Combination Therapy with Oncolytic Viruses and CAR T Cells

Tim Kottke[1], Laura Evgin[1], Jacob van Vloten[1], Rosa Maria Diaz[1], Jill Thompson[1], Jason Tonne[1], Seth Eisenman[1], Gloria Olivier[1], Christopher B Driscoll[1], Gary Archer[2], John Sampson[2], Jose Pulido[3], Luis Sanchez Perez[2], Matt Coffey[4], Richard Vile[1,5,*]

[1]Department of Molecular Medicine, Mayo Clinic, Rochester, MN, USA; [2]Department of Neurosurgery, Duke University [3]Department of Ophthalmology, Mayo Clinic, Rochester, MN, USA; [4]Oncolytics Biotech Incorporated, Calgary, Canada; [5]Department of Immunology, Mayo Clinic, Rochester, MN, USA

Abstract

Oncolytic viruses (OVs) replicate preferentially in cancer cells and initiate a pro-inflammatory cascade that stimulates the production of chemokines and cytokines within the tumour micro-environment. We hypothesized that this intra-tumoral cytokine storm could reverse the immune suppressive micro-environment of solid tumours and change it to one which favours the recruitment of activating immune cells. In this respect, we hypothesized that an inflammatory remodeling of the tumor microenvironment by oncolytic viruses would overcome those factors which restrict CAR T infiltration, persistence and function and would, therefore, enhance both the recruitment and anti-tumor function of CAR T cells. We show here that oncolytic viruses, such as reovirus and VSV, do indeed induce a robust proinflammatory shift in the cytokine and chemokine profile of the tumor. However, intra-tumoral infection was also associated with significant attrition of CAR T cells acting through both T cell intrinsic, and CAR T cell specific, effects. These effects were mediated, in part at least, by type I interferon released upon virus infection of tumors which promoted apoptosis, activation, and inhibitory receptor expression. We also show that specific designer modifications can be made to CAR T cells in order to modify their responsiveness to molecules in the tumor microenvironment to enhance their efficacy. Thus, when the Interferon receptor was knocked out in CAR T cells using CRISP/CAS, these cells were largely refractory to OV associated attrition and provided combinatorial therapy with intra-tumoral OV. As a direct result of these studies, we have also explored additional ways to combine CAR T cell therapy with oncolytic viruses to overcome the problems associated with negative oncolytic virus-derived "heat" in the tumor. In particular, we have shown that CAR T cells can carry oncolytic viruses as cargoes into tumors. This T cell-mediated virus carriage results in significantly enhanced therapy compared to unloaded CAR T cells, partly through enhanced delivery of, and oncolysis by, the carried OV and partly because the presence of virus leads to increased CAR T cell intrinsic functionality. In summary, we believe that OV can be used in combination with CAR T cell therapies to enhance the therapeutic efficacy of either modality alone.

Infection promotes chemokines, but no combination therapy

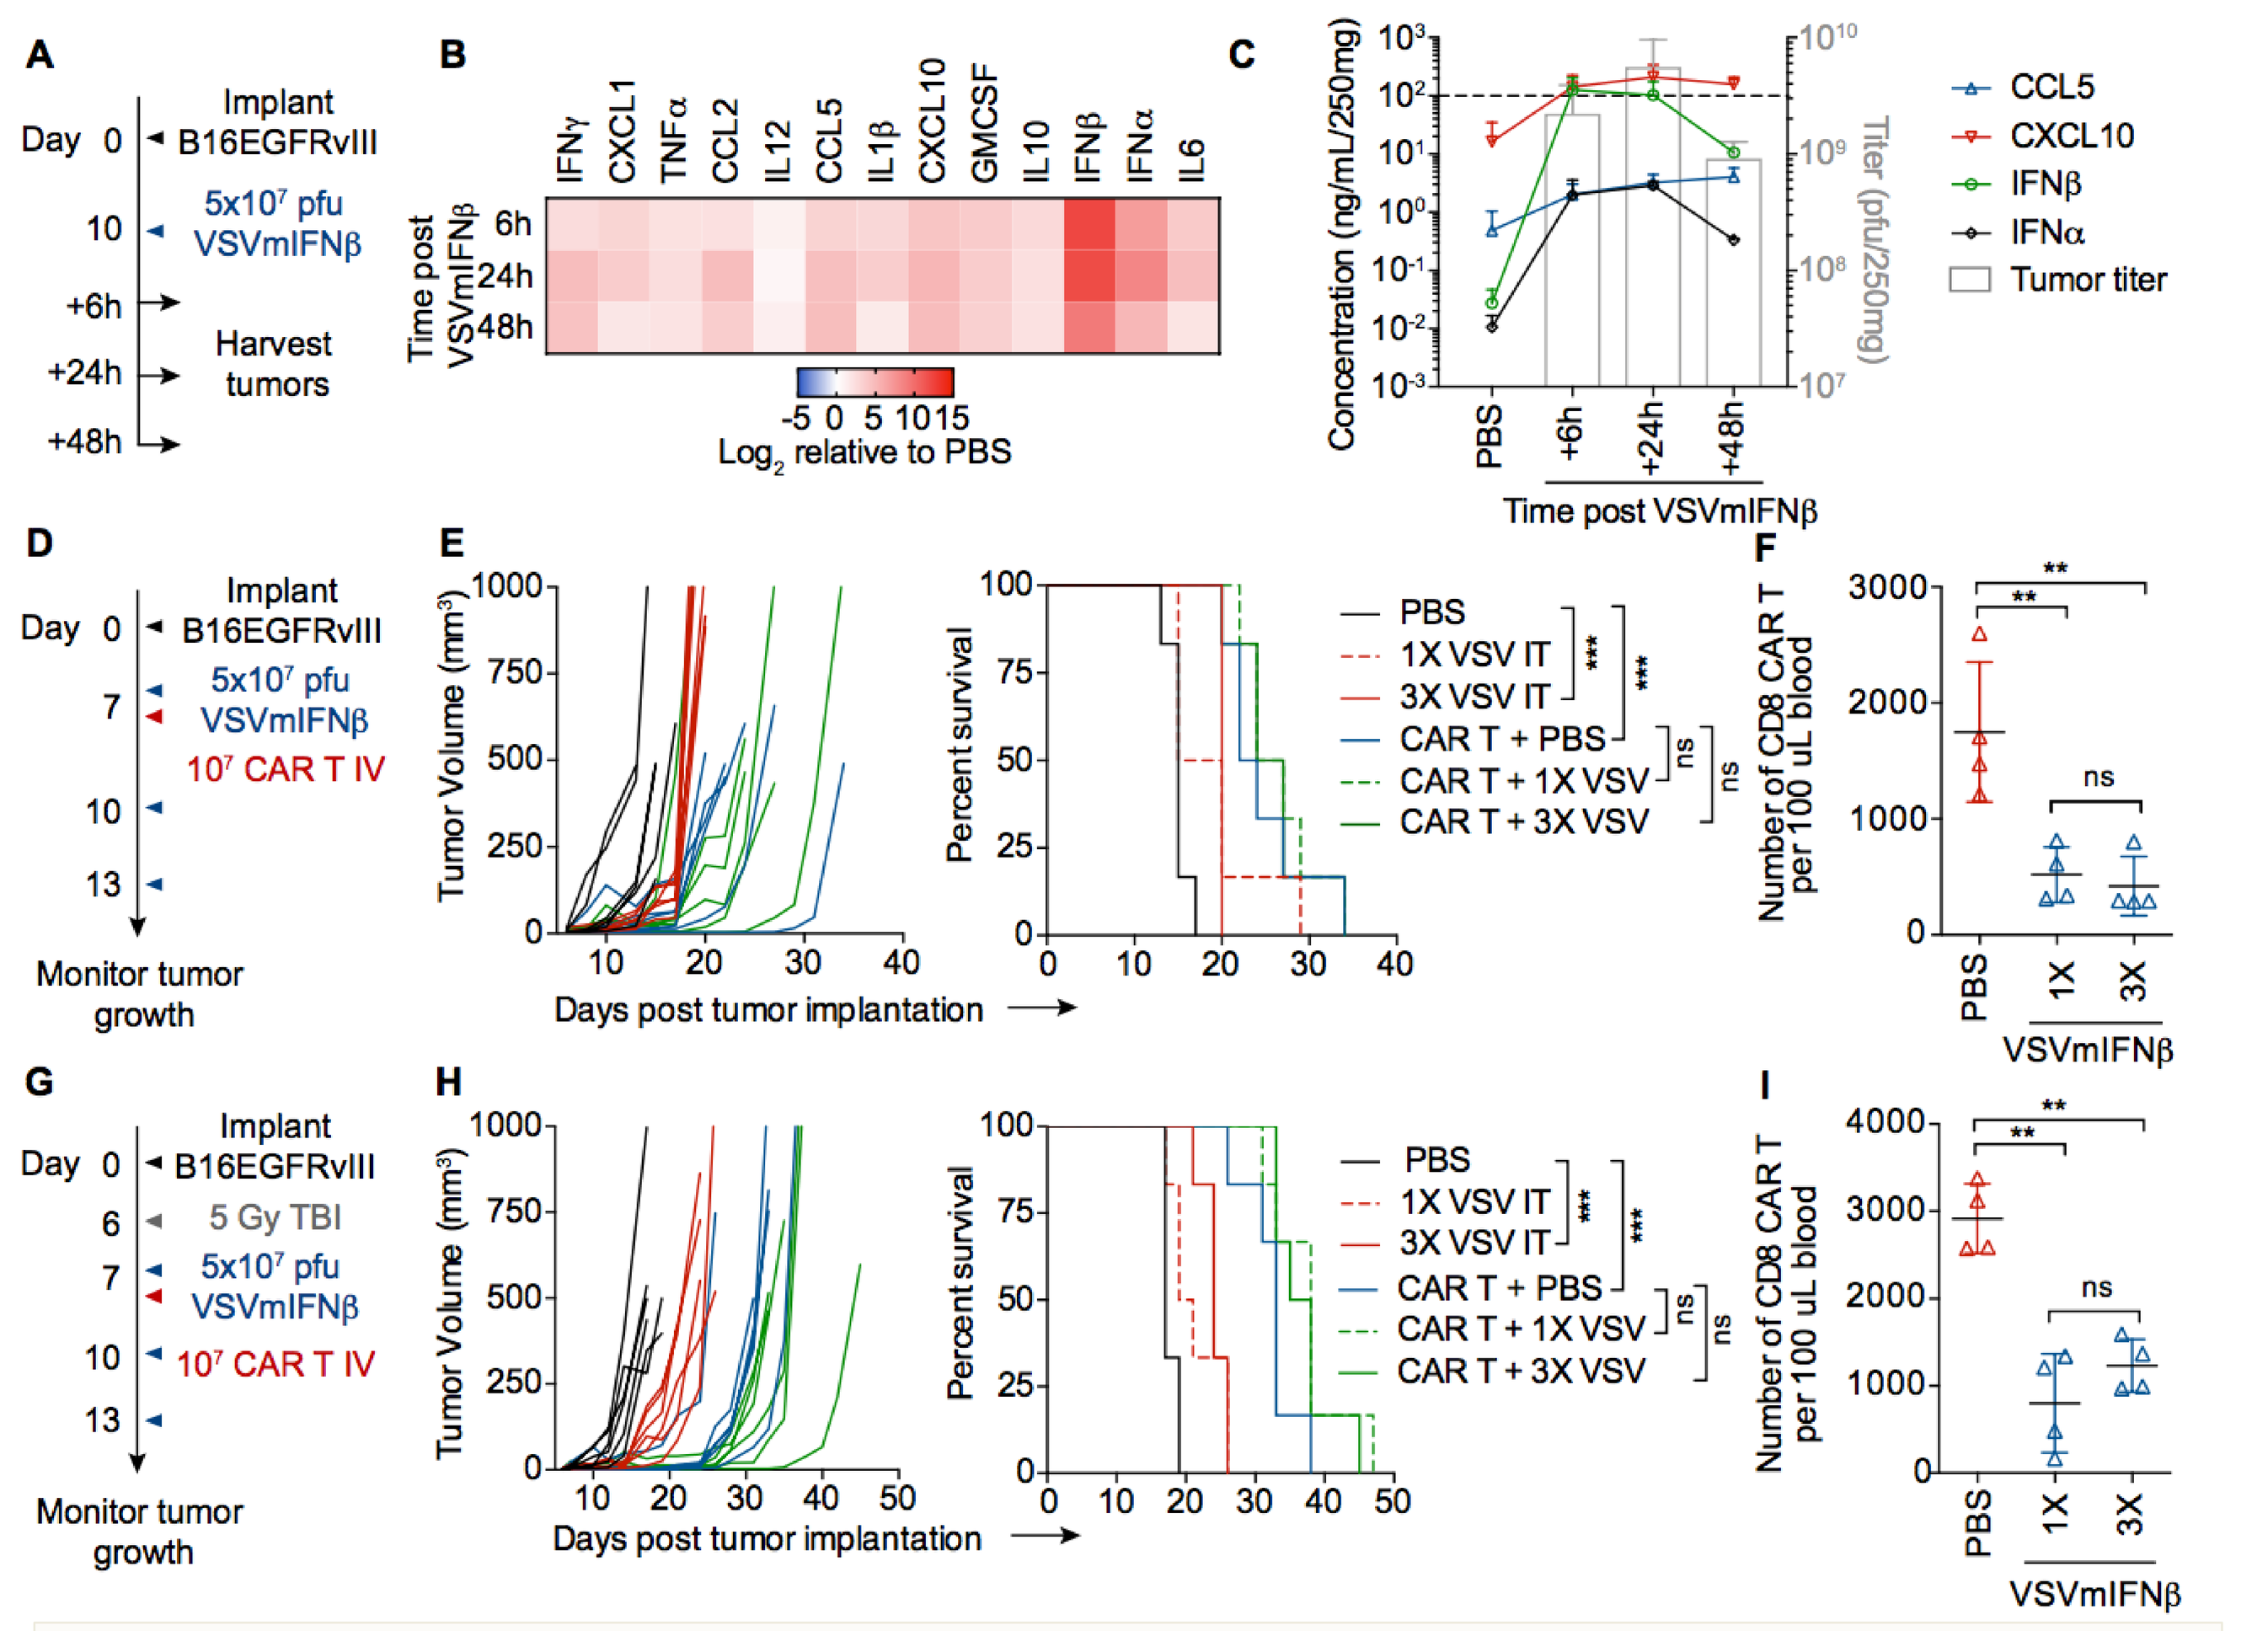

Figure 1. Oncolytic VSVmIFNβ infection has modest efficacy and promotes a favorable chemokine gradient for CAR T cell trafficking, yet combination does not improve survival over either modality. Mice bearing B16EGFRvIII tumors received a single intratumoral injection of 5x10^7 pfu of VSVmIFNβ or PBS and tumors were harvested 6, 24 or 48h post injection (**A**). Cytokine and chemokine concentration and infectious viral titer (**B, C**) was quantified in tumor homogenate and normalized to the average tumor weight. N=3 per group except 48h time point N=2. Dashed line indicates upper limit of standard curve for quantification. (**D**) Mice bearing B16EGFRvIII tumors were treated with 5x10^7 pfu VSVmIFNβ or PBS 6 hours prior to administration of 1x10^7 EGFRvIII CAR T cells on day 7. Select groups received 2 additional doses of 5x10^7 pfu VSVmIFNβ on days 10 and 13. N=6/group. (**E**) Tumor growth for select groups is shown in the left panel and overall survival is shown in the right panel. Circulating CD8 CAR T cells were quantified from the blood 7 days after adoptive transfer (**F**). (**G**) Mice treated as in **D** with a lymphodepleting dose of radiation (5 Gy TBI) on day 6. N=6/group. (**H**) Tumor growth for select groups is shown in the left panel and overall survival is shown in the right panel. Circulating CD8 CAR T cells were quantified from the blood 6 days after adoptive **transfer** (**I**). P-values were determined using the Log-rank Mantel-Cox test (**E,H**) or a one-way ANOVA with a Tukey post-test (**F,I**). ns $P > 0.05$; $^{**}P \le 0.01$; $^{***}P \le 0.001$.

VSVmIFNβ promotes CAR T cell attrition

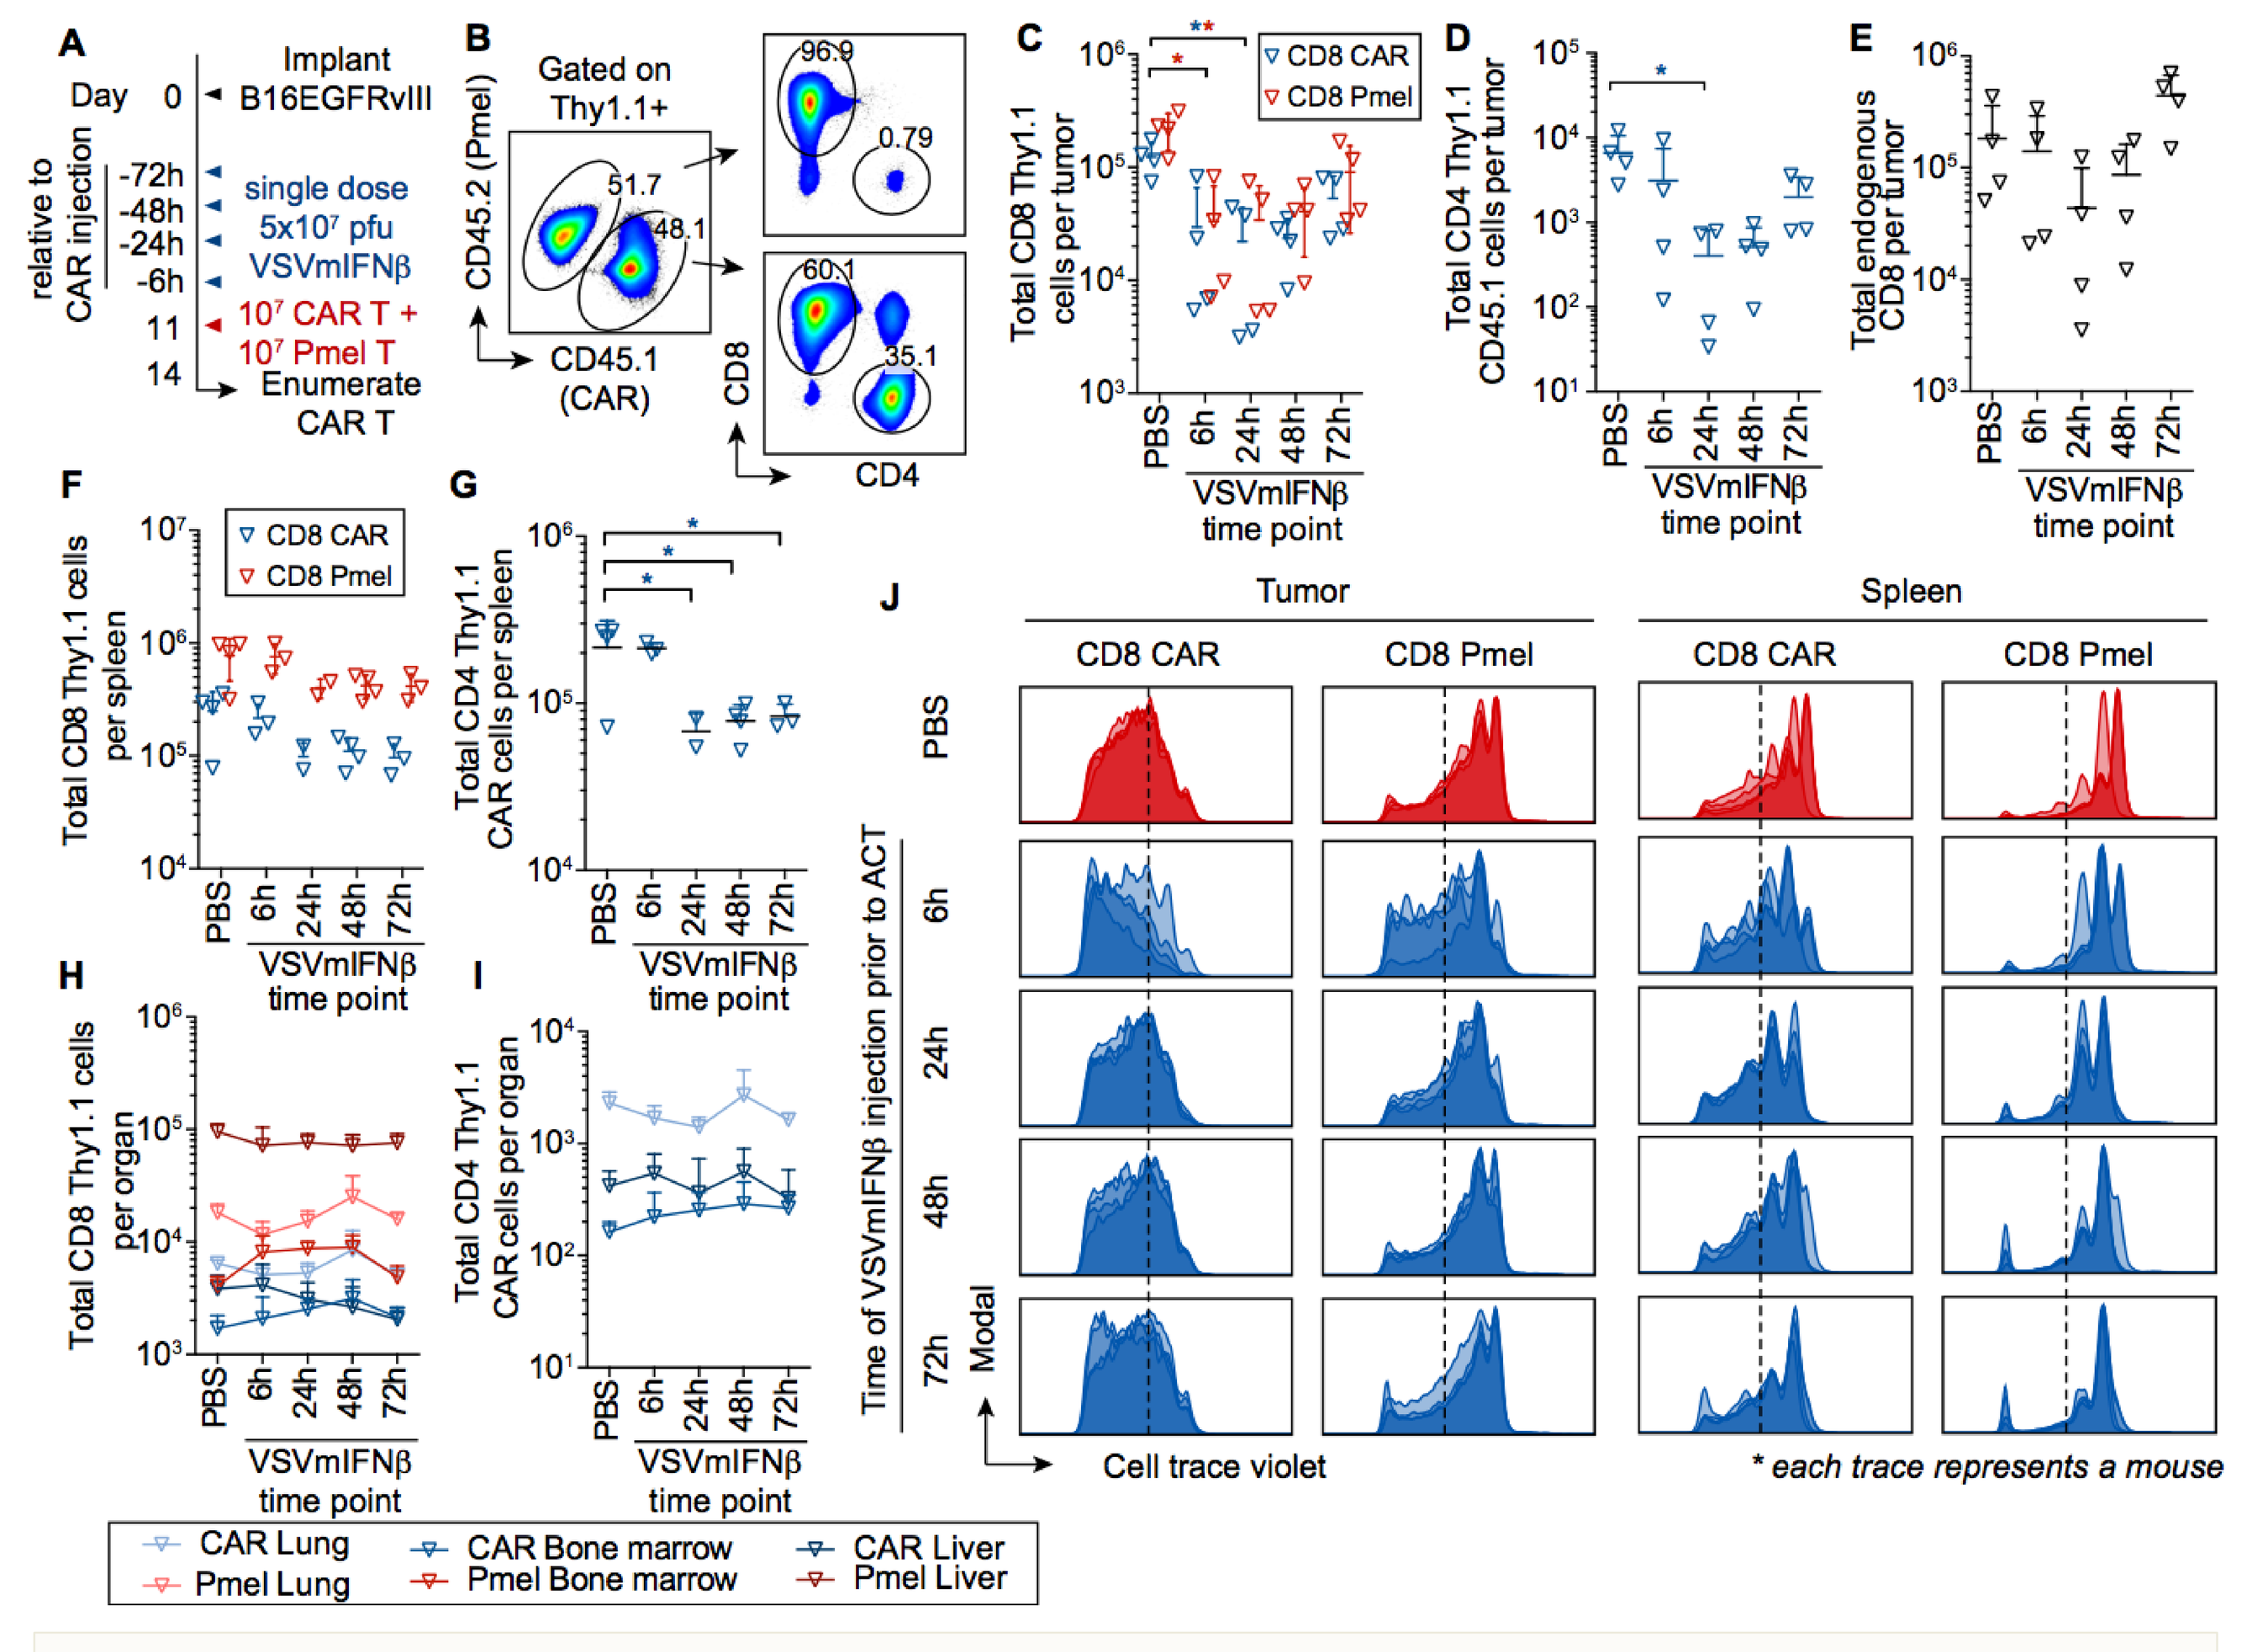

Figure 2. Oncolytic VSVmIFNβ infection promotes T cell attrition in the tumor and spleen. (**A**) Mice bearing B16EGFRvIII tumors received a single intratumoral injection of 5x10^7 pfu of VSVmIFNβ or PBS at various time points (6 to 72h) prior to adoptive transfer of 1x10^7 activated Pmel T cells + 1x10^7 EGFRvIII CAR T cells on day 11. N=2-4/group. (**B**) Both the CAR retrovirus and the Pmel mice express the congenic marker Thy1.1. To distinguish CAR T cells from C57 WT mice and Pmel T cells, CAR T cells were prepared from CD45.1 donor mice. Three days post adoptive transfer, the number of viable Thy1.1 CD8 (**C**) and CD4 (**D**) and endogenous CD8 (**E**) T cells were quantified in the tumor. The number of viable Thy1.1 CD8 (**F**) and CD4 (**G**) T cells were quantified in the spleen. The number of viable Thy1.1 CD8 (**H**) and CD4 (**I**) T cells were quantified in the lungs, liver, and left tibia and femur (bone marrow). (**J**) Pmel and CAR T cells were labeled with Cell trace violet and proliferation is shown in the tumor, and spleen. The Cell trace violet dilution for all four mice per group is overlaid. P-values were determined using a one-way ANOVA with a Tukey post-test (**C,D,G**) or an unpaired t-test (**L**). $^{*}P \le 0.05$; $^{**}P \le 0.01$; $^{***}P \le 0.001$.

Conclusions and perspectives

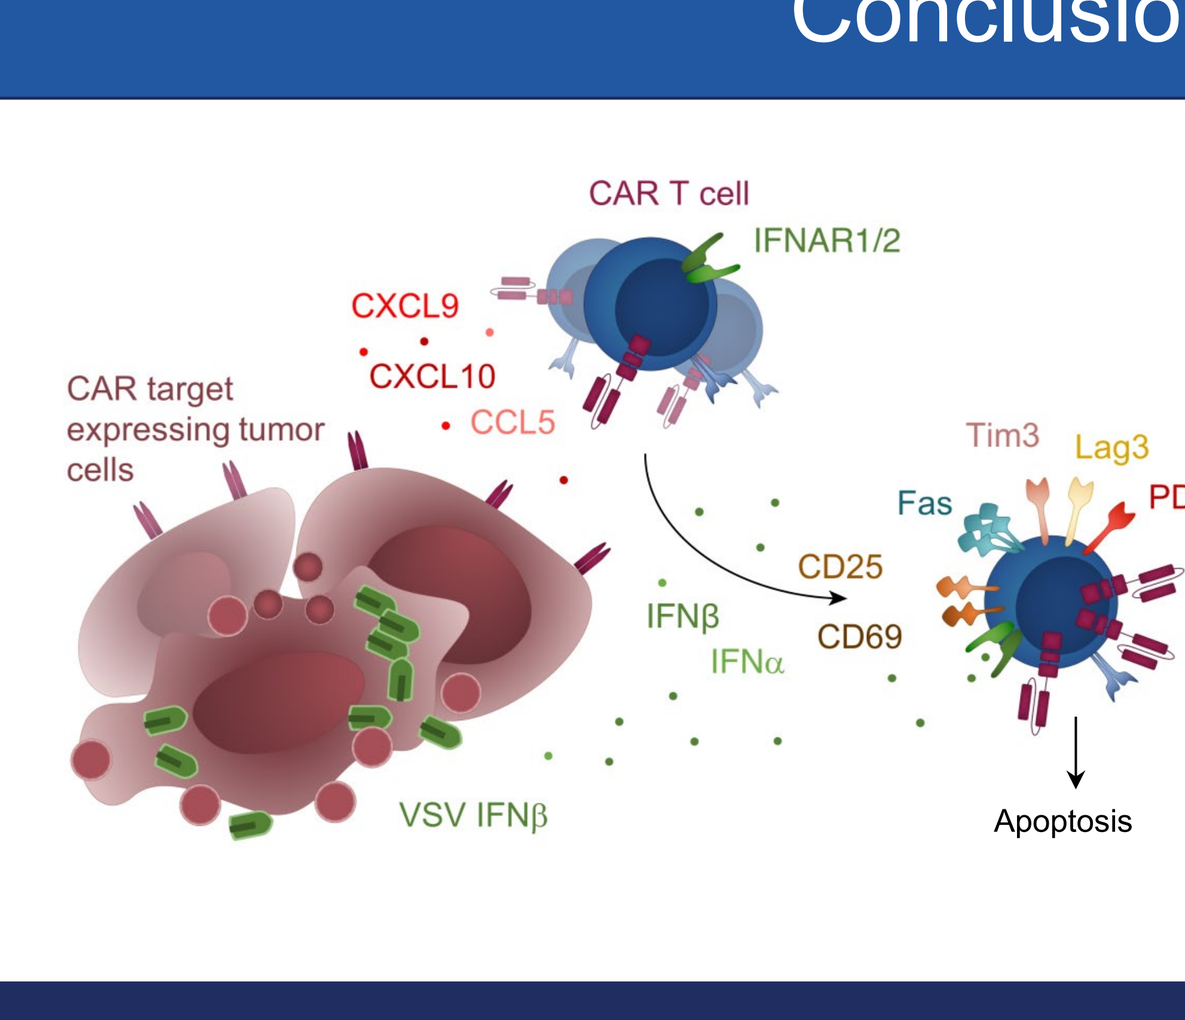
CAR T cell
IFNAR1/2
CXCL9
CXCL10
CCL5
CAR target expressing tumor cells
Tim3
Lag3
PD1
Fas
CD25
CD69
IFNβ
IFNα
VSV IFNβ
Apoptosis

-Tumoral infection with oncolytic viruses, such as VSVmIFNß, infection remodels the TME in complex ways that are both helpful and deleterious to CAR T cell therapy. Therefore, manipulation of CAR T cells to be resistant to those harmful effects, such as to confer type I IFN resistance, will help to fully capitalize on the complementary mechanisms of action of each platform.

-By loading oncolytic virus onto CAR T cells it is possible to generate more therapeutically effective CAR T cell therapy; This may be due to virus carriage and delivery to tumors by the CAR T as well as increasing the CAR T cell intrinsic activity against tumor. Moreover, by providing a further in vivo boost with virus, these CAR T cells can be further/re-activated to generate effective tumor cures.

CAR T cell attrition in virus infected tumors is type I IFN dependent

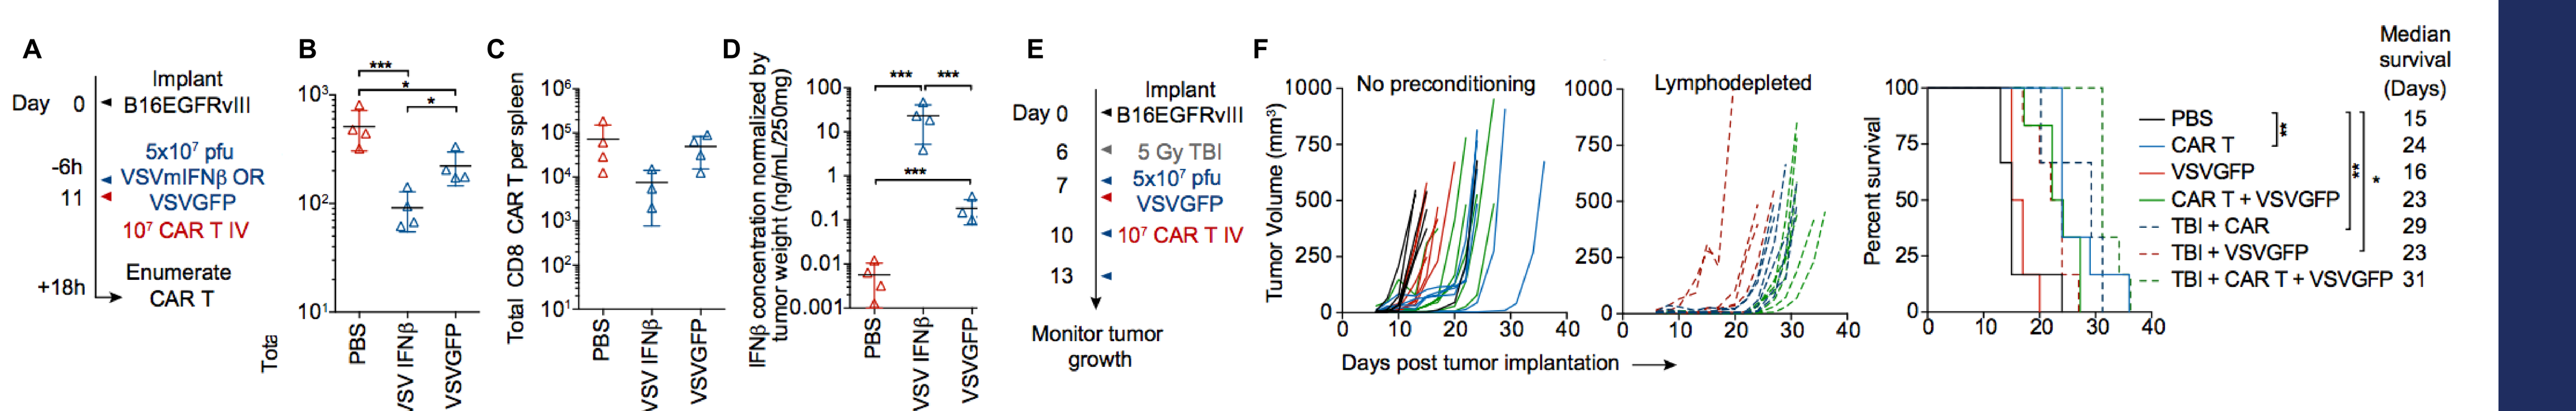

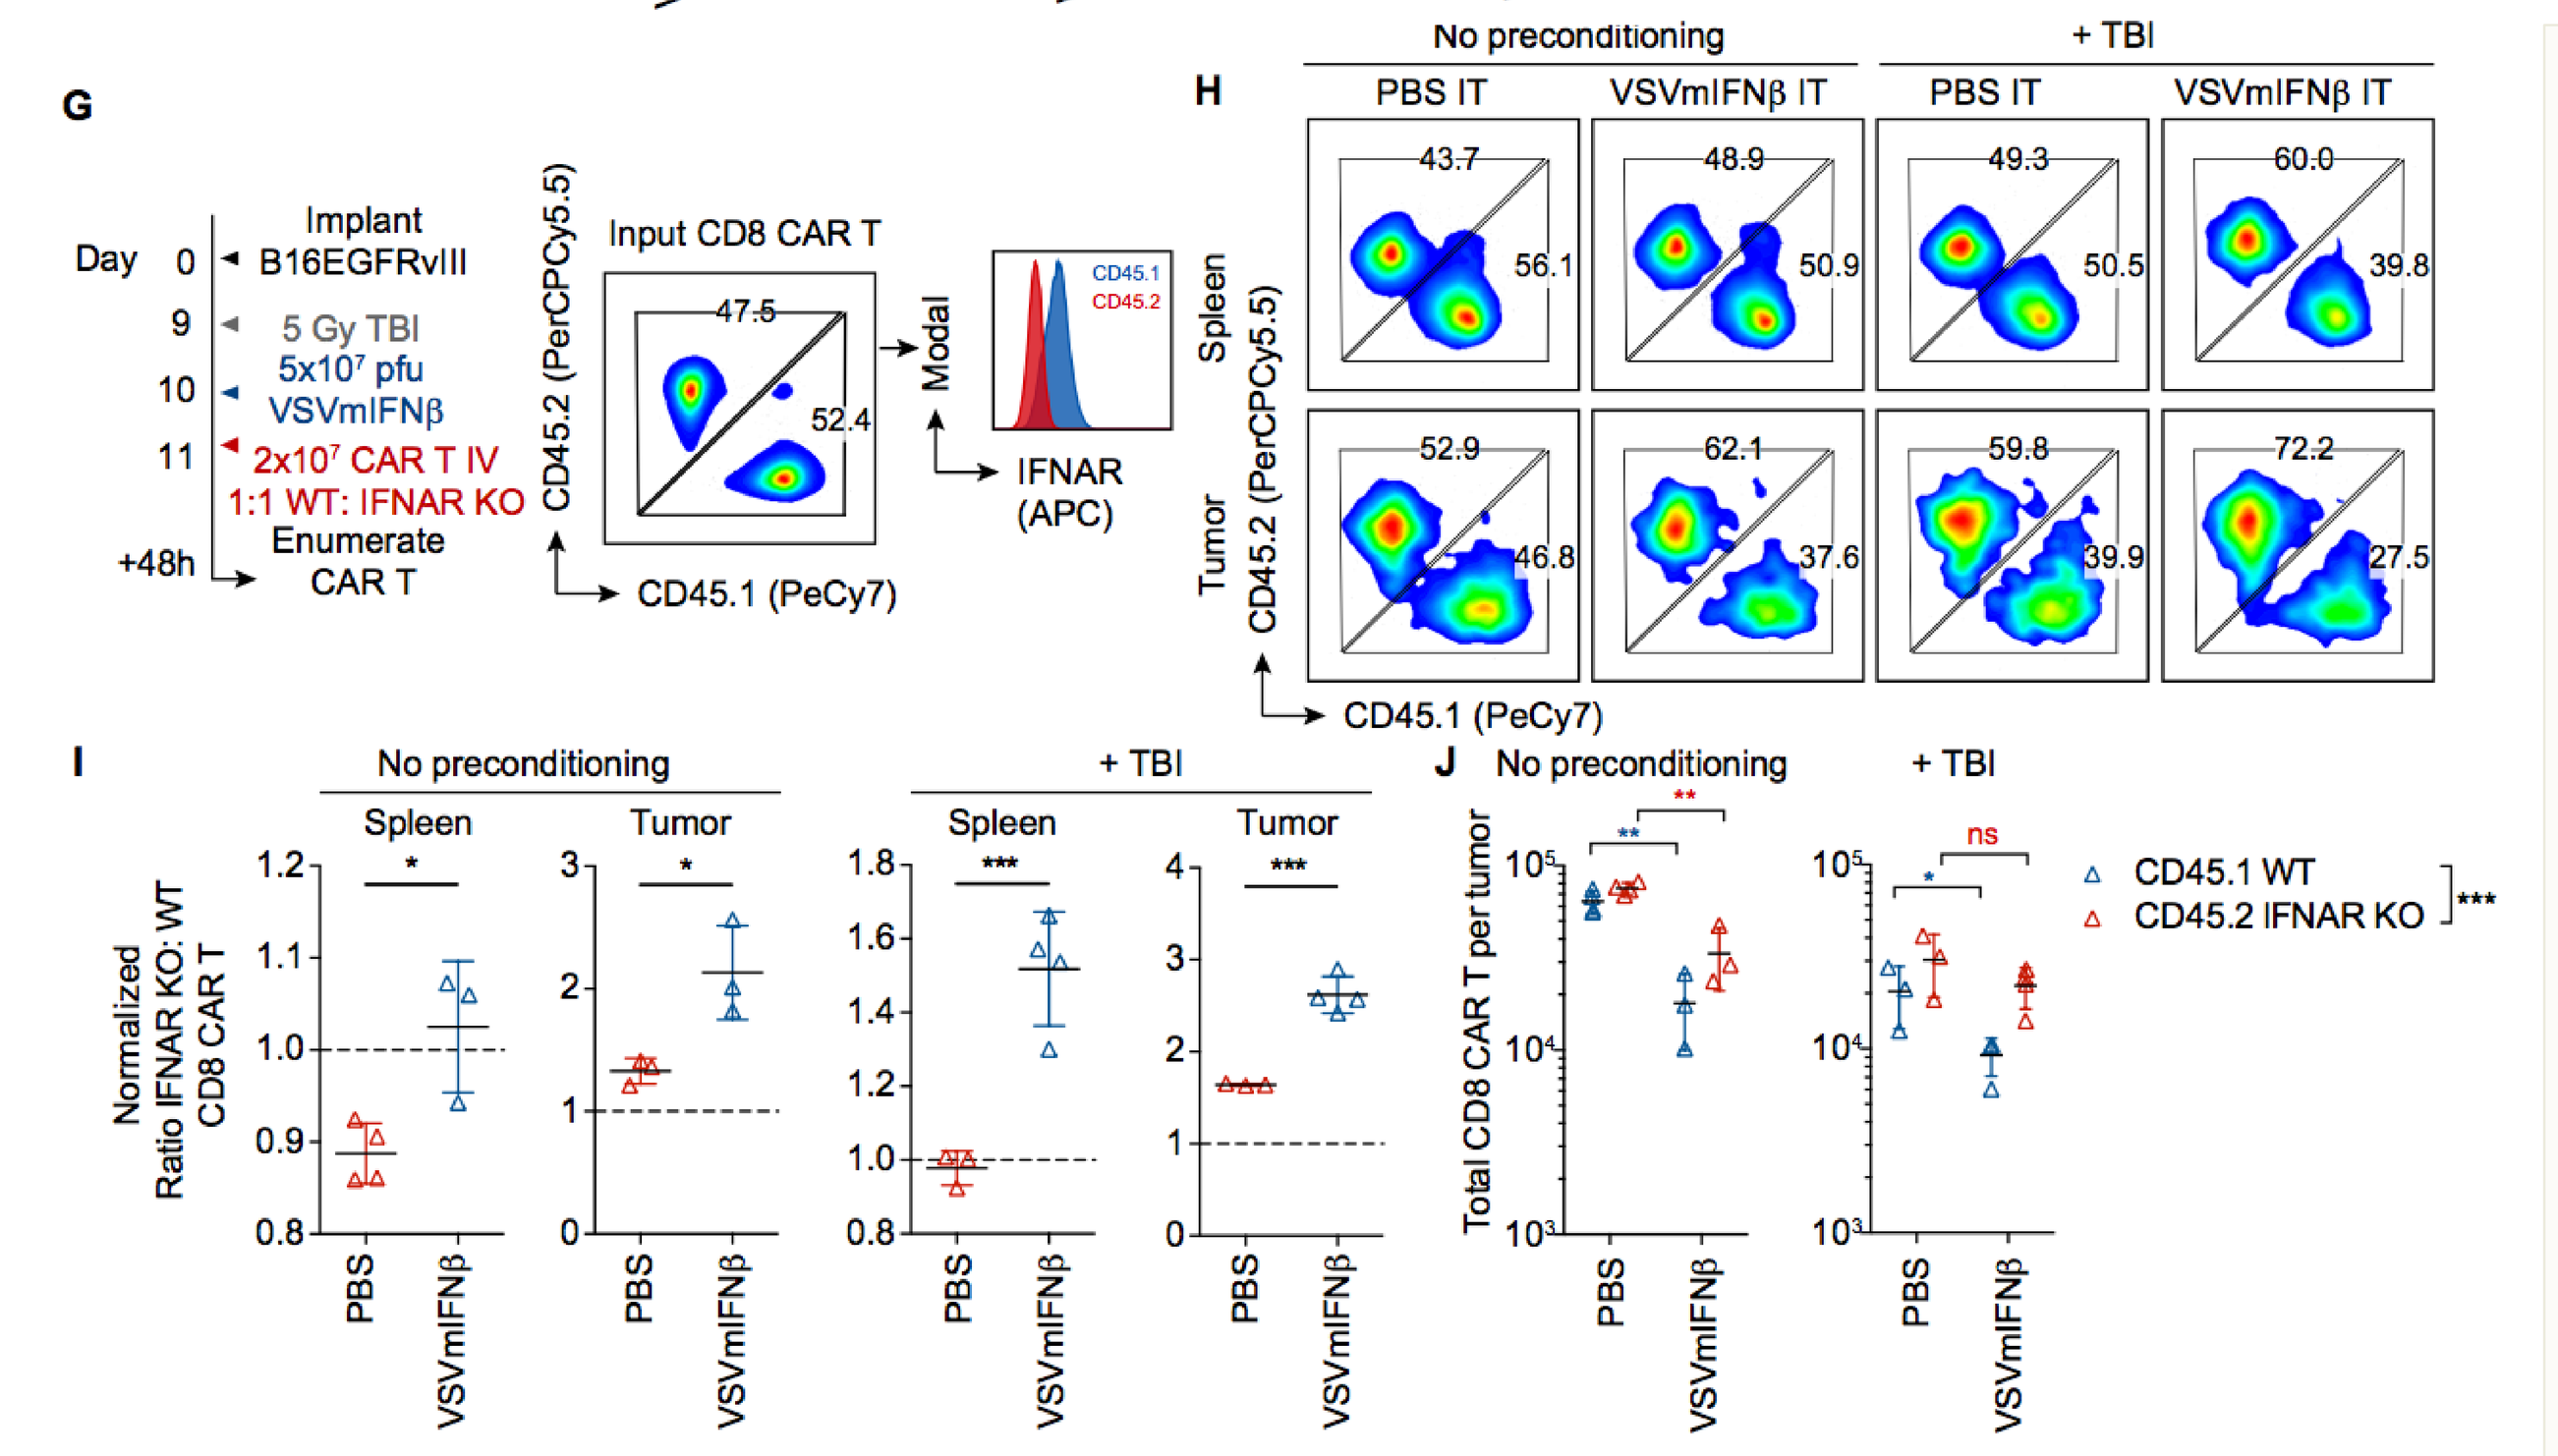

Figure 3. Oncolytic virus-associated type I IFN is deleterious to adoptively transferred CAR T cells. (**A**) Mice bearing B16EGFRvIII tumors received a single intratumoral injection of 5x10^7 pfu of VSVmIFNβ or VSVGFP or PBS 6 hours prior to adoptive transfer of 1x10^7 EGFRvIII CAR T cells on day 11. N=4/group. The next day, the number of viable CD8 CAR T cells was quantified in the tumor (**B**) and spleen (**C**). IFNβ was quantified in tumor homogenate and normalized by tumor weight (**D**). Mice bearing B16EGFRvIII tumors were treated with 5x10^7 pfu VSVGFP or PBS 6 hours prior to administration of 1x10^7 EGFRvIII CAR T cells on day 7 (**E**). Select groups received a lymphodepleting dose of radiation (5 Gy TBI) on day 6, and 2 additional doses of 5x10^7 pfu VSVmIFNβ on days 10 and 13. N=6/group. (**F**) Tumor growth is shown in the left panel and overall survival is shown in right panel. (**G**) Mice bearing B16EGFRvIII tumors received a single intratumoral injection of 5x10^7 pfu of VSVmIFNβ or PBS 48h prior to adoptive transfer of 1x10^7 WT CD45.1 EGFRvIII CAR T cells + 1x10^7 transgenic IFNAR1 KO CD45.2 EGFRvIII CAR T cells. N=3 or 4/group. (**H**) The ratio of IFNAR KO to WT CD8 CAR T cells in the tumor and in the spleen is shown for a single representative mouse per group. (**I**) The IFNAR KO: WT ratio normalized to the input ratio. (**J**)The total number of CD8 CAR T cells of each type was enumerated in the tumor and spleen. P-values were determined using a one-way ANOVA with a Tukey post-test (**B, D, H**), the Log-rank Mantel-Cox test (**F**), unpaired T tests (**I**) or a two-way ANOVA with a Sidak post-test (**J**). ns $P > 0.05$; $^{*}P \le 0.05$; $^{**}P \le 0.01$; $^{***}P \le 0.001$.

Type I IFN promotes apoptosis, activation, and upregulation of the CAR and inhibitory receptors

Figure 4. Recombinant IFNβ promotes apoptosis, activation, inhibitory receptor expression, and dysregulated expression of the CAR. (**A**) Annexin V expression on CD8 CAR T cells or untransduced (UTD) T cells were cultured in IL2 (50 U/mL) in the presence or absence of recombinant murine IFNβ and 1μM Ruxolitinib (JAK1/2 inhibitor) for 48 hours. (**B**) Annexin V expression on CD8 CAR T cells cocultured with B16EGFRvIII cells at an E:T ratio of 1:5 in the presence or absence of recombinant murine IFNβ and/or 1μM Ruxolitinib. Fas (**C**) or cleaved caspase (**D**) expression on CAR T cells or UTD cultured in IL2 with IFNβ and the percent of CD8 CAR T cells. Median fluorescence intensity (MFI) of CAR expression measured using Biotin-Protein L and streptavidin- PE (Protein L + SA PE) is compared to Annexin V in CAR T cells cultured in IL2 with or without IFNβ (**E**). CAR expression on CD8 CAR T cells cultured in IFNβ for 48 hours in the absence (**F**) or presence of target tumor cells (**G**). Representative (**H**) and mean (**I**) expression of CD25 and CD69 for CAR T cells cultured in IL2 with IFNβ in (**H**). Co-expression of the CAR and the activation markers for CD8 CAR T cells (**J**). (**K**) Mean expression of PD1, LAG3, TIM3 on CD8 CAR T or UTD cells cultured in IL2 or in the presence of B16EGFRvIII cells (E:T 1:5). (**L**) The fold change in inhibitory receptor and CAR expression is shown for cells grown in IL2 with additional recombinant IFNβ relative to culture in IL2 alone. (**L**) WT or transgenic IFNAR1 KO CAR T cells were cocultured with B16EGFRvIII (E:T ratio of 1:5) which were mock infected or infected with VSVmIFNβ 6 hours prior to coculture. Surface CAR expression (measured as in **E**) expressed as fold change relative to coculture with mock infected tumor cells. (**M**) Inhibitory receptor expression was quantified on WT or IFNAR1 KO CD8 CAR T cultured as in **L**. Means ± SD of three technical triplicates are shown. P-values were determined using a two-way ANOVA with a Tukey post-test (**A,B,C,D,F,G, I**). ns $P > 0.05$; $^{*}P \le 0.05$; $^{**}P \le 0.01$; $^{***}P \le 0.001$.

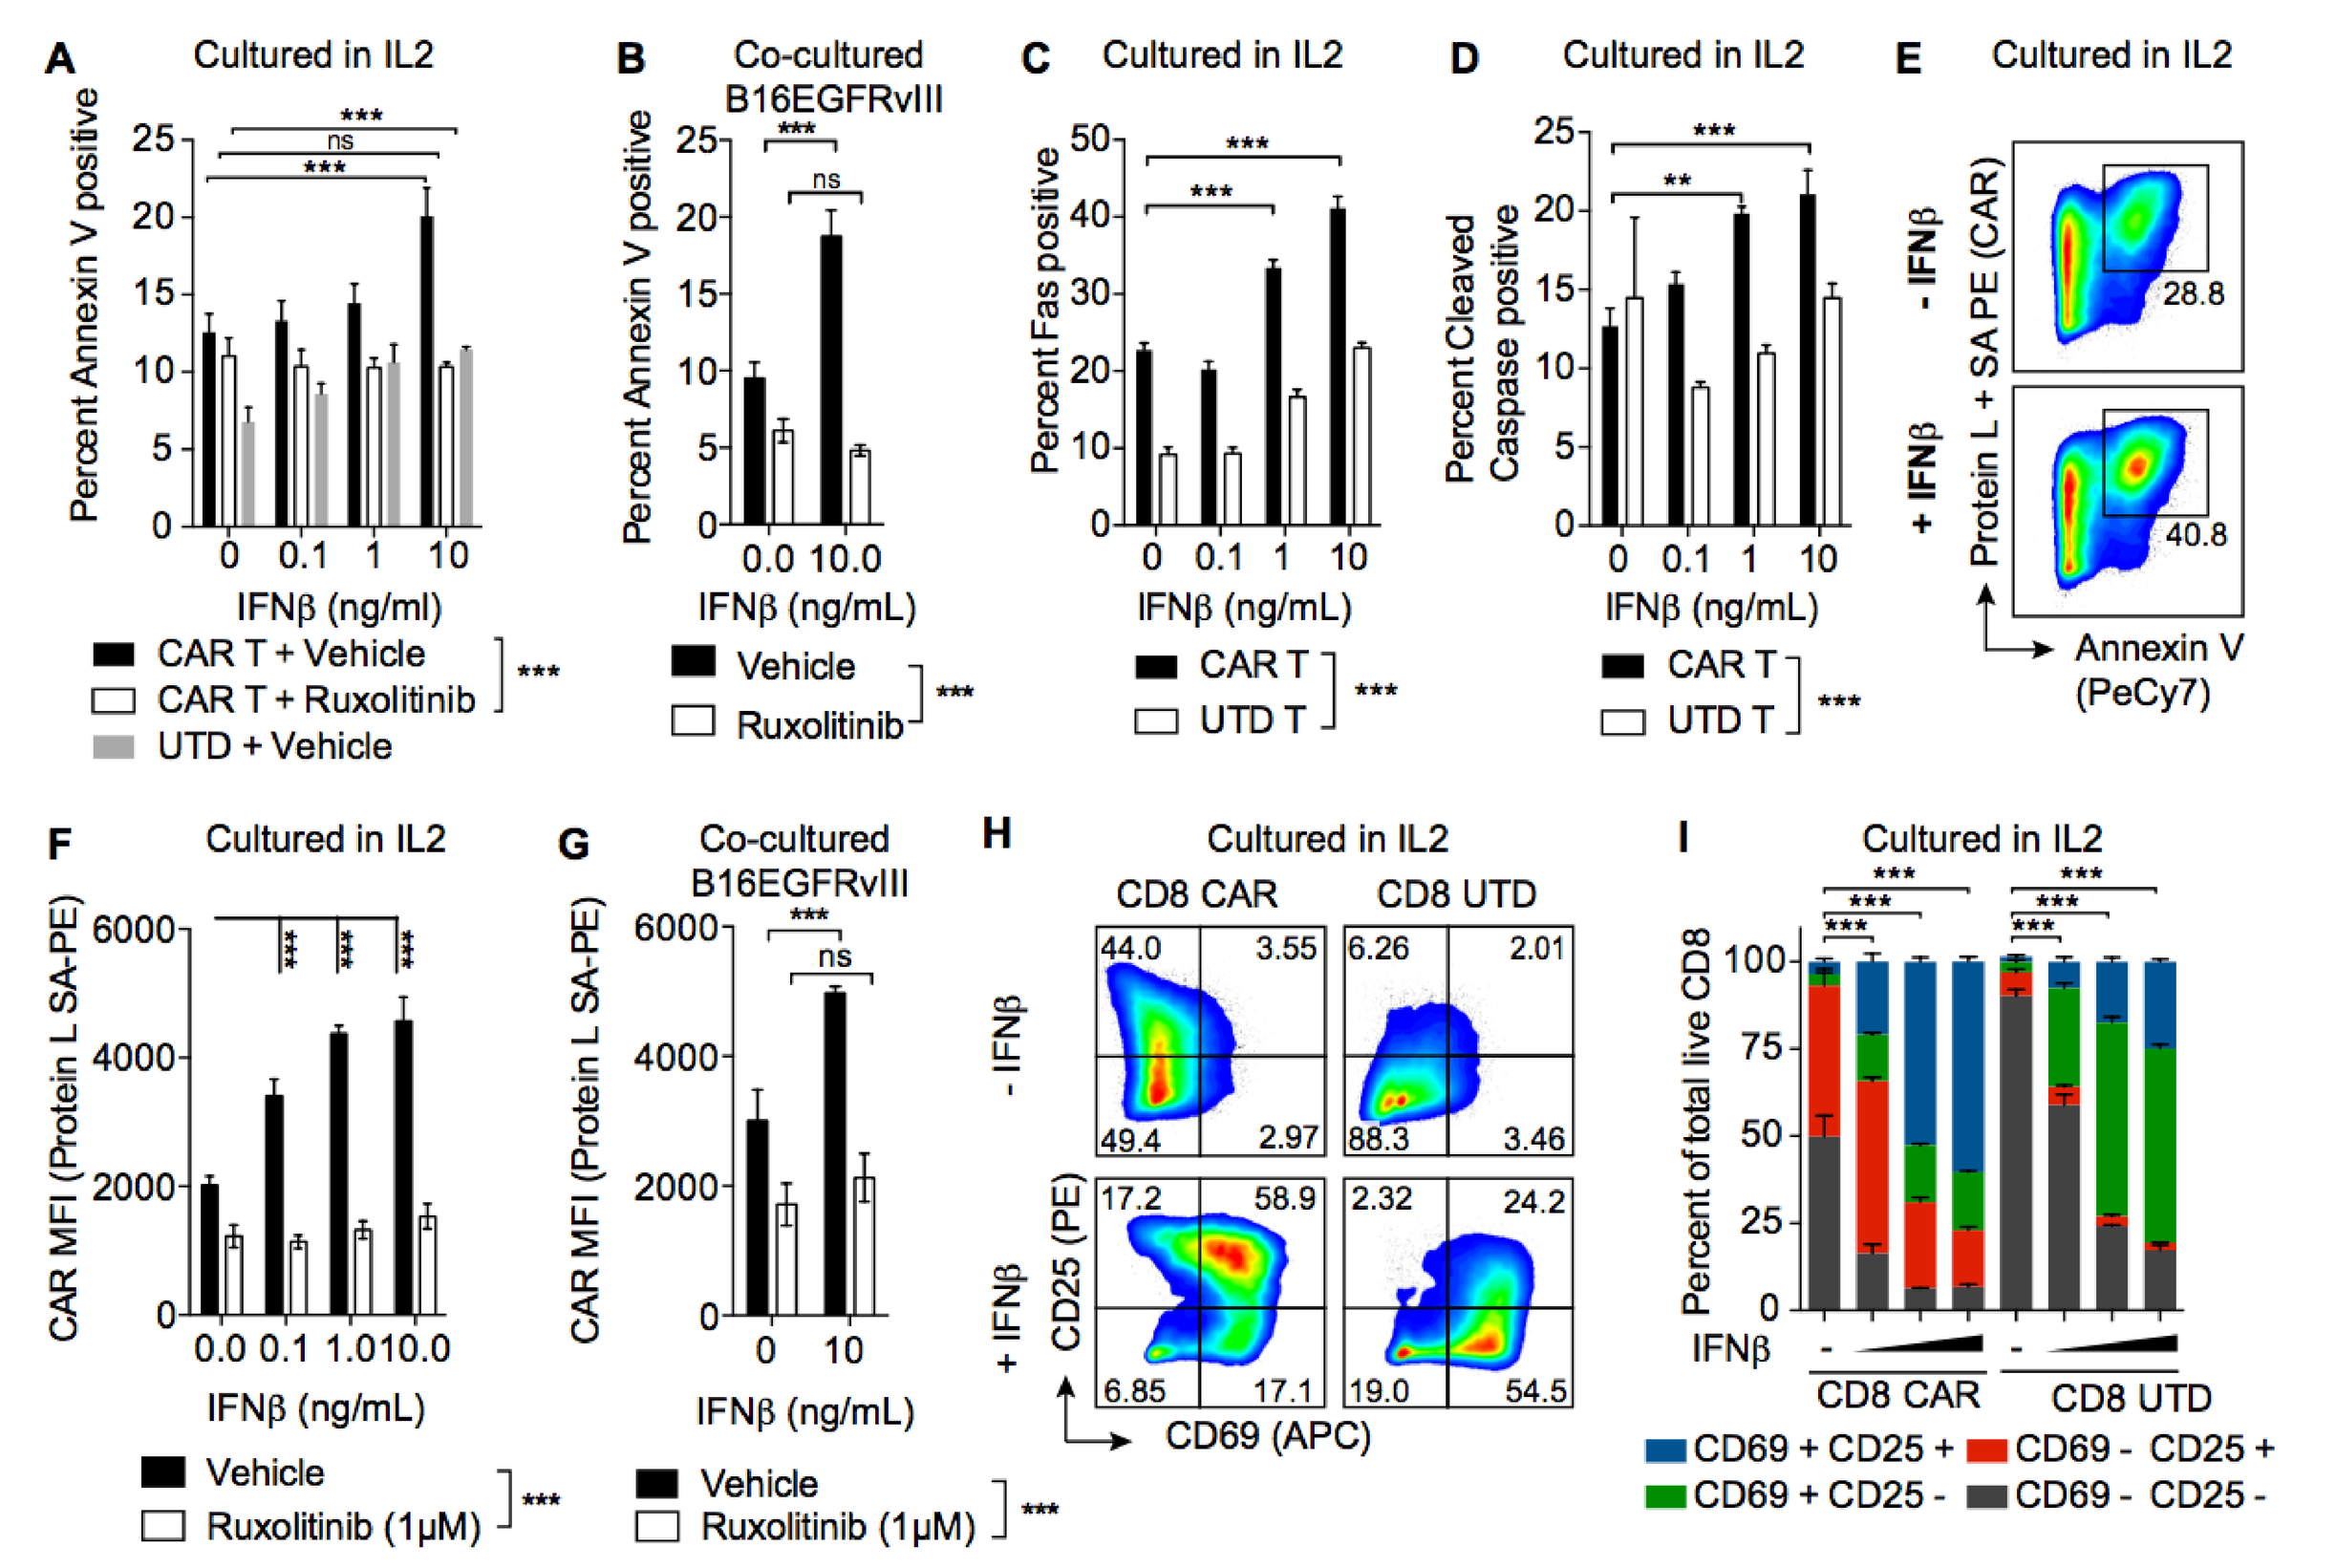

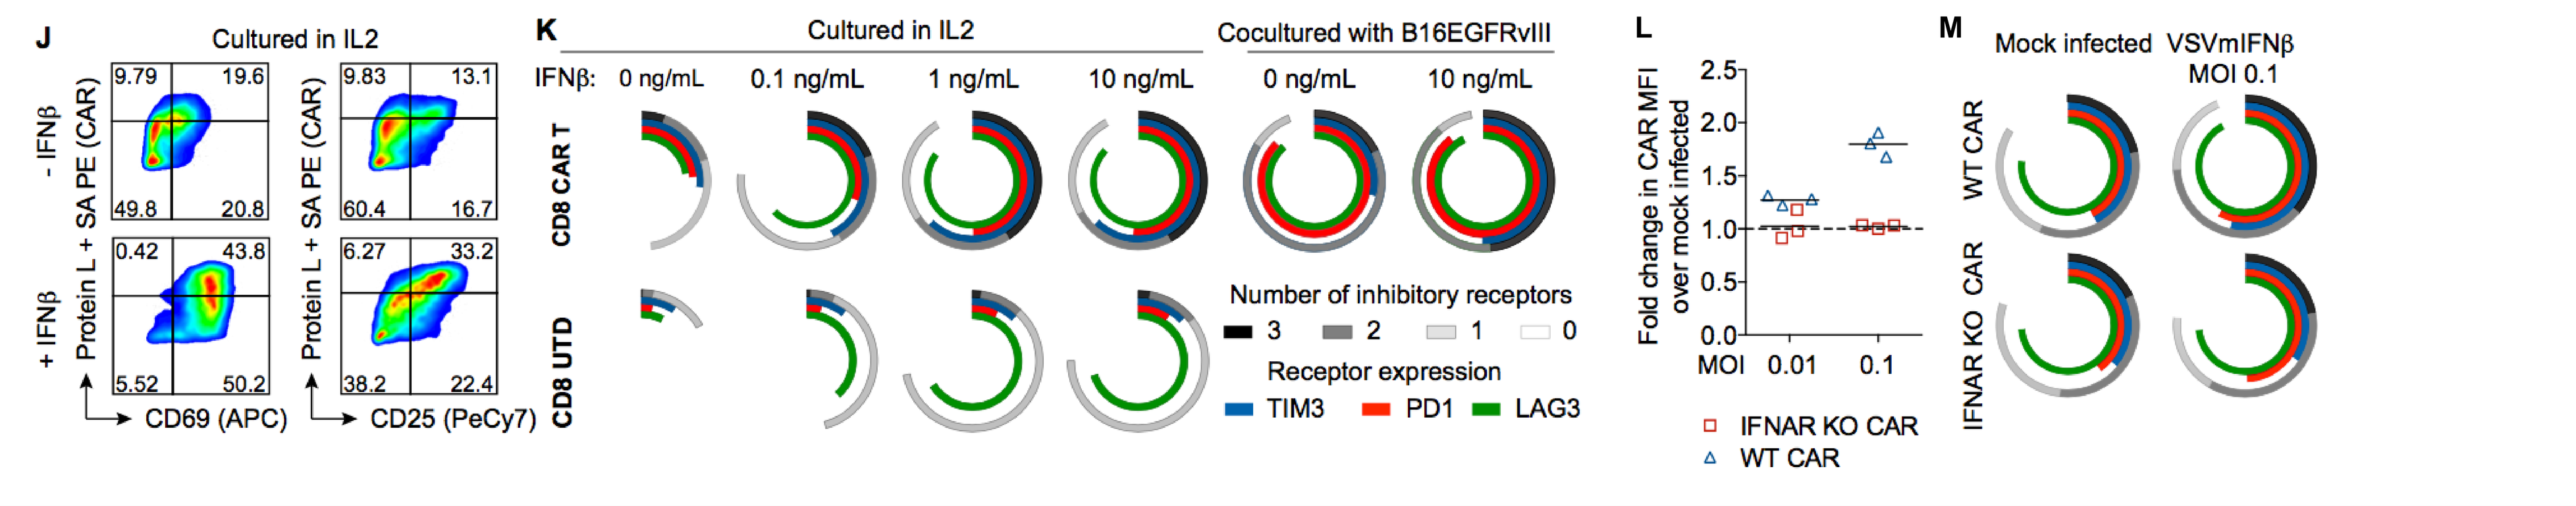

Effective Combination of Oncolytic Virus and CAR T Cell Therapies

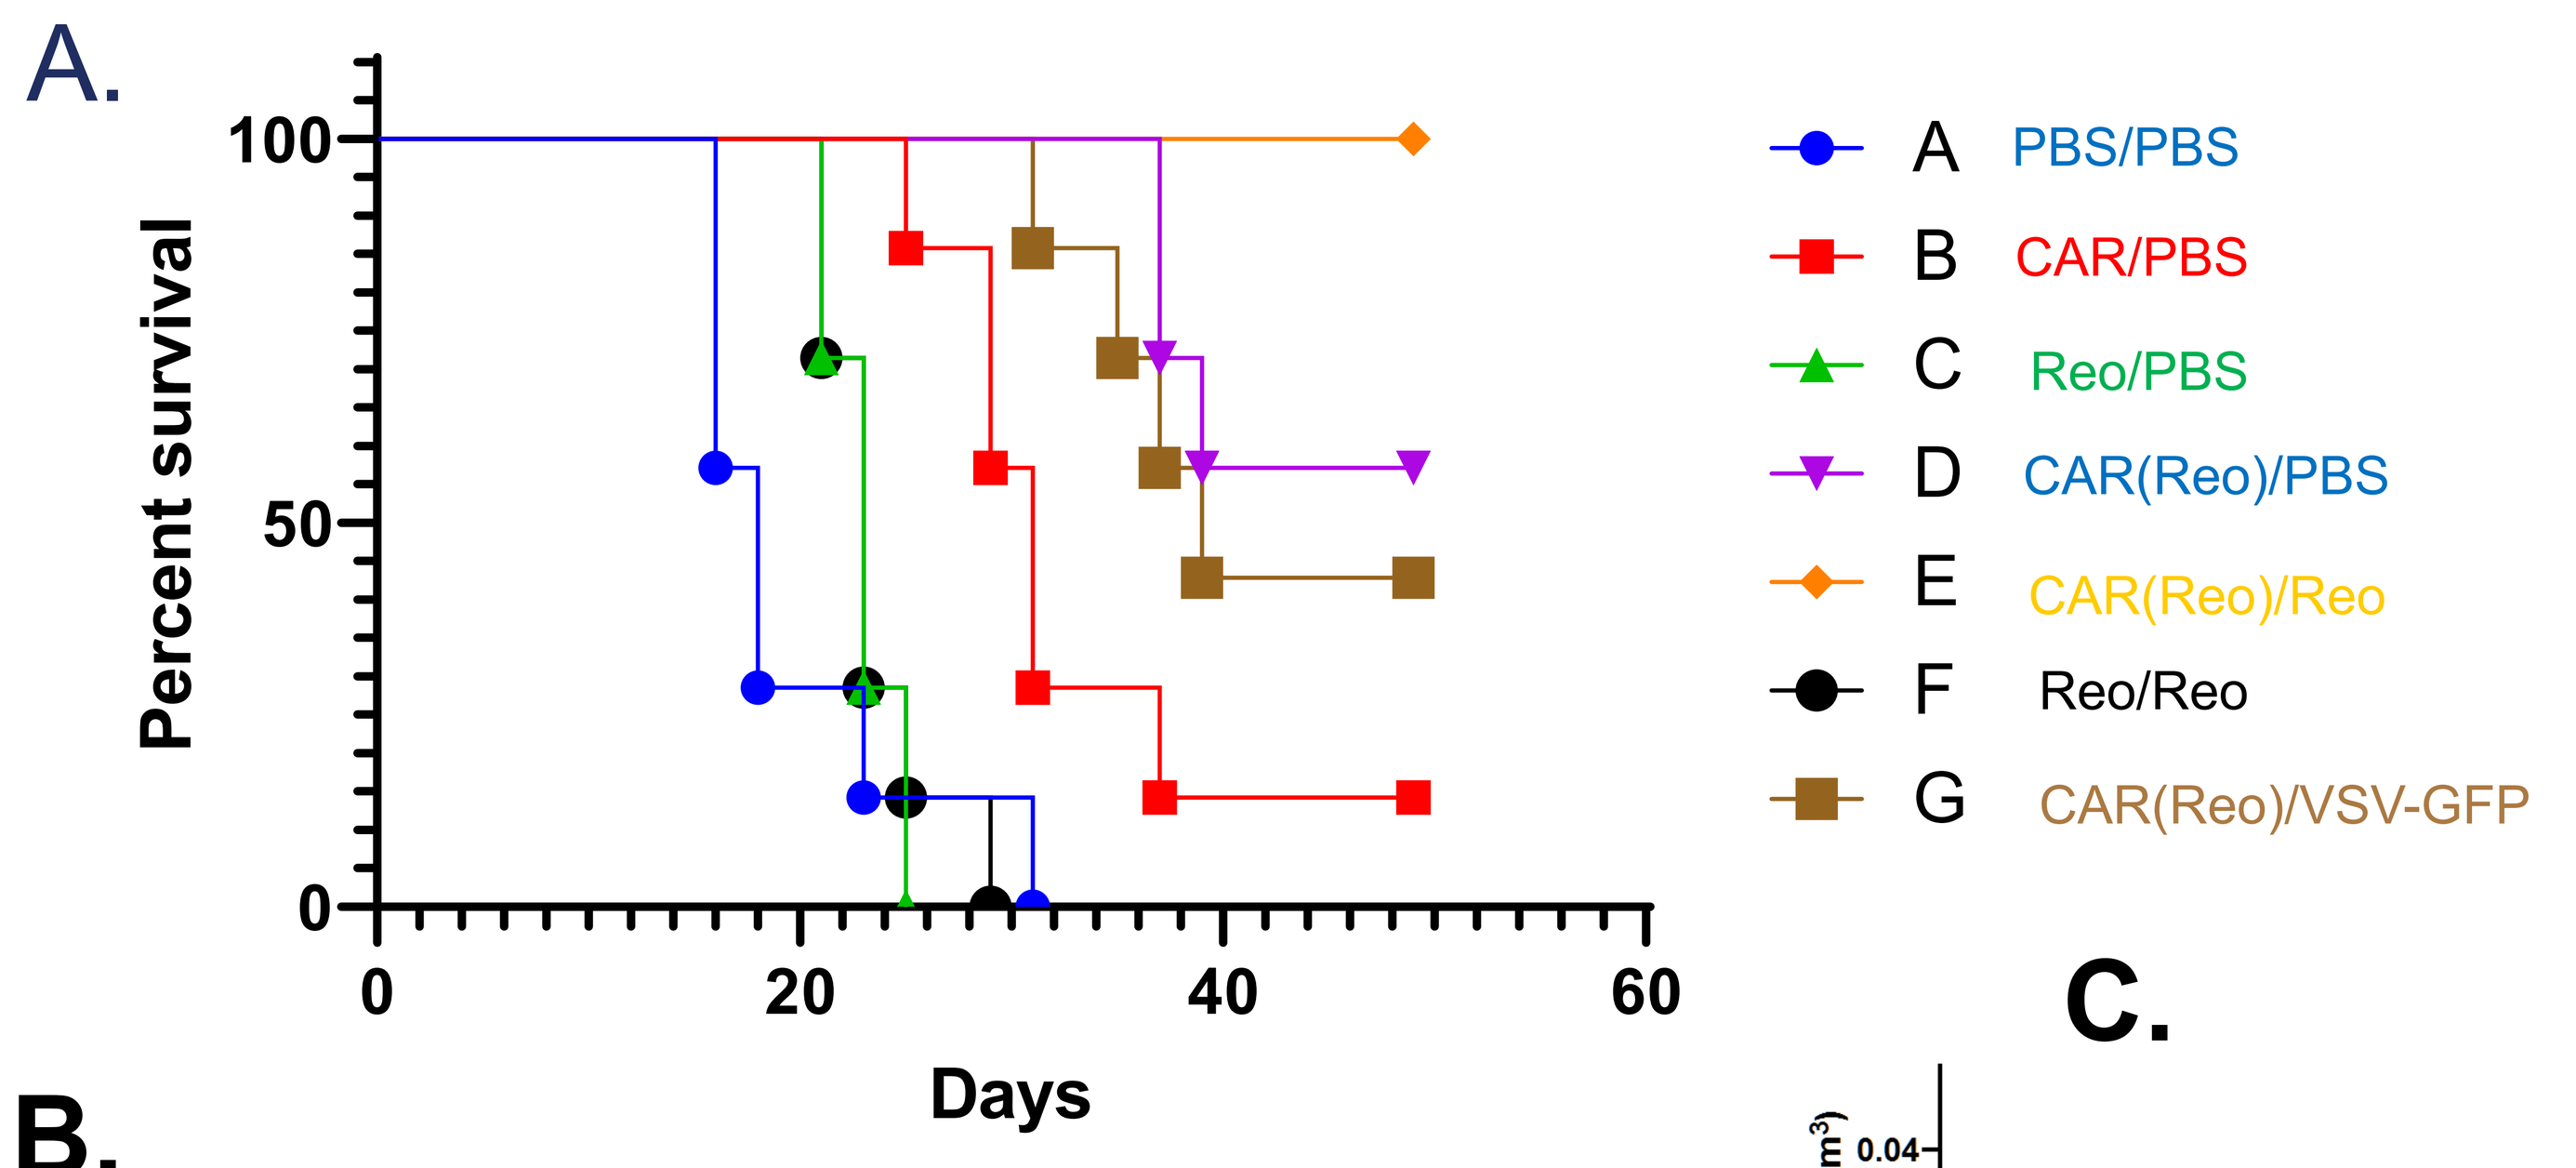
A.
Percent survival
Days
A PBS/PBS
B CAR/PBS
C Reo/PBS
D CAR(Reo)/PBS
E CAR(Reo)/Reo
F Reo/Reo
G CAR(Reo)/VSV-GFP

B.

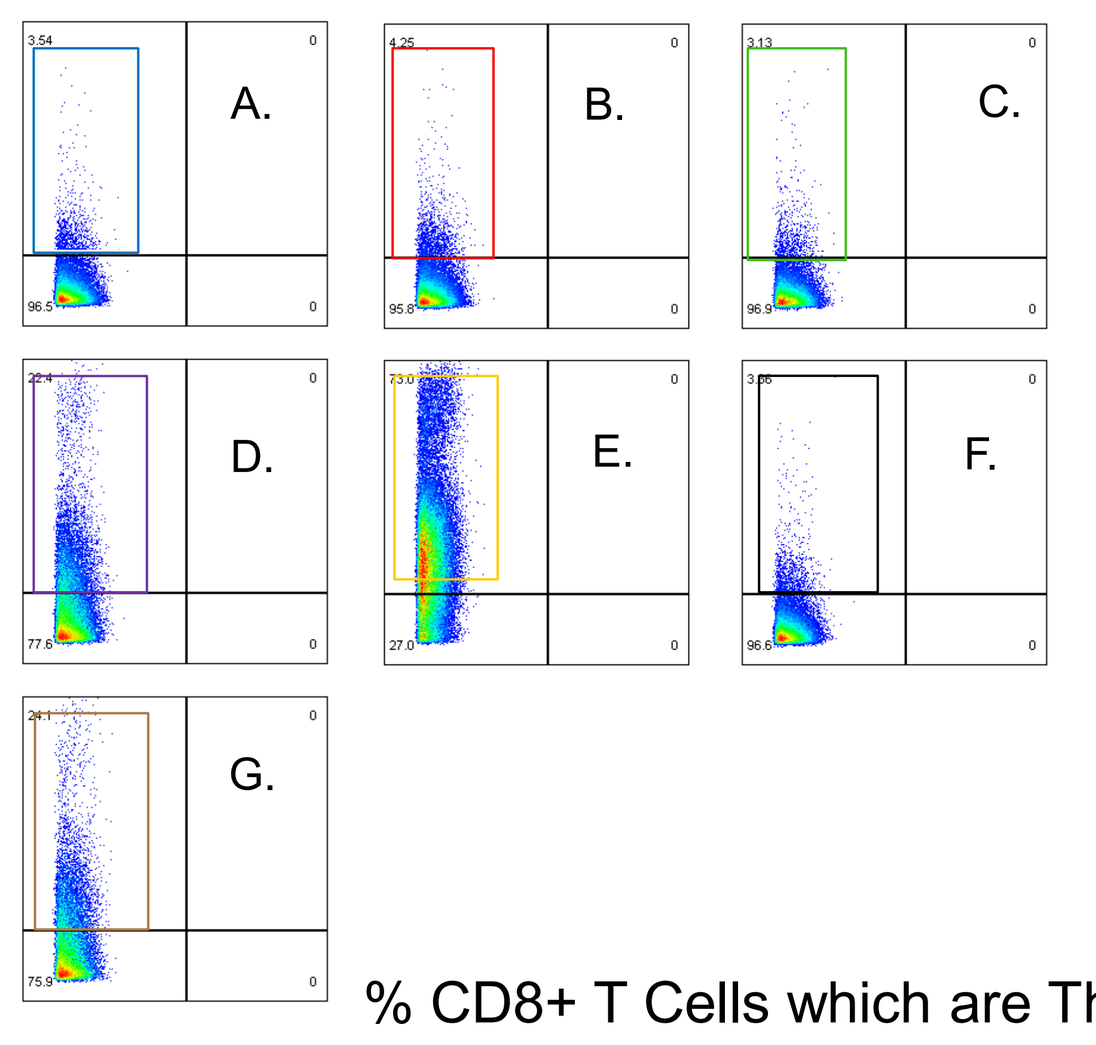

% CD8+ T Cells which are Thy1.1+ve (CD8+ CAR T CELLS):

A. PBS/PBS: 3.54%
B. CAR/PBS: 4.25%
C. Reo/PBS: 3.13%
D. CAR(Reo)/PBS: 22.4%
E. CAR(Reo)/Reo Boost: 73.0%
F. Reo/Reo: 3.36%
G. CAR(Reo)/Heterologous Boost: 24.1%

C.

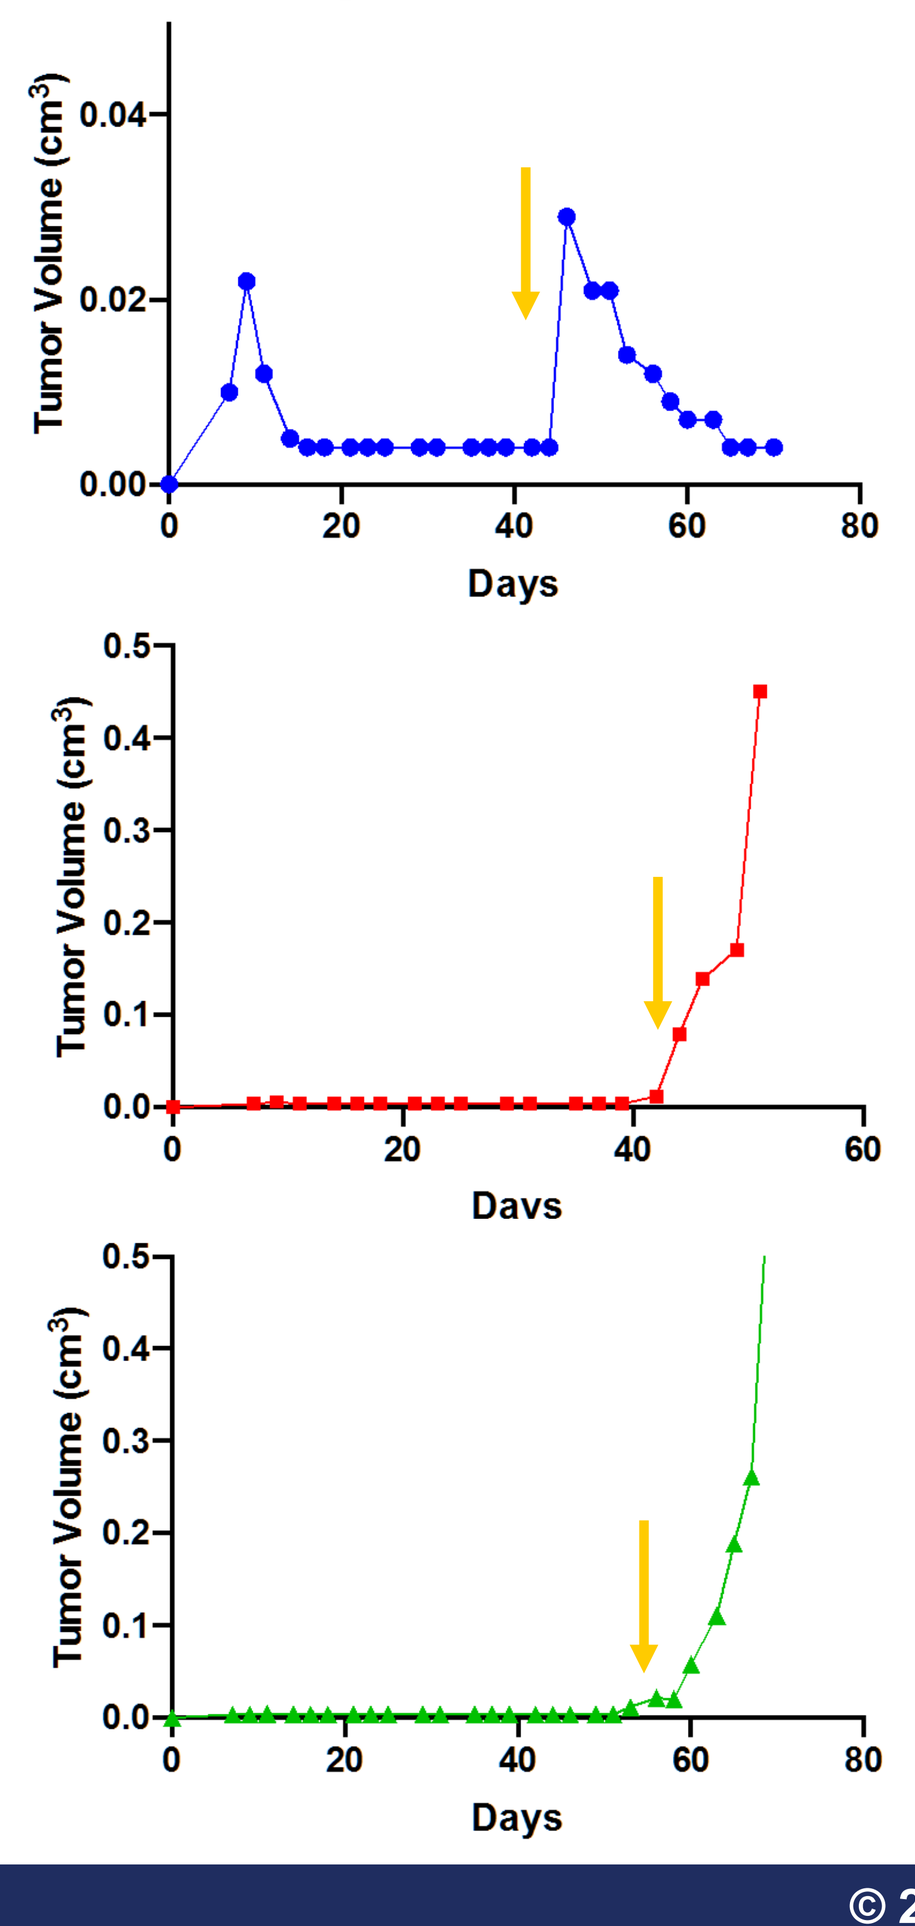
Tumor Volume (cm³)
Days

Figure 5. CAR T can carry oncolytic virus to tumors and generate improved therapy compared to either alone. A. C57Bl/6 mice were seeded with sc B16-EGFRvIII tumors. 8d later, mice were treated iv with (A) PBS, (B) 10^7 anti-EGFRvIII CAR T cells, (C,F) 10^7pfu Reovirus, or (D,E,G) 10^7 CAR T cells pre-loaded with Reovirus at a MOI of 1. On day 15, mice were treated with a second iv injection of PBS, 10^7 pfu Reovirus or a heterologous virus VSV-GFP. Tumor size and mouse survival was followed with time as shown. **B.** Splenocytes from mice treated as in **A.** above were harvested from mice at euthanasia and analyzed by flow cytometry for CD8+/Thy1.1 CAR+ve T cells. **C.** C57Bl/6 mice were seeded with sc B16-EGFRvIII tumors. 8d later, mice were treated iv with a lower dose of 10^6 anti-EGFRvIII CAR T cells pre-loaded with Reovirus at a MOI of 1. On day 15, mice were treated with a second iv injection of 10^7 pfu Reovirus. In three mice tumors recurred at around day 40-50. When recurrent tumors started to grow (>0.2cm in diameter) they were administered a further i.v. injection of 10^7 pfu Reovirus (top), PBS (Middle) or VSV-GFP bottom (arrows). Tumor size was followed with time as shown

Conclusions: Overall, these data show that:

In the absence of host conditioning (TBI), Reovirus loaded CAR T cells are significantly more therapeutic than unloaded CAR T cells in this model;

Reovirus-loaded CAR T cells are highly therapeutic (tumor cures) when boosted in vivo with Reovirus but not with a heterologous virus (VSV-GFP).

CAR T are very poorly persistent, with very poor therapy;

CAR T loaded with Reovirus are significantly more persistent, associated with significantly improved therapy;

CAR T cells loaded with Reovirus and then boosted in vivo with Reovirus generates a highly persistent population of CAR T which is associated with excellent therapy including tumor cures in the solid tumor setting.

Tumors which recur can be treated with a further boost with homologous, but not heterologous, virus

For questions or more information, please contact vile.richard@mayo.edu

This work was supported by a Fraternal Order of Eagles Cancer Fellowship, the Mayo Foundation, and Oncolytics Biotech Incorporated.

© 2021 Mayo Foundation for Medical Education and Research