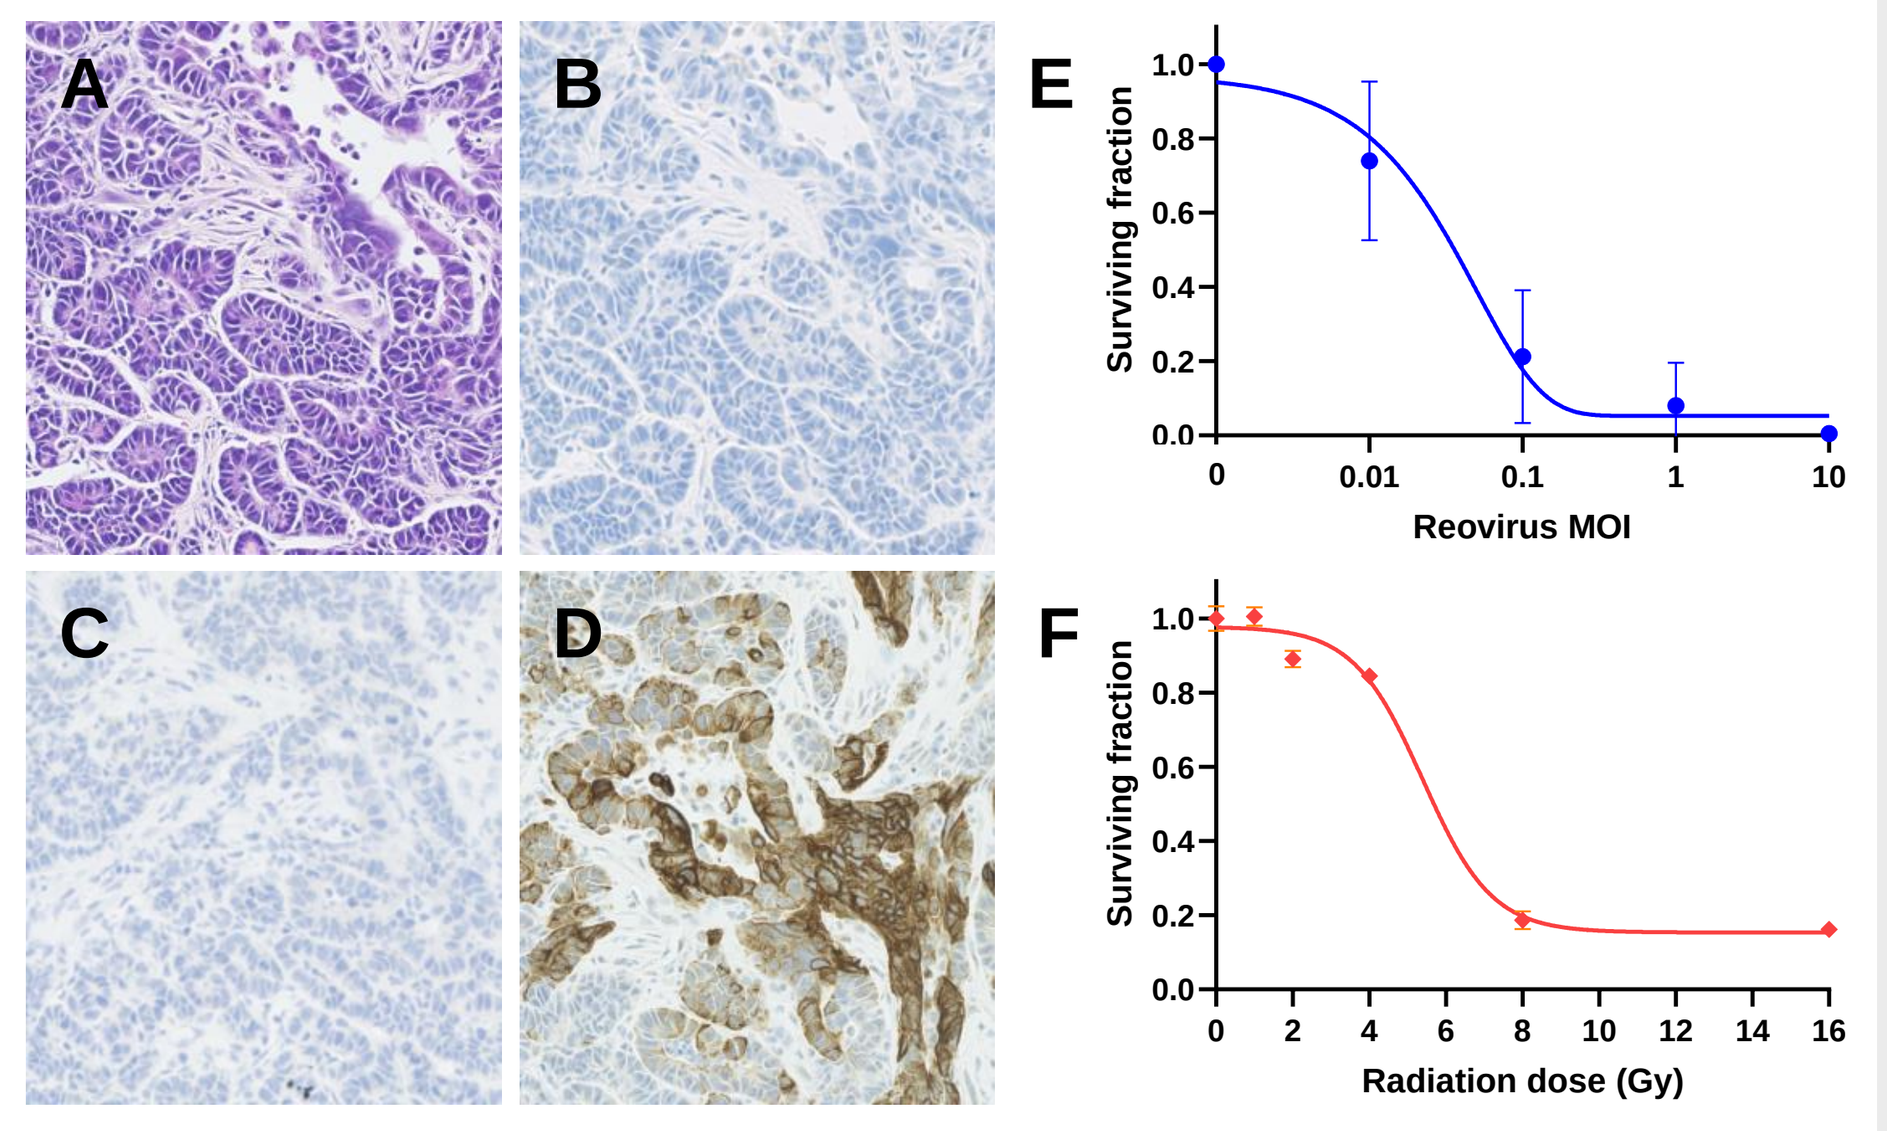
A
B
C
D
E
Surviving fraction
Reovirus MOI
F
Surviving fraction
Radiation dose (Gy)

Combination treatment with radiotherapy and oncolytic reovirus generates CD8+ T cell infiltration in primary and abscopal tumours

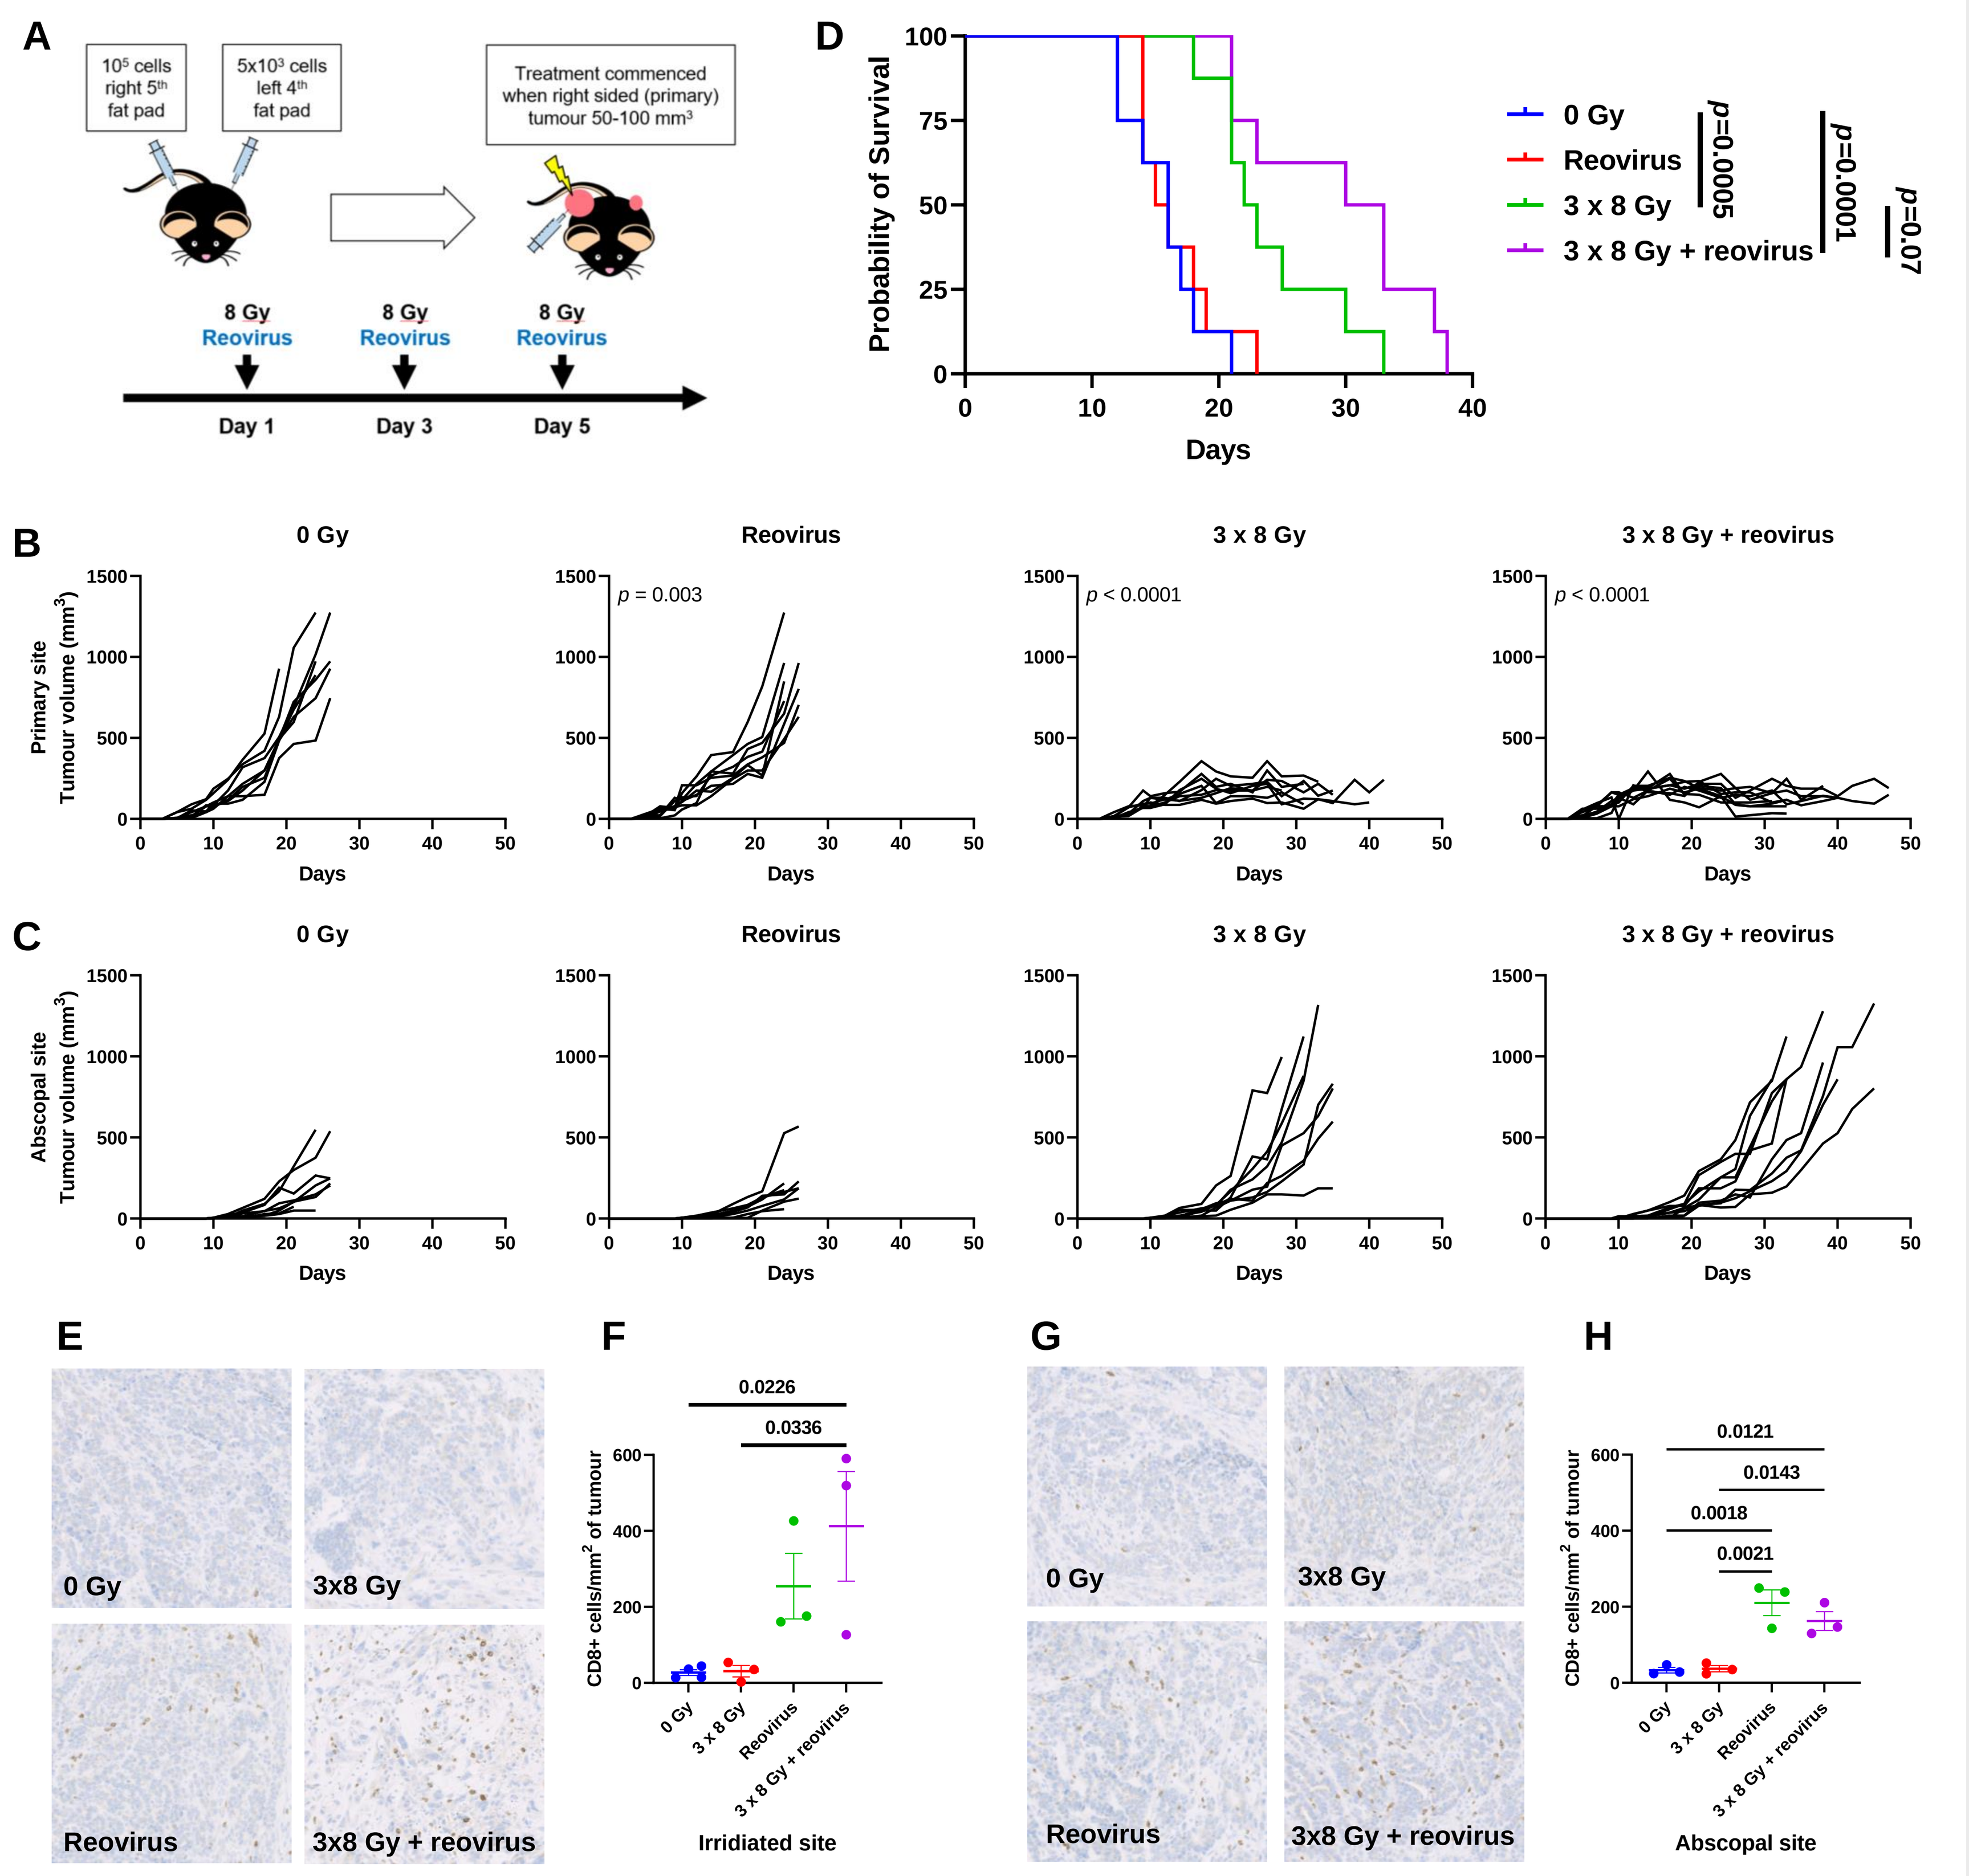
A
10⁵ cells right 5th fat pad
5x10³ cells left 4th fat pad
Treatment commenced when right sided (primary) tumour 50-100 mm³
8 Gy Reovirus
Day 1
Day 3
Day 5
D
Probability of Survival
Days
0 Gy
Reovirus
3 x 8 Gy
3 x 8 Gy + reovirus
p=0.0005
p=0.0001
p=0.07
B
Primary site Tumour volume (mm³)
0 Gy
Reovirus
3 x 8 Gy
3 x 8 Gy + reovirus
p = 0.003
p < 0.0001
p < 0.0001
Days
C
Abscopal site Tumour volume (mm³)
E
0 Gy
3x8 Gy
Reovirus
3x8 Gy + reovirus
F
CD8+ cells/mm² of tumour
0.0226
0.0336
Irridiated site
G
H
0.0121
0.0143
0.0018
0.0021
Abscopal site

- BLBC organoids were orthotopically injected bilaterally in the mammary fat pad of C57BL/6 mice. When the primary tumour reached 50-100 mm^3, mice were randomised to treatment with either sham radiotherapy, IT reovirus (3×10^7 pfu), radiotherapy (3 x 8 Gy), or radiotherapy + reovirus (A).
- Single-agent treatment with either radiotherapy or reovirus significantly reduced the growth rate of primary (treated) tumours compared with controls, though there was no additive effect of combination treatment (B). There were no significant differences between treatment groups in abscopal tumour growth rates (C). *P* values represent comparison between treatment groups by two-way ANOVA calculated using TumGrowth[5]. Piecewise growth curve regression was applied with a breakpoint at the time of the first measurement after the start of treatment. All groups compared to all other groups and Holm–Bonferroni method applied for multiple comparisons (adjusted *p* values shown).
- Radiotherapy and radiotherapy + reovirus treatment led to significantly longer survival than controls. Combination treatment generated longer survival than radiotherapy alone, though this difference was not statistically significant (D). Survival comparisons were by log-rank test with all groups compared to all other groups, and Bonferroni correction for multiple comparisons (adjusted alpha level: 0.0083).
- In primary tumours, neither single treatment significantly changed the CD8+ T cell infiltration, however combination treatment generated a 15-fold increase in CD8+ cell infiltration compared with controls (478 vs 31 cells per mm^2 of tumour; $p = 0.02$ [E, F]). In abscopal tumours, CD8+ T cell infiltration was increased by either reovirus alone, or combination treatment, though the effect was smaller than in primary tumours (G, H). Comparisons by one-way ANOVA with Tukey's test for multiple comparisons (adjusted *p* values shown).

Combination treatment with radiotherapy, reovirus and α-PD-1 prolongs survival

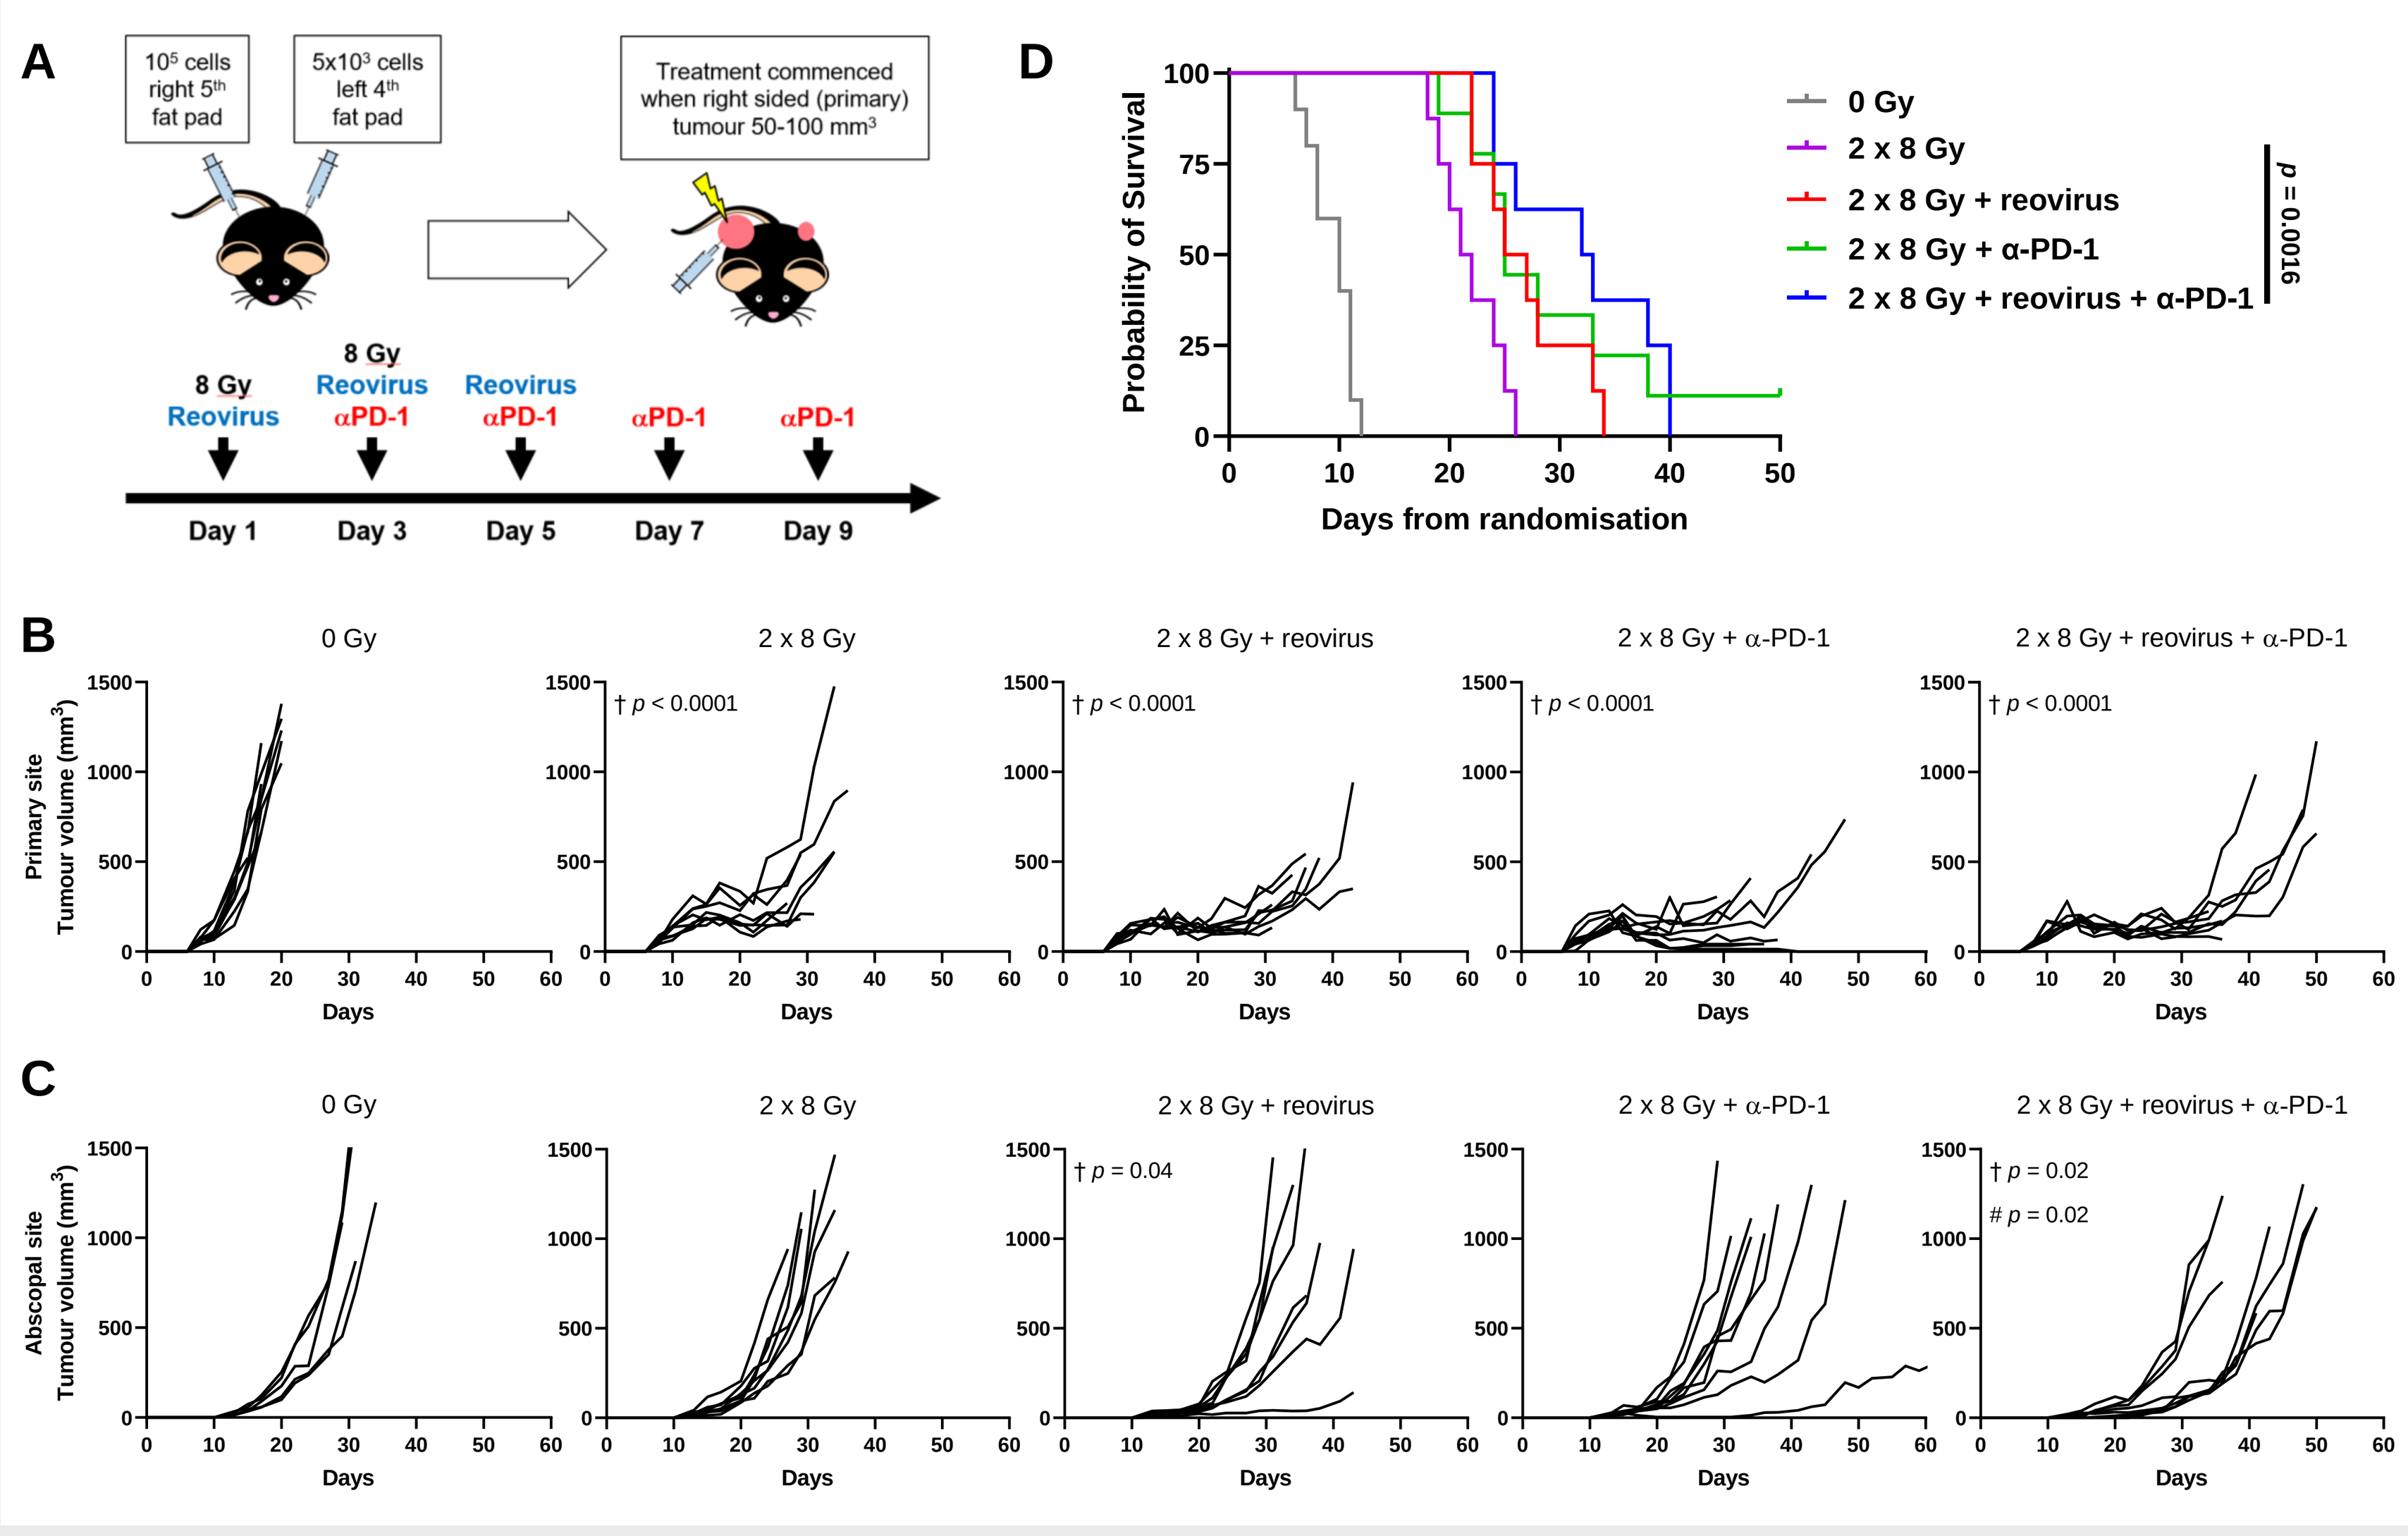
A
10⁵ cells right 5th fat pad
5x10³ cells left 4th fat pad
Treatment commenced when right sided (primary) tumour 50-100 mm³
8 Gy Reovirus
8 Gy Reovirus αPD-1
Reovirus αPD-1
αPD-1
αPD-1
Day 1
Day 3
Day 5
Day 7
Day 9
D
Probability of Survival
Days from randomisation
0 Gy
2 x 8 Gy
2 x 8 Gy + reovirus
2 x 8 Gy + α-PD-1
2 x 8 Gy + reovirus + α-PD-1
p = 0.0016
B
Primary site Tumour volume (mm³)
† p < 0.0001
Days
C
Abscopal site Tumour volume (mm³)
† p = 0.04
† p = 0.02
p = 0.02

- BLBC organoids were orthotopically injected bilaterally in the mammary fat pad of C57BL/6 mice. When the primary tumour reached 50-100 mm^3, mice were randomised to treatment with either sham radiotherapy, radiotherapy (2 x 8 Gy), radiotherapy + IT reovirus (3×10^8 pfu), radiotherapy + α-PD-1, or radiotherapy + reovirus + α-PD-1 (A).
- In primary tumours all treatment groups showed significant growth delay compared with controls, but no combination treatment was significantly different to radiotherapy alone (B).
- In abscopal tumours, radiotherapy + reovirus and radiotherapy + reovirus + α-PD-1 generated significantly reduced tumour growth rate compared to controls.
- Of all treatment combinations tested, only radiotherapy + reovirus + α-PD-1 led to reduced abscopal tumour growth and longer survival compared with radiotherapy alone (C, D).
- † = comparison with controls, # = comparison with radiotherapy alone

Conclusions

- In an organoid model of murine BLBC, combination treatment with reovirus and radiotherapy generated CD8+ infiltrates in primary and abscopal tumours, suggesting a local and systemic immune response to treatment.
- Combination treatment with radiotherapy, reovirus and α-PD-1 significantly prolonged survival though did not generate cures or long-term tumour control.
- Investigations are ongoing to explore the mechanisms for these effects and identify immune pathways which may be targeted to further improve treatment responses.

References

1. Sørlie et al. Gene expression patterns of breast carcinomas distinguish tumor subclasses with clinical implications. PNAS. 2001;98(19):10869–74.
2. Schmid et al. Atezolizumab and Nab-Paclitaxel in Advanced Triple-Negative Breast Cancer. NEJM. 2018; 379(22):2108-2121.
3. Samson et al. Intravenous delivery of oncolytic reovirus to brain tumor patients immunologically primes for subsequent checkpoint blockade. Sci Transl Med. 2018;10(422)
4. Twigger et al. Enhanced in vitro and in vivo cytotoxicity of combined reovirus and radiotherapy. Clin Cancer Res. 2008;14(3):912-23.
5. Enot et al. TumGrowth: An open-access web tool for the statistical analysis of tumor growth curves. Oncoimmunology. 2018;7(9):e1462431

Contact

The Institute of Cancer Research
Division of Breast Cancer Research
237 Fulham Road, London, SW3 6JB
T +44 207 153 5077
E ramsay.singer@icr.ac.uk