Treatment with pembrolizumab in combination with the oncolytic virus pelareorep promotes anti-tumor immunity in patients with advanced pancreatic adenocarcinoma

Devalingam Mahalingam[1], Siqi Chen[2], Ping Xie[2], Aparna Kalyan[1], Sheetal Mehta Kircher[3], Masha Kocherginsky[2], Jiahui Xu[1], Victoria Maurer[3], Pedro Antonio Hermida de Viveiros[3], Elena Vagia[1], Grey A Wilkinson[4], Matthew C. Coffey[4], Mary Frances Mulcahy[3], Al Bowen Benson III[1], Bin Zhang[2]

[1]Northwestern University, Chicago, IL; [2]Northwestern University Feinberg School of Medicine, Chicago, IL; [3]Robert H. Lurie Comprehensive Cancer Center of Northwestern University, Chicago, IL; [4]Oncolytics Biotech Inc, Calgary, AB

Abstract #343107

Abstract

Background: Pelareorep is an intravenously delivered oncolytic reovirus that can induce a T cell-inflamed phenotype in pancreatic ductal adenocarcinoma (PDAC). In prior studies, tumor tissue analysis from patients treated with pelareorep shows pelareorep replication, increased T cell infiltration, and upregulation of PD-L1. We hypothesized that pelareorep in combination with pembrolizumab in patients with PDAC would lead to improved responses and anti-tumor immunological changes within peripheral blood and tumor biopsies in responding patients.

Methods: PDAC patients who progressed after first-line treatment received pelareorep at a dose of 4.5×10^{10} $TCID_{50}$ IV on days 1, 2, 3 & 8 of Cycle (C) 1, and days 1 & 8 with C2 onwards. Pembrolizumab was administered on day 1 of each 21-day cycle at 200 mg IV. The primary objective was overall response rate by RECIST v 1.1 criteria. Secondary objectives included evaluating immunological changes within tumor tissue and peripheral blood, performed by multi-plex immunohistochemistry and spectral flow cytometry (Cytek), respectively.

Results: Thirteen patients were enrolled. Disease control was achieved in 42% of the 12 efficacy-evaluable patients. One patient achieved a partial response (PR). Four additional patients achieved stable disease (SD). On-treatment tumor biopsies, collected during C1, showed pelareorep replication, increased infiltration of CD8+ T cells and PD-L1+ cells, and decreased expression of VDAC1, a mitochondrial gatekeeper for tumor promotion, relative to archival tissue. Reduced infiltration of Foxp3+ regulatory T cells (Treg) was observed in patients showing tumor response. Peripheral blood was collected at day 1 of each cycle and on C1 day 8. Relative to pretreatment samples, the number of CD8+ effector memory T cells and B cells tend to increase while the number of Treg cells declined in C2 onwards in patients with tumor response. Furthermore, these patients had increased expression of the mitochondrial protein TOMM20 in CD8+ T cells and decreased expression of PD-1 and the H3K27me3 epigenetic mark in Treg. Treatment was well tolerated with most treatment-related adverse events, including flu-like symptoms, being grade 1 or 2.

Conclusions: The combination of pelareorep and pembrolizumab showed a manageable safety profile and modest efficacy in an unselected PDAC population. Additional correlation analyses between treatment efficacy and immunological changes will be presented. The anti-tumor activity of pelareorep and checkpoint blockade therapy is being evaluated further in ongoing studies.

Background and Rationale

Figure 1. Mechanism of Action

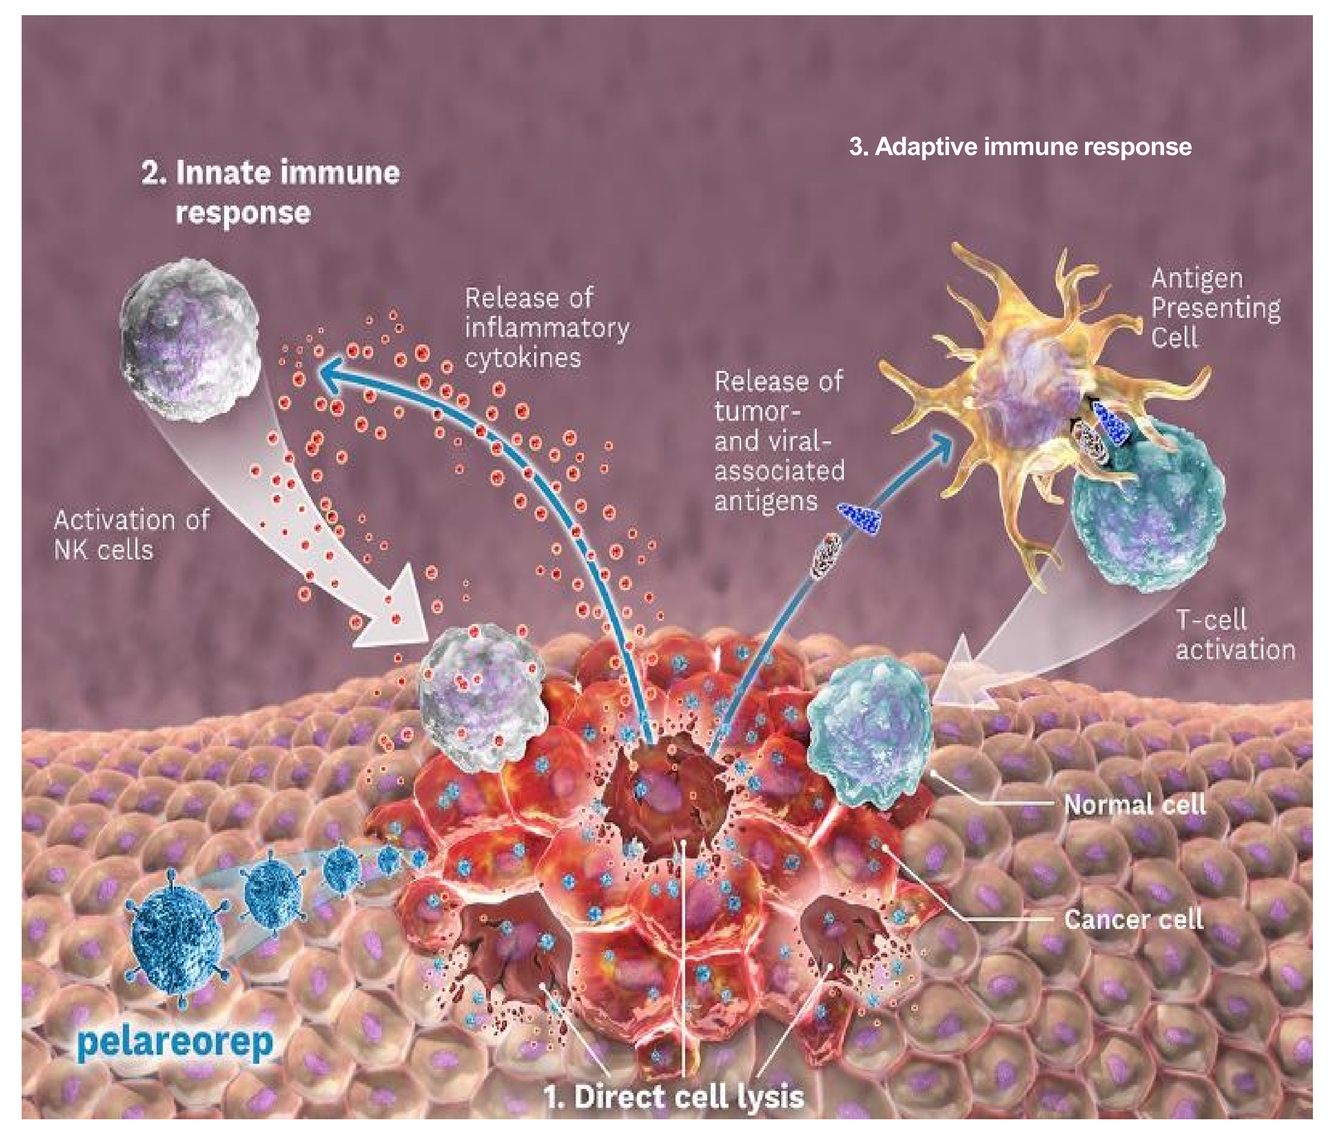
2. Innate immune response
3. Adaptive immune response
Release of inflammatory cytokines
Release of tumor- and viral-associated antigens
Antigen Presenting Cell
Activation of NK cells
T-cell activation
Normal cell
Cancer cell
pelareorep
1. Direct cell lysis

- In prior studies, pelareorep and pembrolizumab did not add significant toxicity to chemotherapy and showed encouraging efficacy in second line PDAC patients[1]
- Prior studies in first-line PDAC achieved encouraging 1 & 2 year-survival rates of 46% & 24%, respectively with chemotherapy and pelareorep treatment[2]
- On-treatment biopsies have shown that pelareorep induces an inflammatory phenotype in the tumor micro-environment that facilitates synergy with checkpoint blockade therapy (see pelareorep mechanism of action, Fig 1)[1,3,4]
- We hypothesized that pelareorep in combination with pembrolizumab in patients with PDAC would lead to improved responses and anti-tumor immunological changes within peripheral blood and tumor biopsies in responding patients.

Methods

- This phase 2 study (NCT03723915) enrolled PDAC patients who progressed after first line treatment. Patients received therapy until disease progression or unacceptable toxicity as follows: pelareorep ($4.5\times10^{10}TCID_{50}$ IV) plus pembrolizumab (200 mg IV)
- The primary objective was overall response rate by RECIST v 1.1 criteria. Secondary objectives included evaluating immunological changes within tumor tissue and peripheral blood, performed by multi-plex immunohistochemistry and spectral flow cytometry (Cytek), respectively.

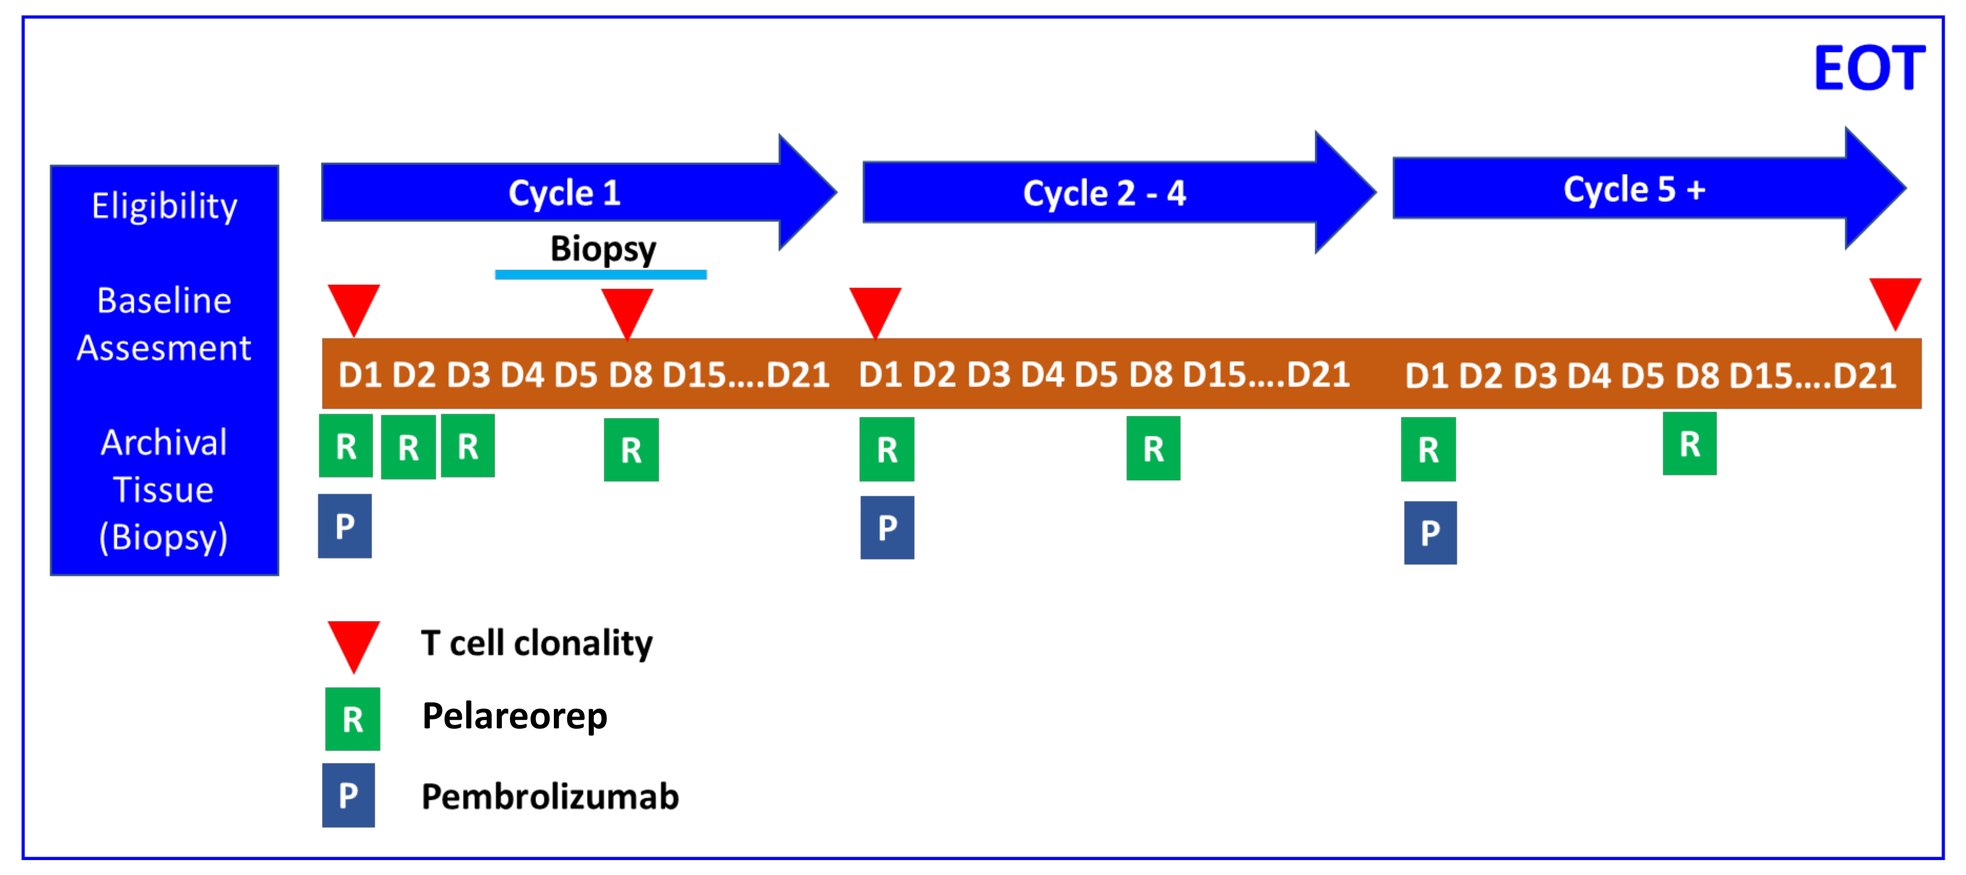
EOT
Eligibility
Baseline Assesment
Archival Tissue (Biopsy)
Cycle 1
Biopsy
Cycle 2 - 4
Cycle 5 +
D1 D2 D3 D4 D5 D8 D15....D21 D1 D2 D3 D4 D5 D8 D15....D21 D1 D2 D3 D4 D5 D8 D15....D21
T cell clonality
Pelareorep
Pembrolizumab

Figure 2. Study Schedule.

Results

Figure 3. Swimmer's plot of best response.

Thirteen patients were enrolled. Disease control was achieved in 42% of the 12 efficacy-evaluable patients. One patient achieved a partial response (PR). Four additional patients achieved stable disease (SD).

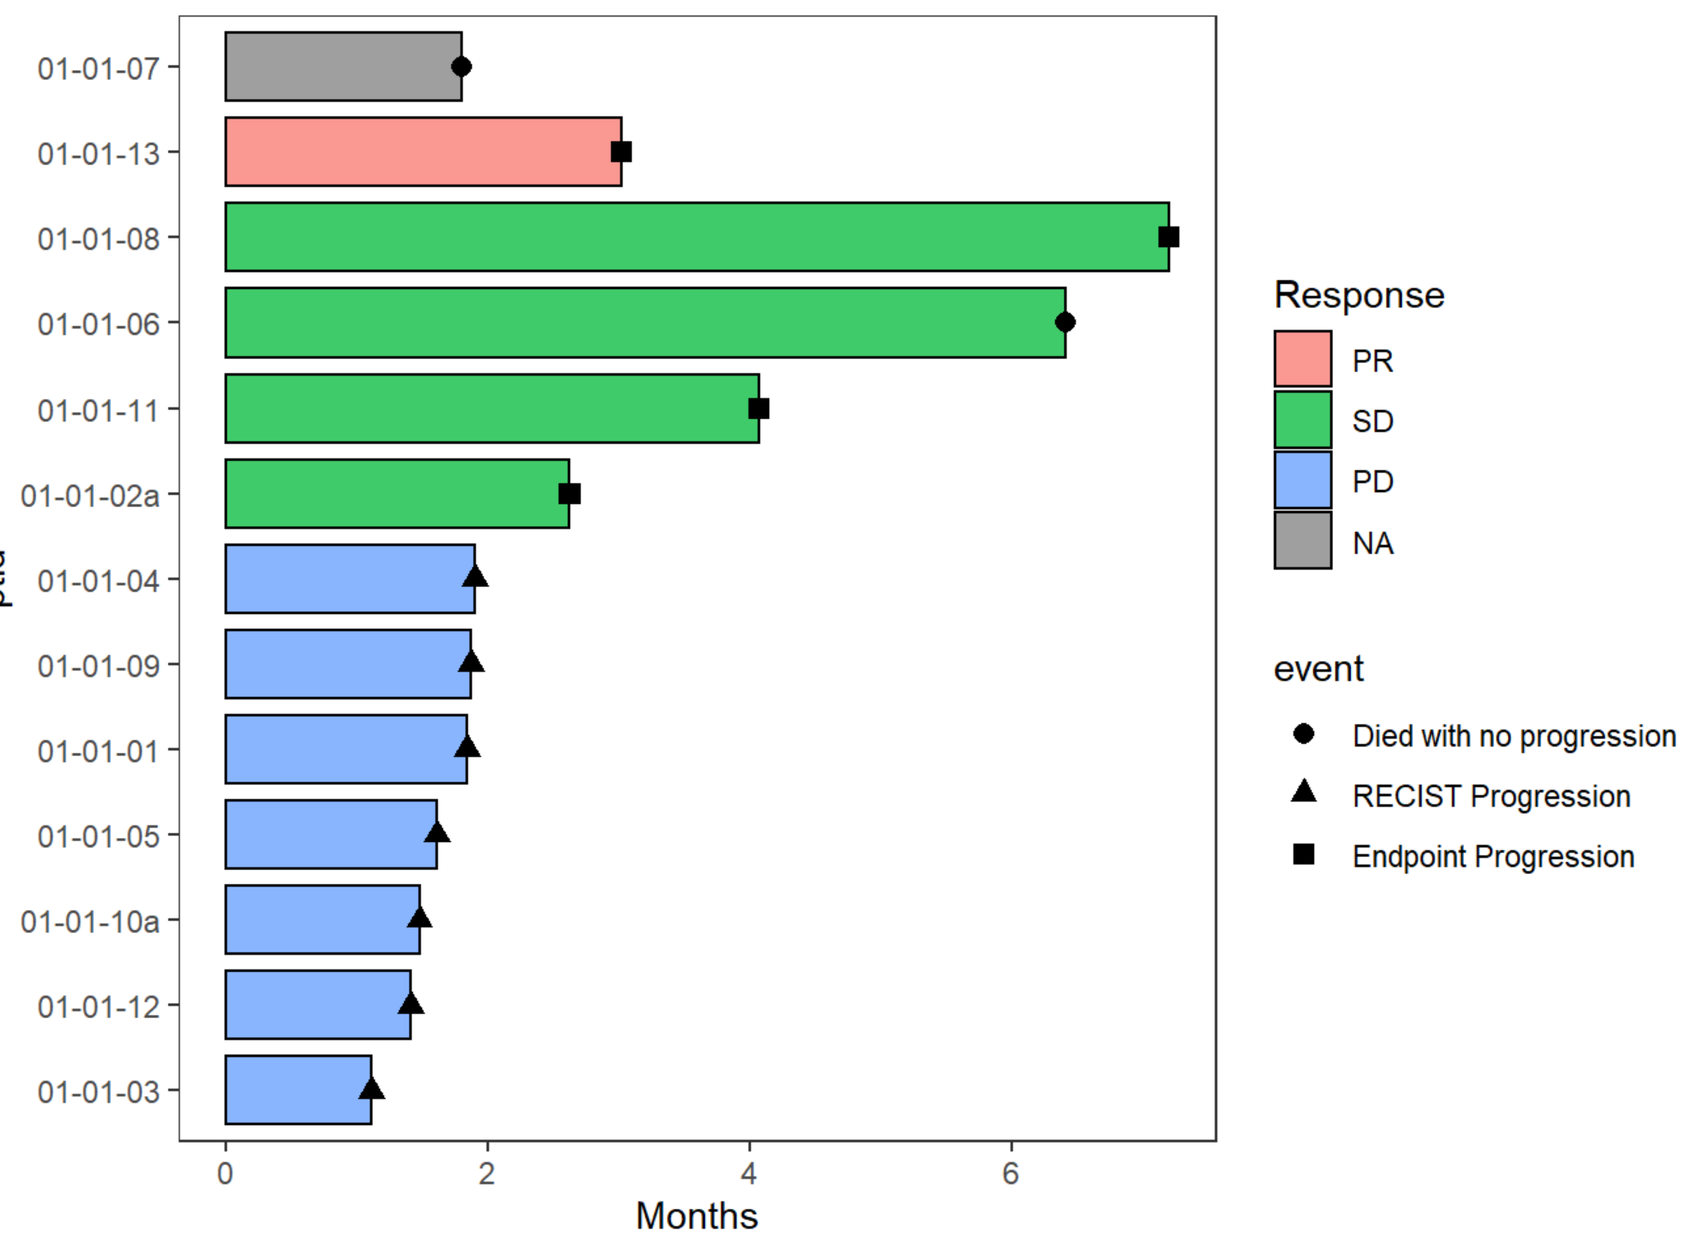
01-01-07
01-01-13
01-01-08
01-01-06
01-01-11
01-01-02a
01-01-04
01-01-09
01-01-01
01-01-05
01-01-10a
01-01-12
01-01-03
ptid
Months
Response
PR
SD
PD
NA
event
Died with no progression
RECIST Progression
Endpoint Progression

Results

Figure 4. (A) Progression Free Survival and (B) Overall Survival.

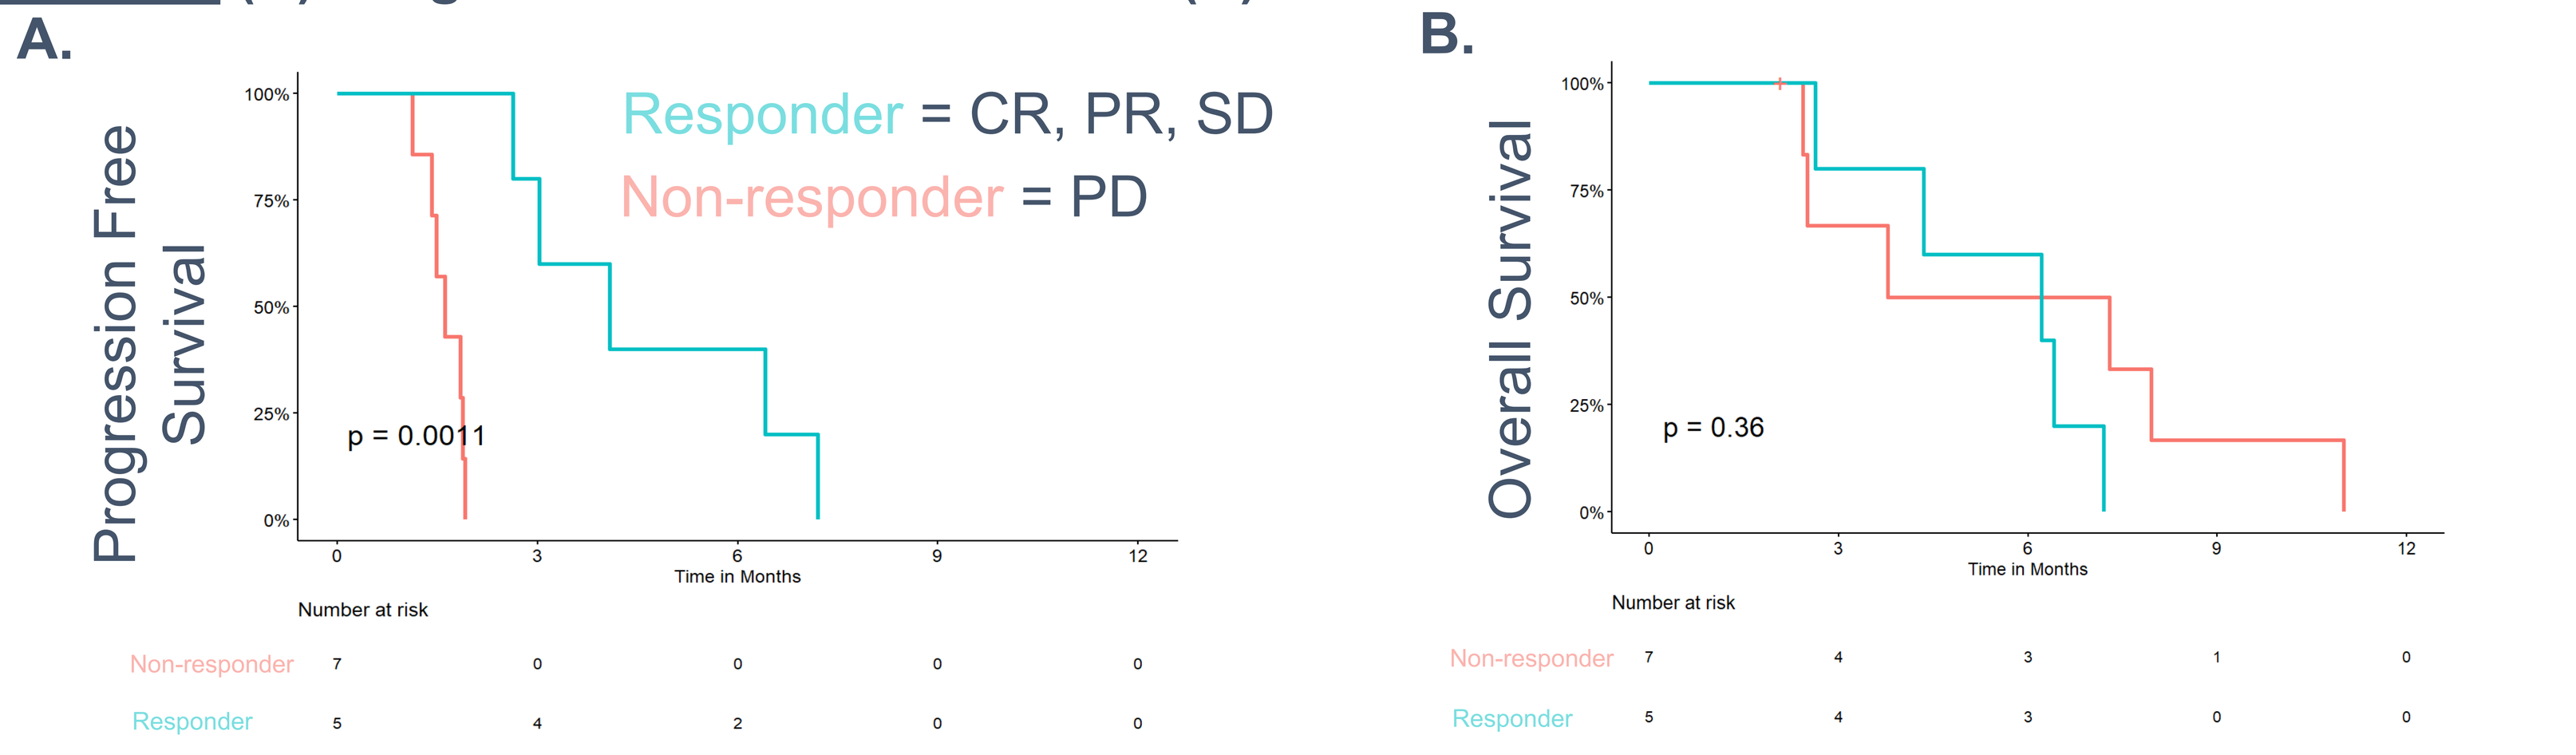
A.
Progression Free Survival
Responder = CR, PR, SD
Non-responder = PD
p = 0.0011
Time in Months
Number at risk
Non-responder 7 0 0 0 0
Responder 5 4 2 0 0
B.
Overall Survival
p = 0.36
Time in Months
Number at risk
Non-responder 7 4 3 1 0
Responder 5 4 3 0 0

	Median PFS (months)	95% LCL for median PFS	95% UCL for median PFS
Non-responders	1.6099	1.4141	1.9042
Responders	4.0753	3.0240	7.1964

	Median OS (months)	95% LCL for median OS	95% UCL for median OS
Non-responders	5.53593	2.4956	11.0061
Responders	6.2094	4.3381	7.1964

Table 1. Adverse events.

The safety and tolerability of pembrolizumab and pelareorep when administered in combination as determined by NCI CTCAE v 4.03 are evaluated by the frequencies and percentages of adverse events for all grades and separately for grades 1, 2, and 3 for the 13 evaluable patients. Data is presented in descending order of frequencies of adverse events > 10 percent for all grades.

AE	All Grades	Grade 1	Grade 2	Grade 3
Chills	11 (85%)	10 (77%)	1 (8%)	0 (0%)
Fever	10 (77%)	7 (54%)	2 (15%)	1 (8%)
Vomiting	9 (69%)	8 (62%)	0 (0%)	1 (8%)
Nausea	7 (54%)	3 (23%)	3 (23%)	1 (8%)
Fatigue	6 (46%)	4 (31%)	0 (0%)	2 (15%)
Diarrhea	5 (38%)	5 (38%)	0 (0%)	0 (0%)
Hyperglycemia	5 (38%)	0 (0%)	5 (38%)	0 (0%)
Anemia	4 (31%)	3 (23%)	0 (0%)	1 (8%)
Headache	4 (31%)	4 (31%)	0 (0%)	0 (0%)
White blood cell decreased	4 (31%)	2 (15%)	1 (8%)	1 (8%)
Abdominal pain	3 (23%)	2 (15%)	1 (8%)	0 (0%)
Alkaline phosphatase increased	3 (23%)	1 (8%)	1 (8%)	1 (8%)
Anorexia	3 (23%)	3 (23%)	0 (0%)	0 (0%)
Aspartate aminotransferase increased	3 (23%)	3 (23%)	0 (0%)	0 (0%)
Blood bilirubin increased	3 (23%)	3 (23%)	0 (0%)	0 (0%)
Lymphocyte count decreased	3 (23%)	0 (0%)	1 (8%)	2 (15%)
Neutrophil count decreased	3 (23%)	0 (0%)	2 (15%)	1 (8%)
Platelet count decreased	3 (23%)	2 (15%)	1 (8%)	0 (0%)
Proteinuria	3 (23%)	2 (15%)	1 (8%)	0 (0%)
Hyponatremia	2 (15%)	0 (0%)	0 (0%)	2 (15%)
Hypothyroidism	2 (15%)	2 (15%)	0 (0%)	0 (0%)
Musculoskeletal and connective tissue disorder - Other, specify	2 (15%)	2 (15%)	0 (0%)	0 (0%)
Myalgia	2 (15%)	2 (15%)	0 (0%)	0 (0%)
Nasal congestion	2 (15%)	2 (15%)	0 (0%)	0 (0%)

Figure 5. **Identification of differences in immune cell populations between responders (R) and non-responders (NR) using spectral flow cytometry (Cytek).**
(A) Exemplified tSNE visualization of merged events from PBMCs of NR (n = 6) and R (n = 4), before (C1D1) and after therapy (C2D1). **(B)** The summarized immune cell composition of PBMCs of NR and R, before (C1D1) and after therapy (C2D1). There was a higher frequency of myeloid cells or B cells and a lower frequency of NKT cells or CD4+ effector T cells at C2D1 than at C1D1 in both NR and R. The frequency of CD4+ Treg cells was significantly lower in R after therapy but not in NR.

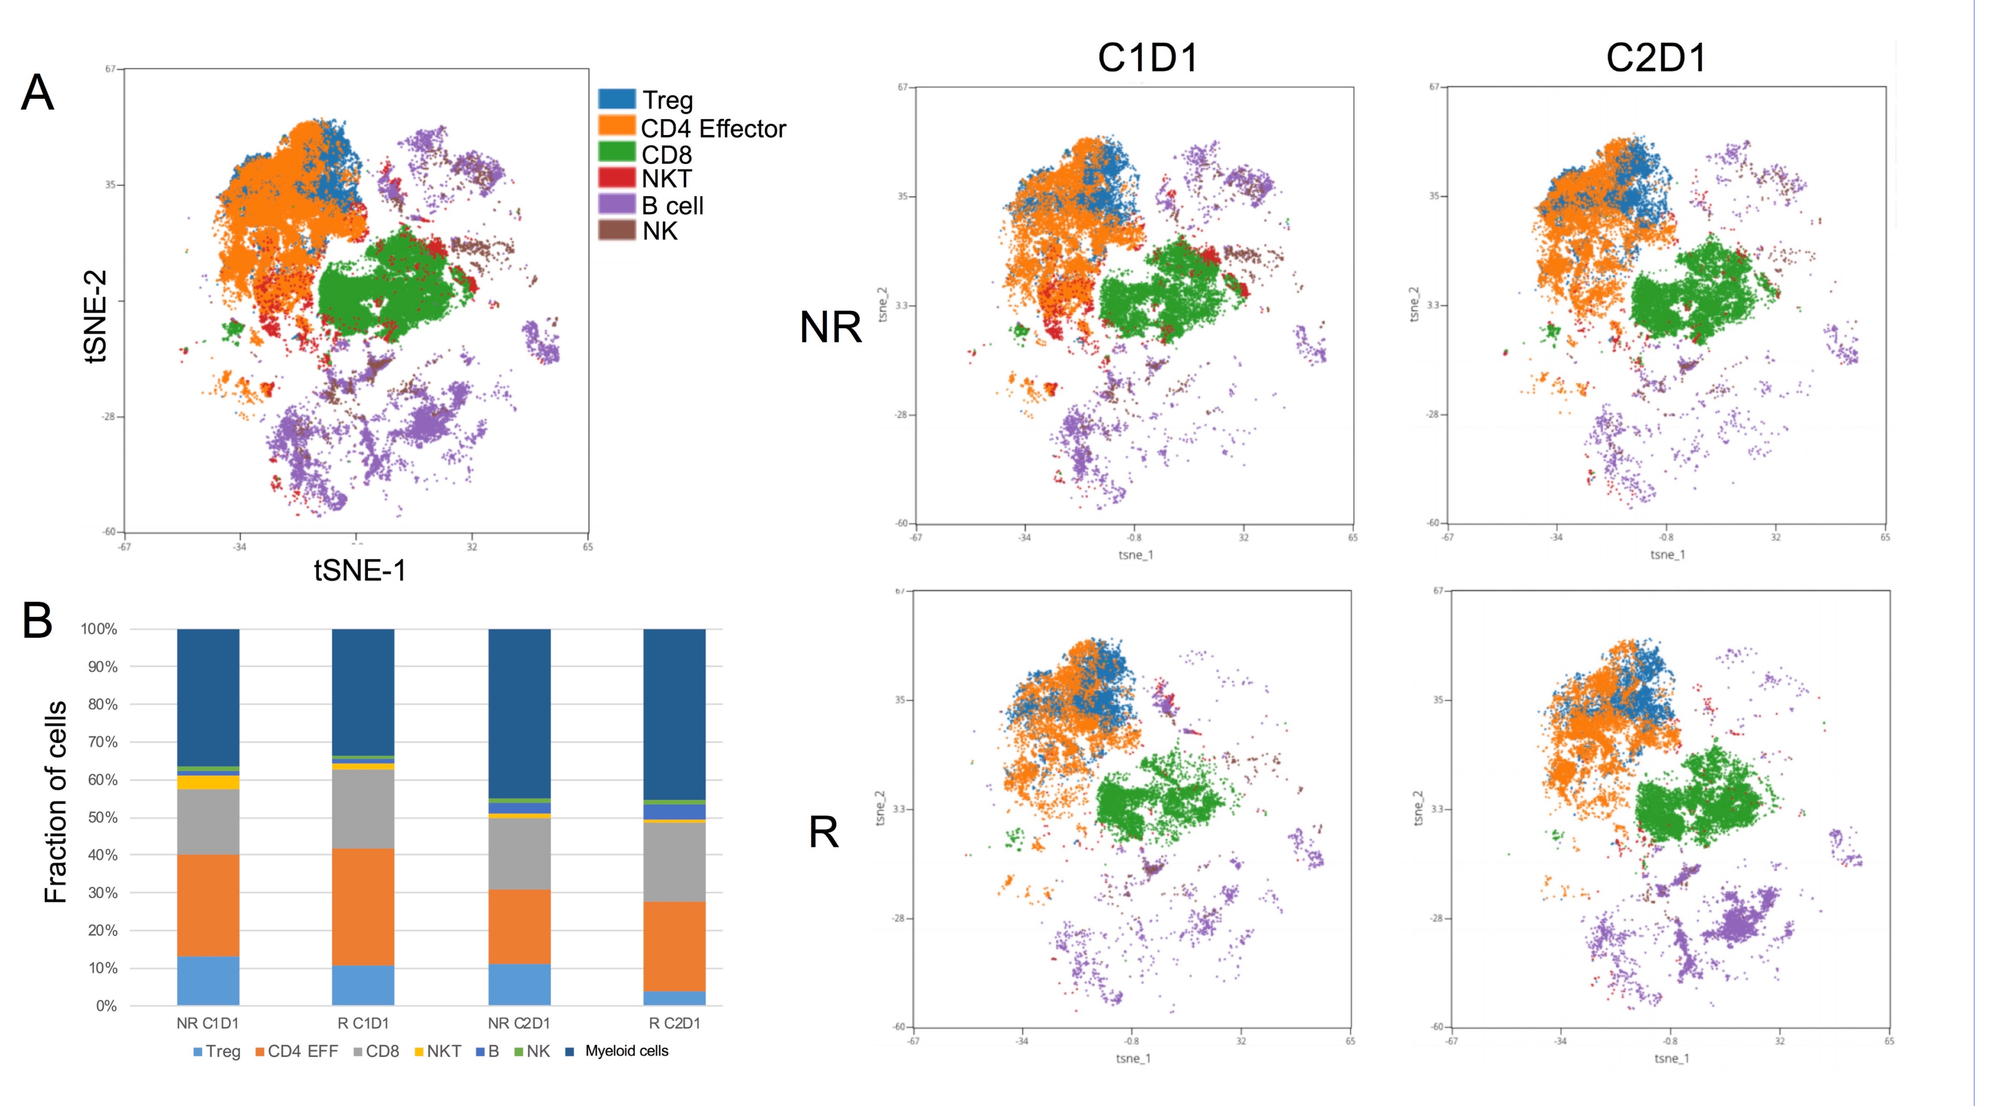
A
Treg
CD4 Effector
CD8
NKT
B cell
NK
tSNE-2
tSNE-1
C1D1
C2D1
NR
R
B
Fraction of cells
NR C1D1
R C1D1
NR C2D1
R C2D1
Treg
CD4 EFF
CD8
NKT
B
NK
Myeloid cells

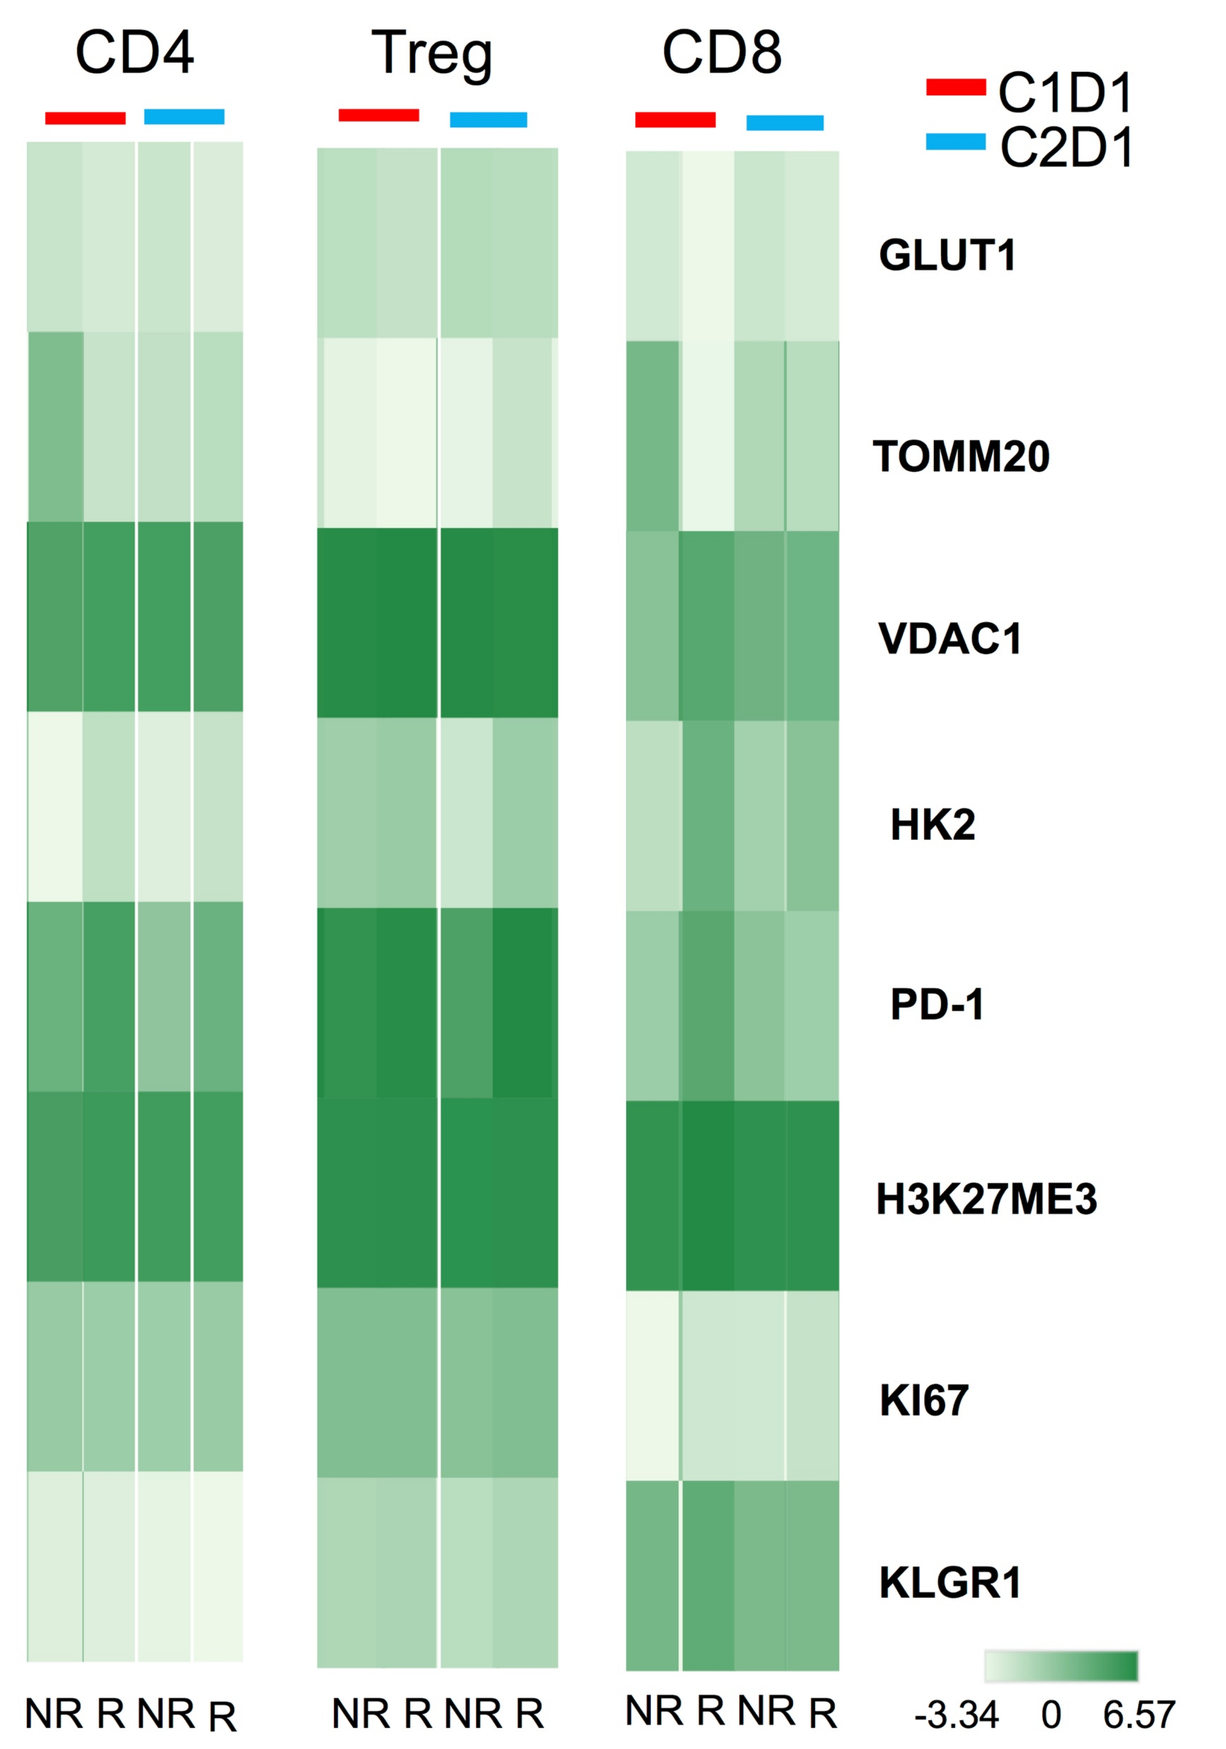
CD4
Treg
CD8
C1D1
C2D1
GLUT1
TOMM20
VDAC1
HK2
PD-1
H3K27ME3
KI67
KLGR1
NR R NR R
NR R NR R
NR R NR R
-3.34 0 6.57

Figure 6. **Improved mitochondria function in CD8+ T cells after the initiation of immunotherapy in R.** The heat map represents the median expression for metabolic markers GLUT1, TOMM20, VDAC1, and HK2, the epigenetic mark H3K27me3, the proliferation marker Ki67 as well as the exhausting markers PD-1 and KLRG1 within CD4+ effector T cells, Treg cells and CD8+ T cells from NR (n = 6) and R (n = 4), before (C1D1) and after therapy (C2D1). Global differential marker expression on merged samples in CD8+ T cells in R showed an upregulation of the mitochondrial protein import marker TOMM20, and the glucose transporter GLUT1, as well as downregulation of the mitochondrial cell death markers VDAC1 and HK2 with a concomitant reduction in expression levels of the exhausting markers PD-1 and KLRG1. Additionally, the CD4+ effector T cell and CD8+ T cell subpopulations of NR had a lower expression levels of TOMM20 after treatment initiation compared to those before therapy.

Results

Figure 7. **Increased activation in CD8+ T cells after the initiation of immunotherapy in R.** The heat map represents the median expression for activation markers within CD4+ T cells **(A)** and CD8+ T cells **(B)** from NR (n = 6) and R (n = 4), before (C1D1) and after therapy (C2D1). Global differential marker expression on merged samples in CD8+ T cells in R showed an upregulation of the activation markers HLA-DR, CD69 and CXCR3 after therapy. We further identified naive, central memory (CM), effector memory (EM) and terminally differentiated effector memory (TEMRA) T cell subsets on the basis of their expression of CCR7 and CD45RA within CD4+ T cells **(C)** and CD8+ T cells **(D)**. The patients who eventually responded to therapy showed a significantly lower frequency of circulating CD4+ TEMRA T cells after therapy.

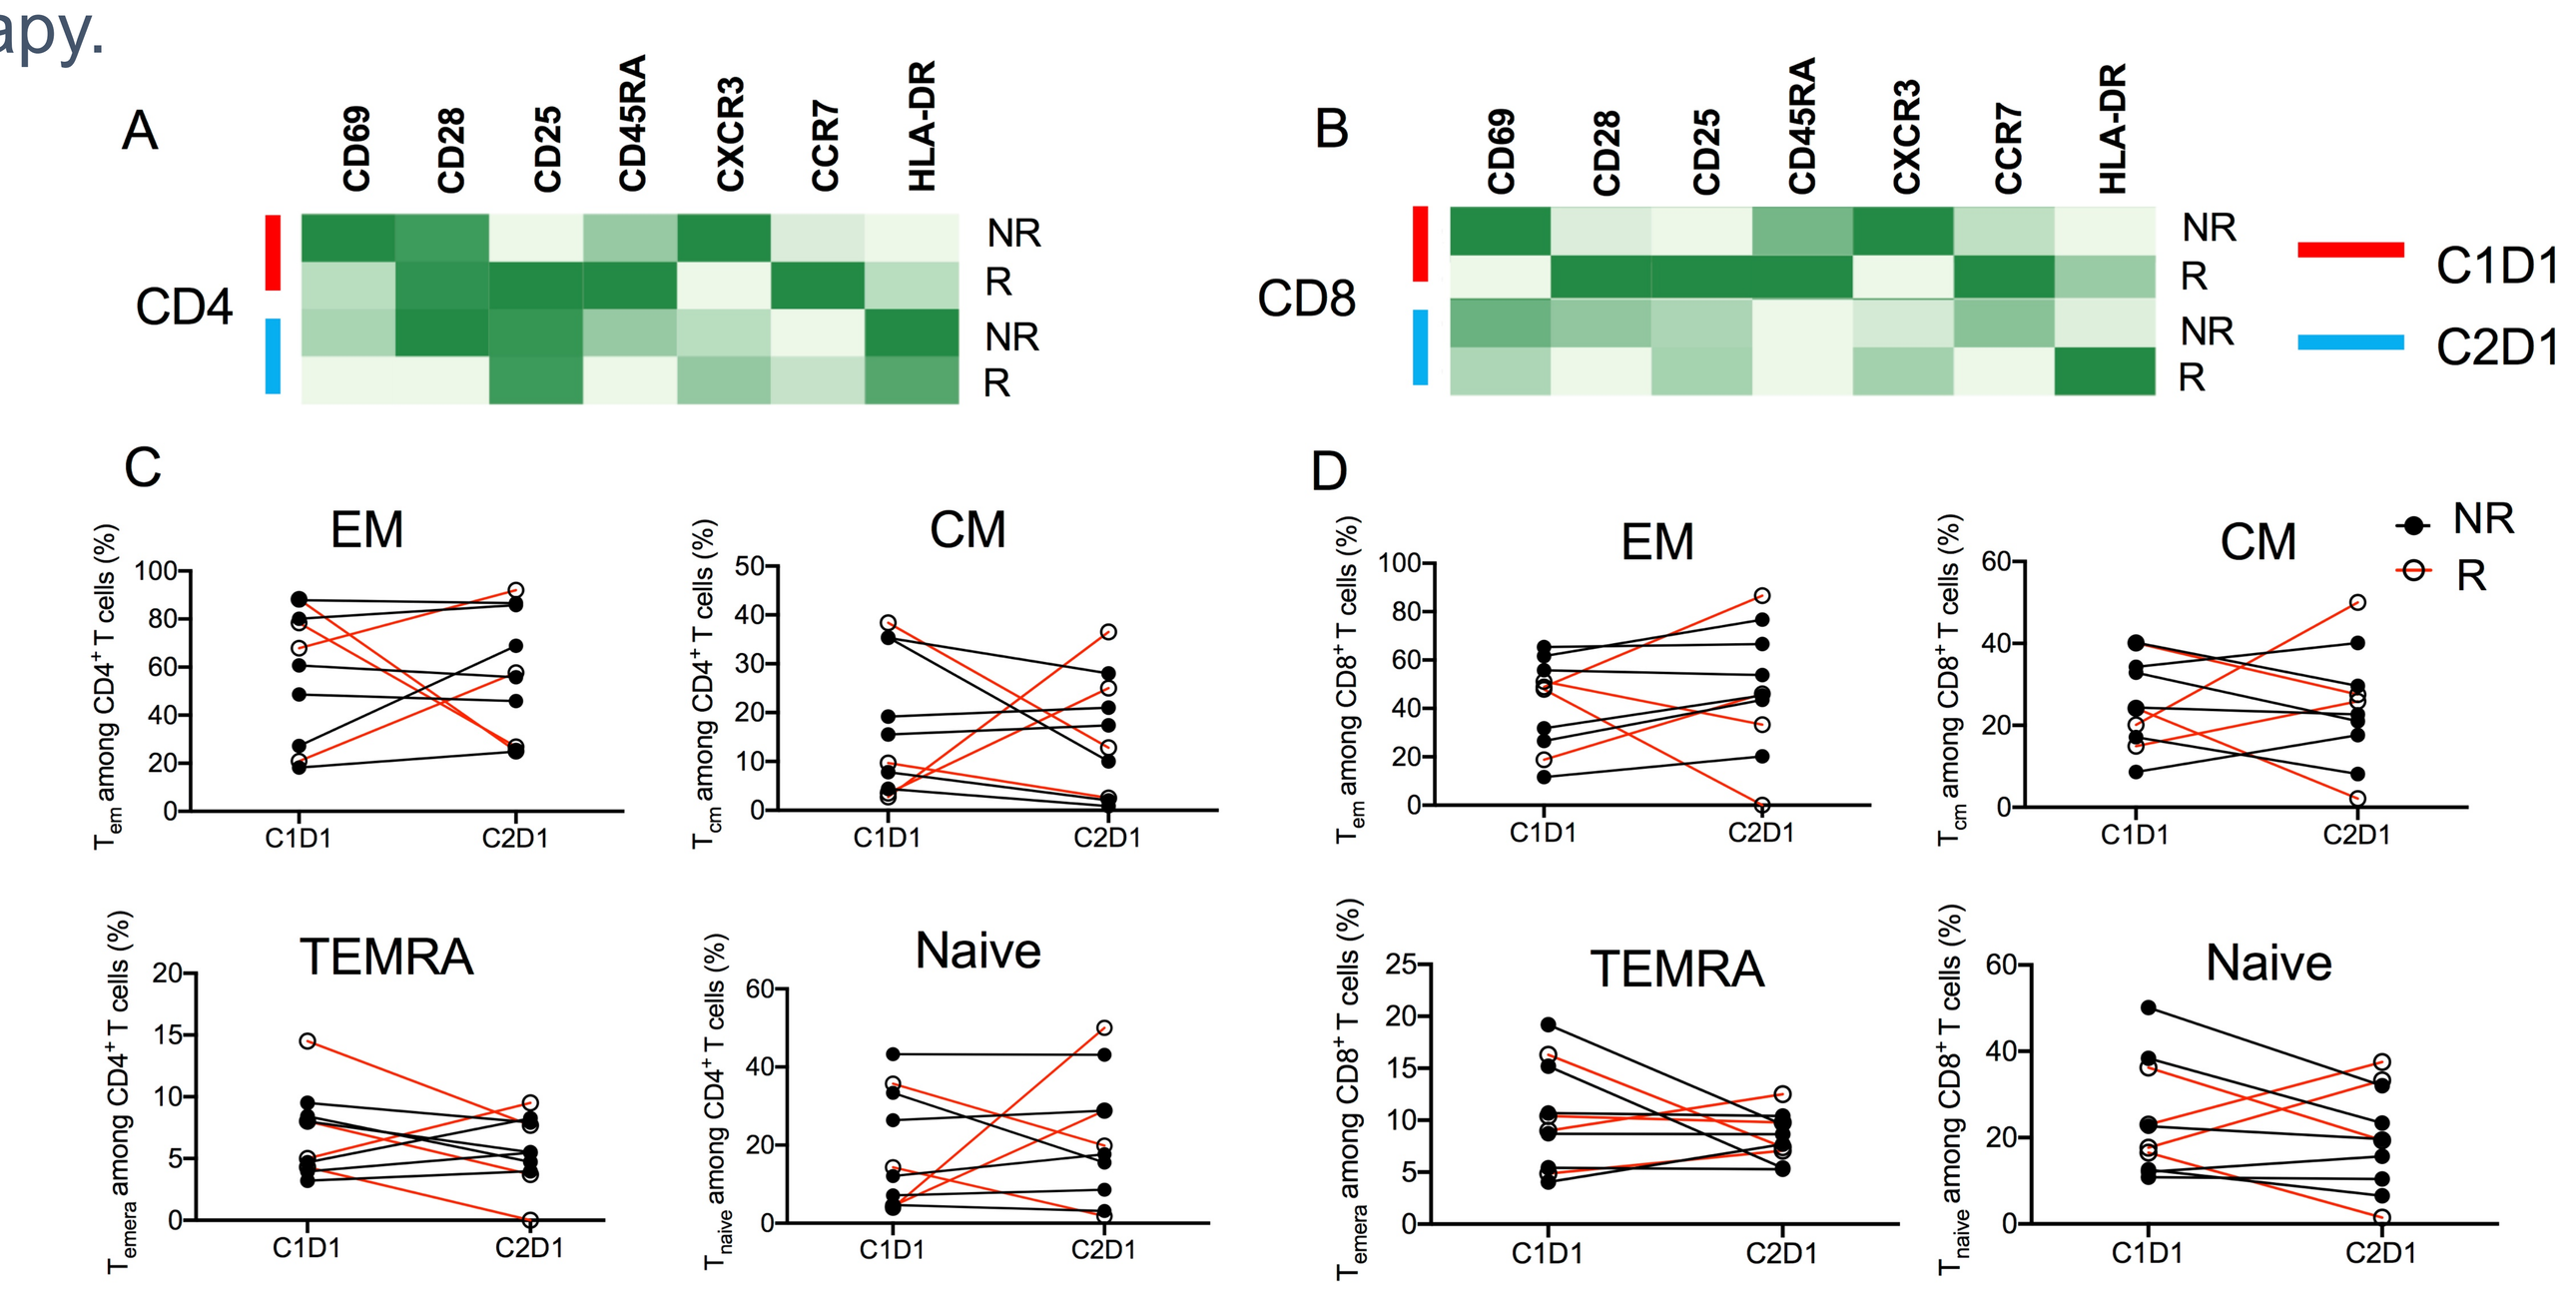
A
CD69
CD28
CD25
CD45RA
CXCR3
CCR7
HLA-DR
CD4
NR
R
NR
R
B
CD8
C1D1
C2D1
C
D
EM
CM
TEMRA
Naive
NR
R
C1D1
C2D1

Figure 8. **Distinct distribution of tumor-infiltrating leukocyte (TIL) populations and metabolic alteration across NR and R.** **(A)** Representative multi-plex immunohistochemical staining of reoviral capsid protein, the mitochondrial cell death marker VDAC1, and different immune markers as indicated for detection of TIL infiltration with the nuclear stain DAPI on FFPE biopsy specimen of R. **(B)** Overall density of selected cellular populations in a pair of SD biopsies and a pair of PD biopsies between screen and after therapy. An increase of the overall CD8+ T cell density or PD-L1+ cell density was observed in both PD and SD biopsies after therapy. There was a reduction of Treg density in SD but not PD biopsy after therapy. **(C)** Relative mean intensity of single marker PD-L1 or VDAC1 in each PD-L1+ or VDAC1+ cell, PD-L1 in Capsid+ tumor cells, and VDAC1 in different immune cell subsets as indicated. There was a greater reduction in cellular expression levels of VDAC1 in SD biopsy than that in PD biopsy after therapy. **(D)** Relative cellular counts of CD8+, Foxp3+, CD56+ NK cells touching other cells. There were more cellular counts of CD8+ T cells touching Capsid+ tumor cells and PD-L1+ cells, and Foxp3+ cells touching CD8+ T cells in PD biopsies after therapy than those in SD biopsies after therapy.

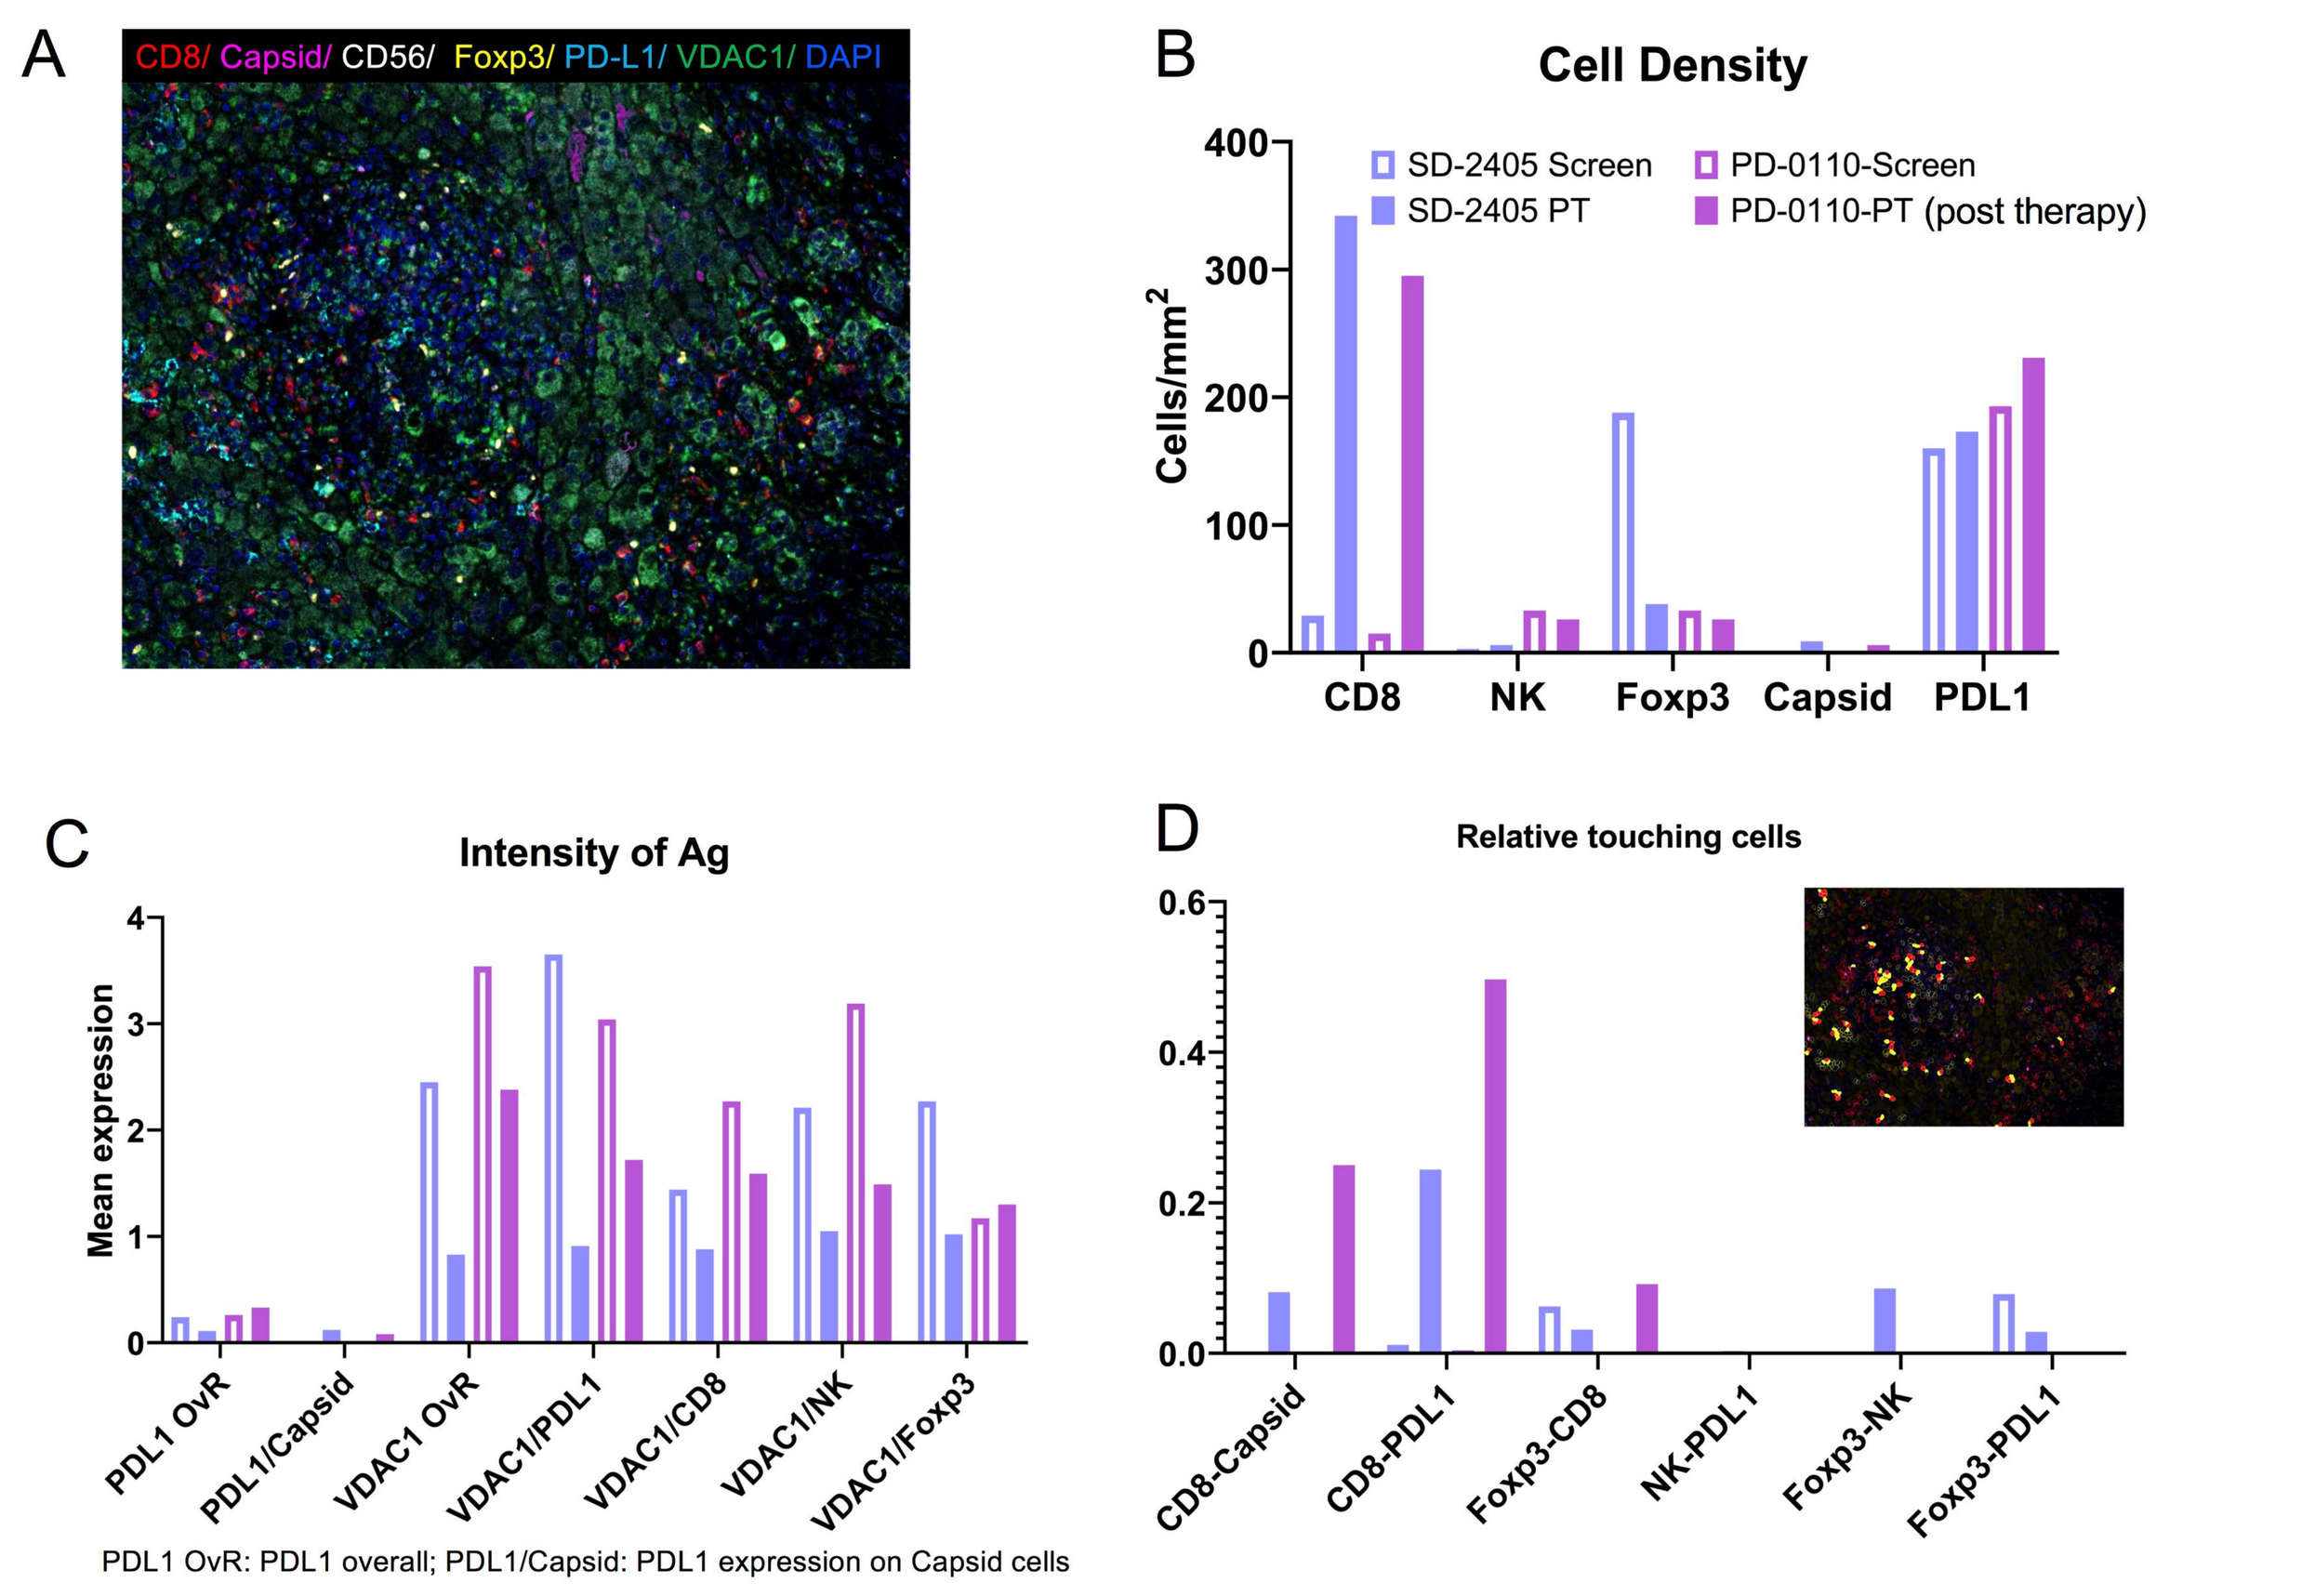
A
CD8/ Capsid/ CD56/ Foxp3/ PD-L1/ VDAC1/ DAPI
B
Cell Density
SD-2405 Screen
SD-2405 PT
PD-0110-Screen
PD-0110-PT (post therapy)
Cells/mm2
CD8
NK
Foxp3
Capsid
PDL1
C
Intensity of Ag
Mean expression
PDL1 OvR
PDL1/Capsid
VDAC1 OvR
VDAC1/PDL1
VDAC1/CD8
VDAC1/NK
VDAC1/Foxp3
PDL1 OvR: PDL1 overall; PDL1/Capsid: PDL1 expression on Capsid cells
D
Relative touching cells
CD8-Capsid
CD8-PDL1
Foxp3-CD8
NK-PDL1
Foxp3-NK
Foxp3-PDL1

Conclusion

- The combination of pelareorep and pembrolizumab demonstrated a manageable safety profile consistent with prior studies.[1]
- The efficacy results, both disease control and overall survival, are also consistent with prior studies in second-line PDAC patients despite the absence of chemotherapy in this study.[1]
- Responding patients show synergy between pelareorep and checkpoint blockade therapy as demonstrated by reductions in regulatory T cells and improved CD8+ effector T cell activation and metabolic function after the start of therapy in both PBMCs and TILs.
- Future research will examine the role of metabolic modulation added to pelareorep and checkpoint blockade therapy.

References

(1) Mahalingam, D., et al. Clin Cancer Res, 2020. 26(1): p. 71-81. **(2)** Mahalingam, D., et al. Cancers (Basel), 2018. 10(6).
(3) Samson, A., et al. Sci Transl Med, 2018. 10(422). **(4)** Manso et al. AACR Annual Meeting, April 9-14, 2021. Abstract number: CT191