Reovirus infection of prostate cancer induces upregulation of the negative regulators PD-L1 and BTLA

Nicola E Annels[1], Guy Simpson[1], Mehreen Arif[1], Mick Denyer[1], Kevin Harrington[2], Richard Vile[3], Matt Coffey[4], Alan Melcher[2], Hardev Pandha[1]

[1]Oncology, Department of Clinical and Experimental Medicine, University of Surrey, [2]Department of Radiotherapy and Imaging, Institute of Cancer Research, London, UK, [3]Department of Molecular Medicine, Mayo Clinic, Rochester, USA, [4]Oncolytics Biotech Incorporated, Calgary, Alberta, Canada.

Background:

Prostate Cancers (PCa) are generally considered to be immunologically 'cold' tumours, and this non-inflamed phenotype thought to be largely responsible for the disappointing lack of sensitivity of PCa patients to immune checkpoint blockade (ICB) therapy. However, the use of oncolytic viruses can overcome pre-existing mechanisms of resistance to ICB in PCas by transforming these cold tumours into 'hot', immune cell infiltrated, tumours. Such biological therapy can be further enhanced with the use of relevant immune checkpoint blockade that can overcome any constitutive or compensatory inhibitory resistance mechanisms. In this study, we investigated whether the effectiveness of oncolytic viral therapy for PCa could be improved with targeted blockade of PD-1 and/or CD73.

Methods:

The susceptibility of PCa cell lines to reovirus infection was tested *in vitro* using MTS assays. The immunogenic cell death profile of reovirus-infected TRAMP-C2 cells was determined by analysing the cell-surface expressed ICD determinants, calreticulin and HSP70 by FACS, and the secreted determinants by ELISA (HMGB1) or ATP assay. The capacity of reovirus to target TRAMP-C2 tumours *in vivo* were evaluated using immunocompetent C57BL/6. Anti-PD-1 and/or anti-CD73 blockade in the TRAMP-C2 PCa murine model was tested as a monotherapy and in combination with reovirus infection. Nanostring's PanCancer Immune Profiling RNA Panel was used to investigate the impact of reovirus therapy on the tumour microenvironment.

Results:

Figure1: *In vitro* susceptibility to Reovirus infection of a panel of PCa cell lines.

Cell monolayers of human PCa cell lines (PC3 and DU145), a human prostatic stromal myofibroblast cell line WPMY-1 and the transgenic adenocarcinoma mouse prostate cell line TRAMP-C2 were infected with doubling dilutions of a stock preparation of Reovirus (3x109pfu/ml). Following incubation at 37°C for 72h, cell survival was determined by MTS assay. Data is presented as the average ± SD (n=2).

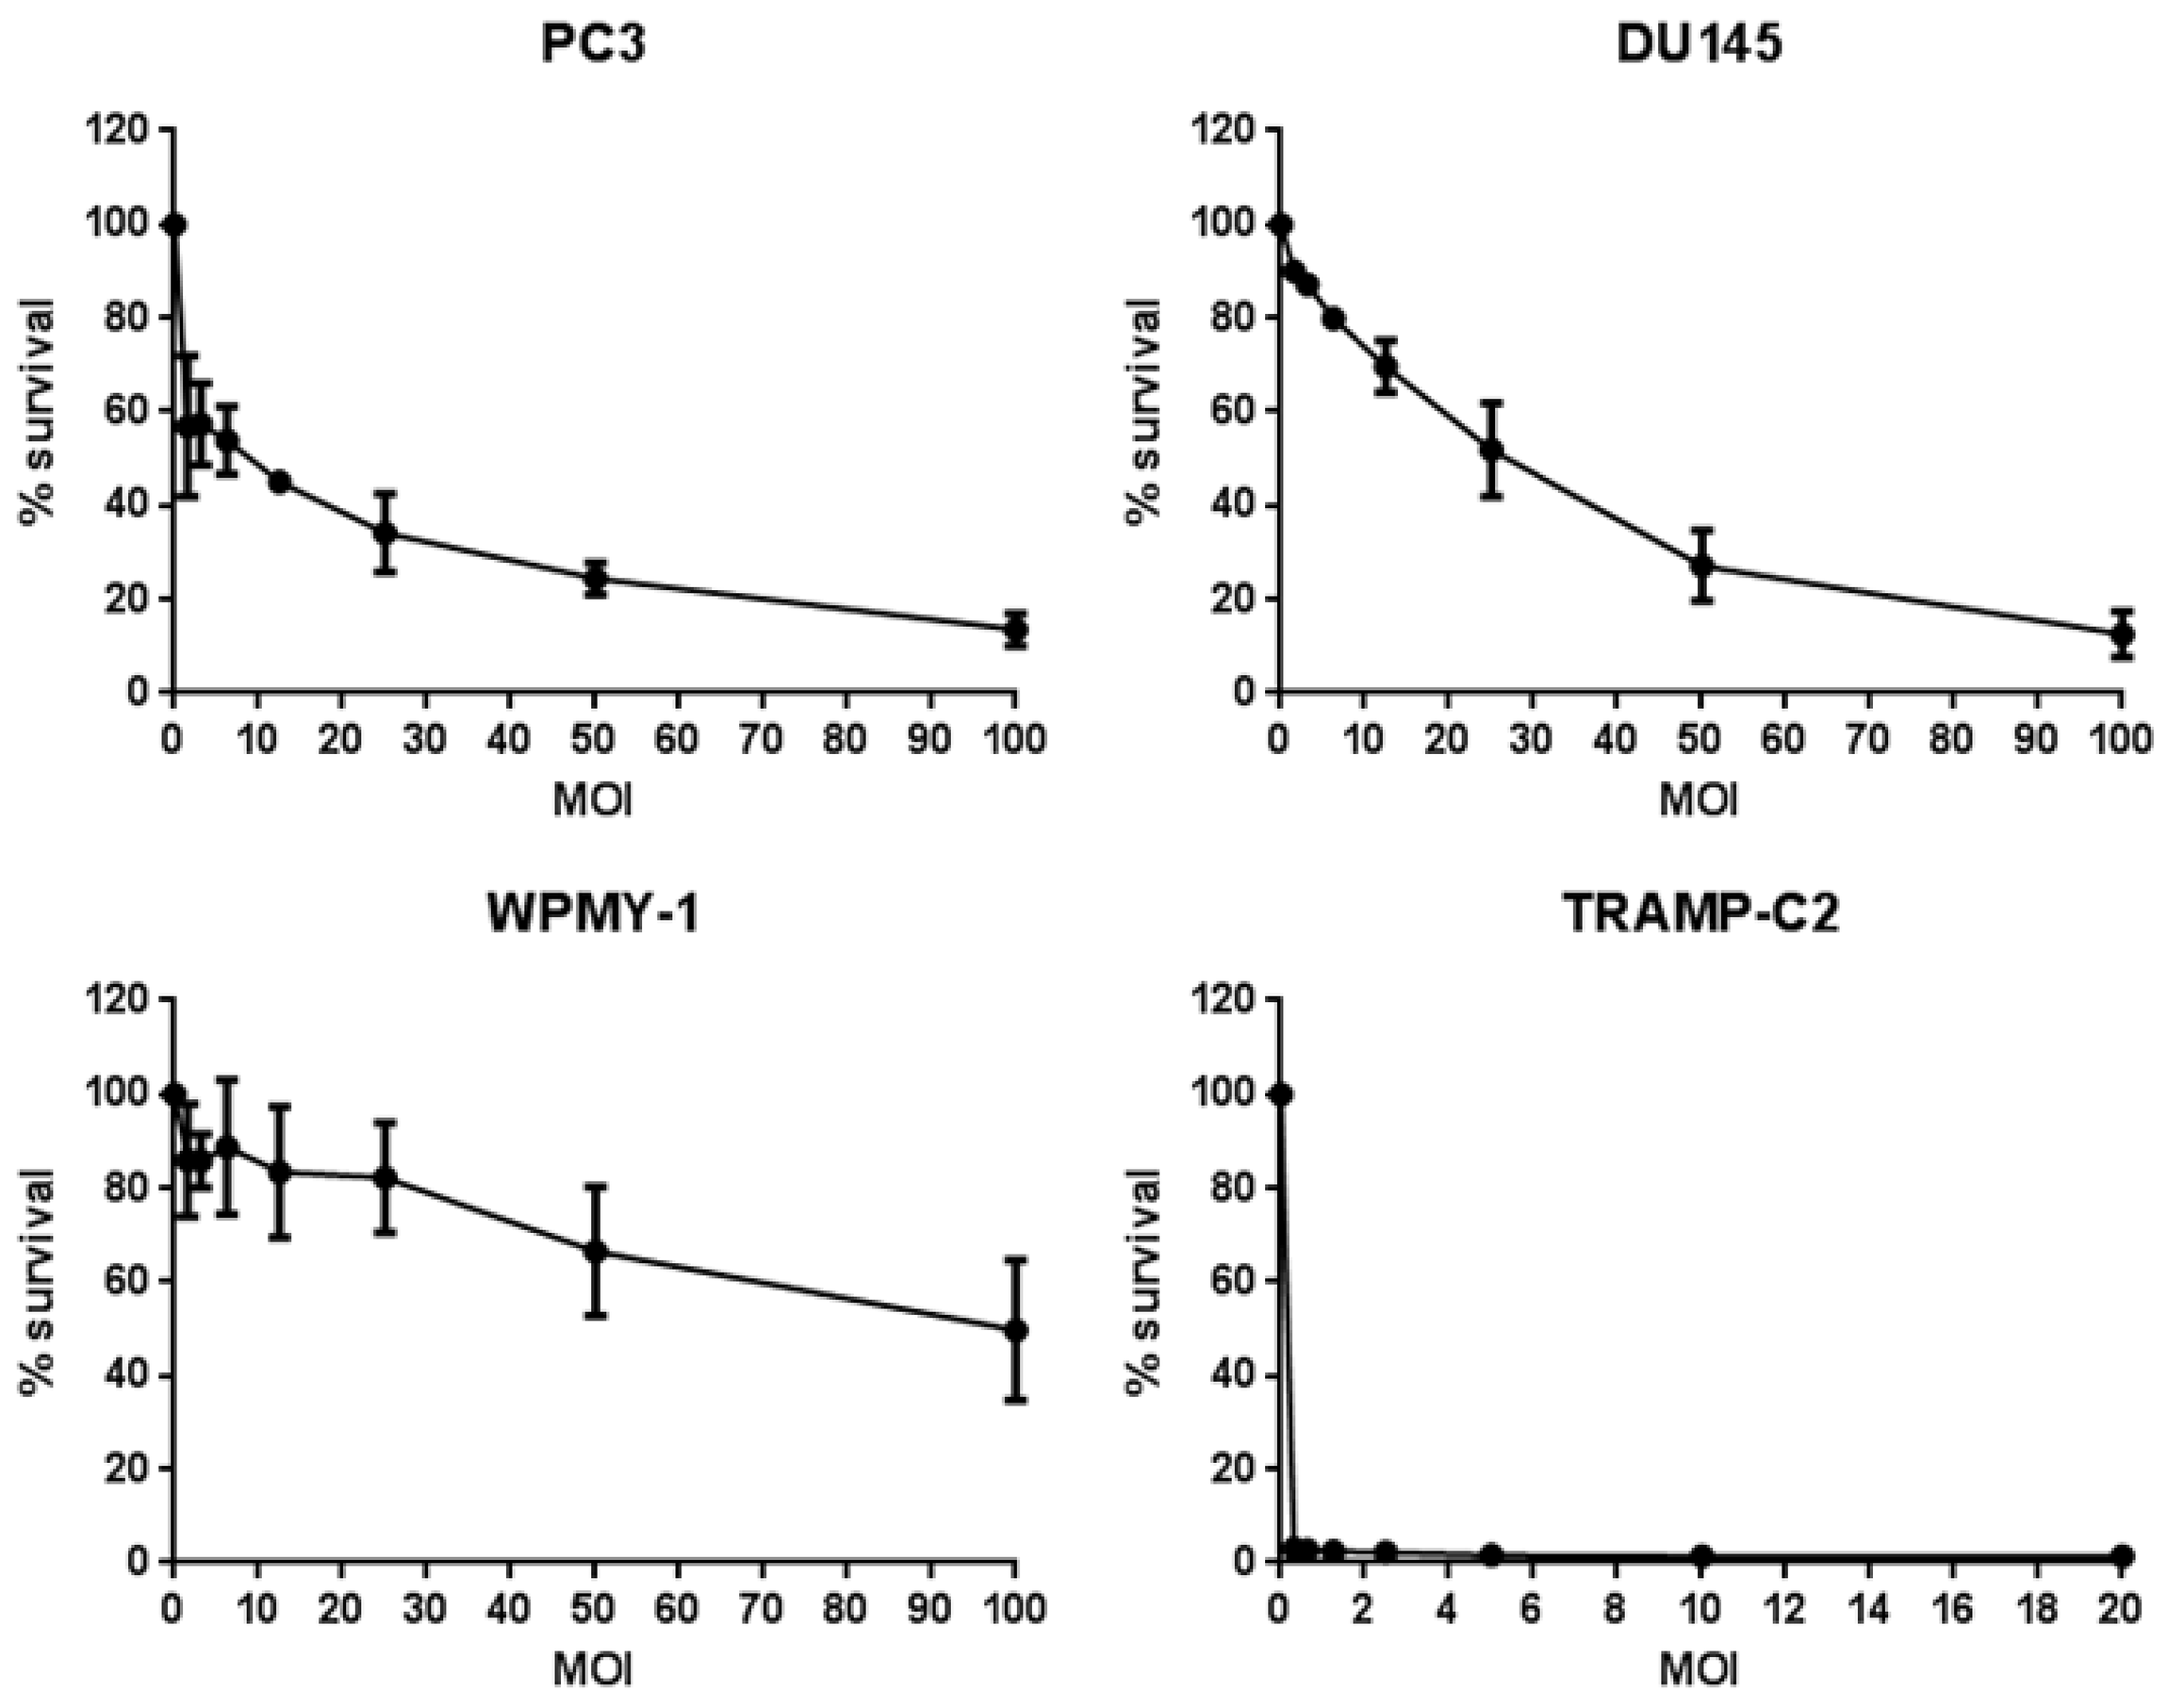
PC3
DU145
WPMY-1
TRAMP-C2
% survival
MOI

Figure 2: Induction of immunogenic cell death markers in response to reovirus infection in PCa cell lines.

The human PCa cell lines PC3 and DU145 and the mouse transgenic adenocarcinoma prostate cell line TRAMP-C2 were treated with reovirus at an MOI of 3 for PC3, 40 for DU145 and 0.06 for TRAMP-C2. (A) Cells were harvested at 16, 24, 48 and 72hour time-points and flow cytometry was performed. The mean fluorescent intensity (MFI) of calreticulin positive cells was gated on viable cells (ViViD negative cells) thus detecting surface exposed calreticulin rather than total calreticulin. (B) Supernatants were harvested at 16, 24, 48 and 72hour time-points. Reovirus triggered extracellular HMGB1 accumulation was determined by ELISA analysis of supernatants (significant differences between untreated or inactivated virus and reovirus-infected cultures as determined by two-way ANOVA; ** p<0.01,*** p<0.001, ****p<0.0001). Results are from two independent experiments (mean ± SD).

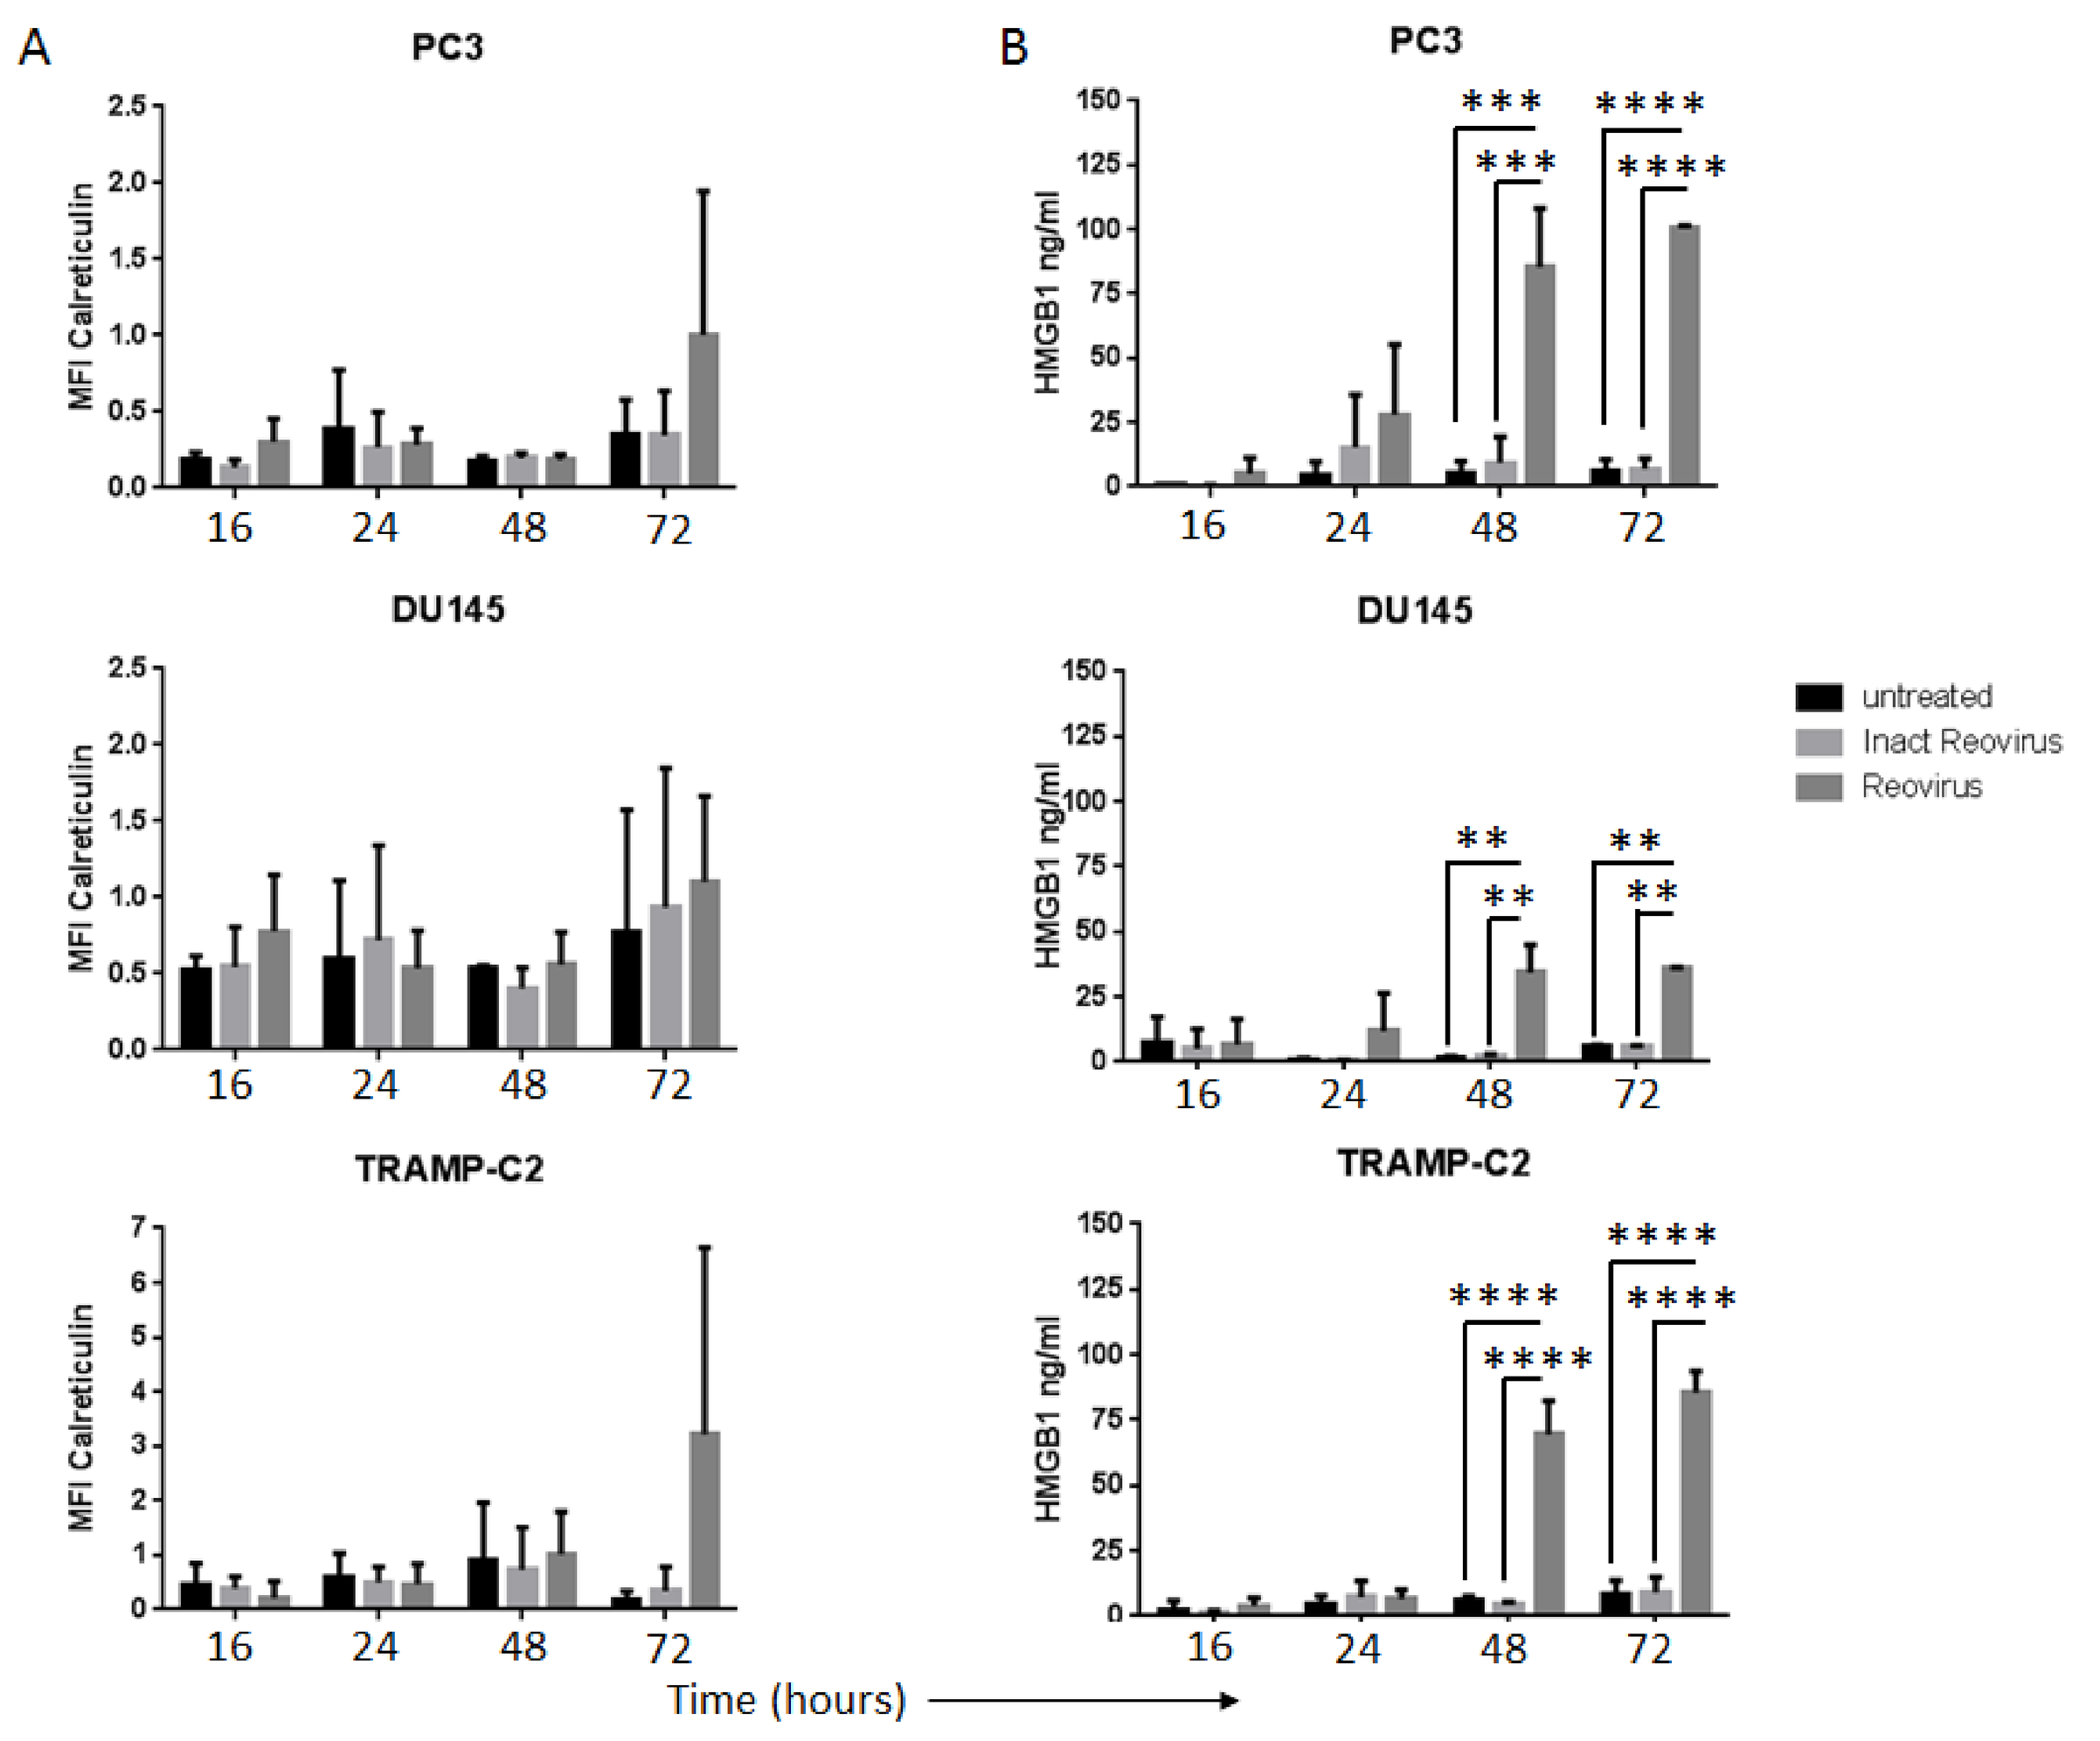
A
B
PC3
DU145
TRAMP-C2
MFI Calreticulin
HMGB1 ng/ml
untreated
Inact Reovirus
Reovirus
Time (hours)

Figure 3: Reovirus infection of PCa cell lines up-regulates cell surface molecules associated with susceptibility to immune attack

The human PCa cell lines PC3 and DU145 and the mouse transgenic adenocarcinoma prostate cell line TRAMP-C2 were treated with reovirus at an MOI of 3 for PC3, 40 for DU145 and 0.06 for TRAMP-C2. Cells were harvested at 16, 24, 48 and 72hour time-points and HLA/H-2, CD80 and Fas expression were assessed by flow cytometry. Results are from two independent experiments (mean ± SD).

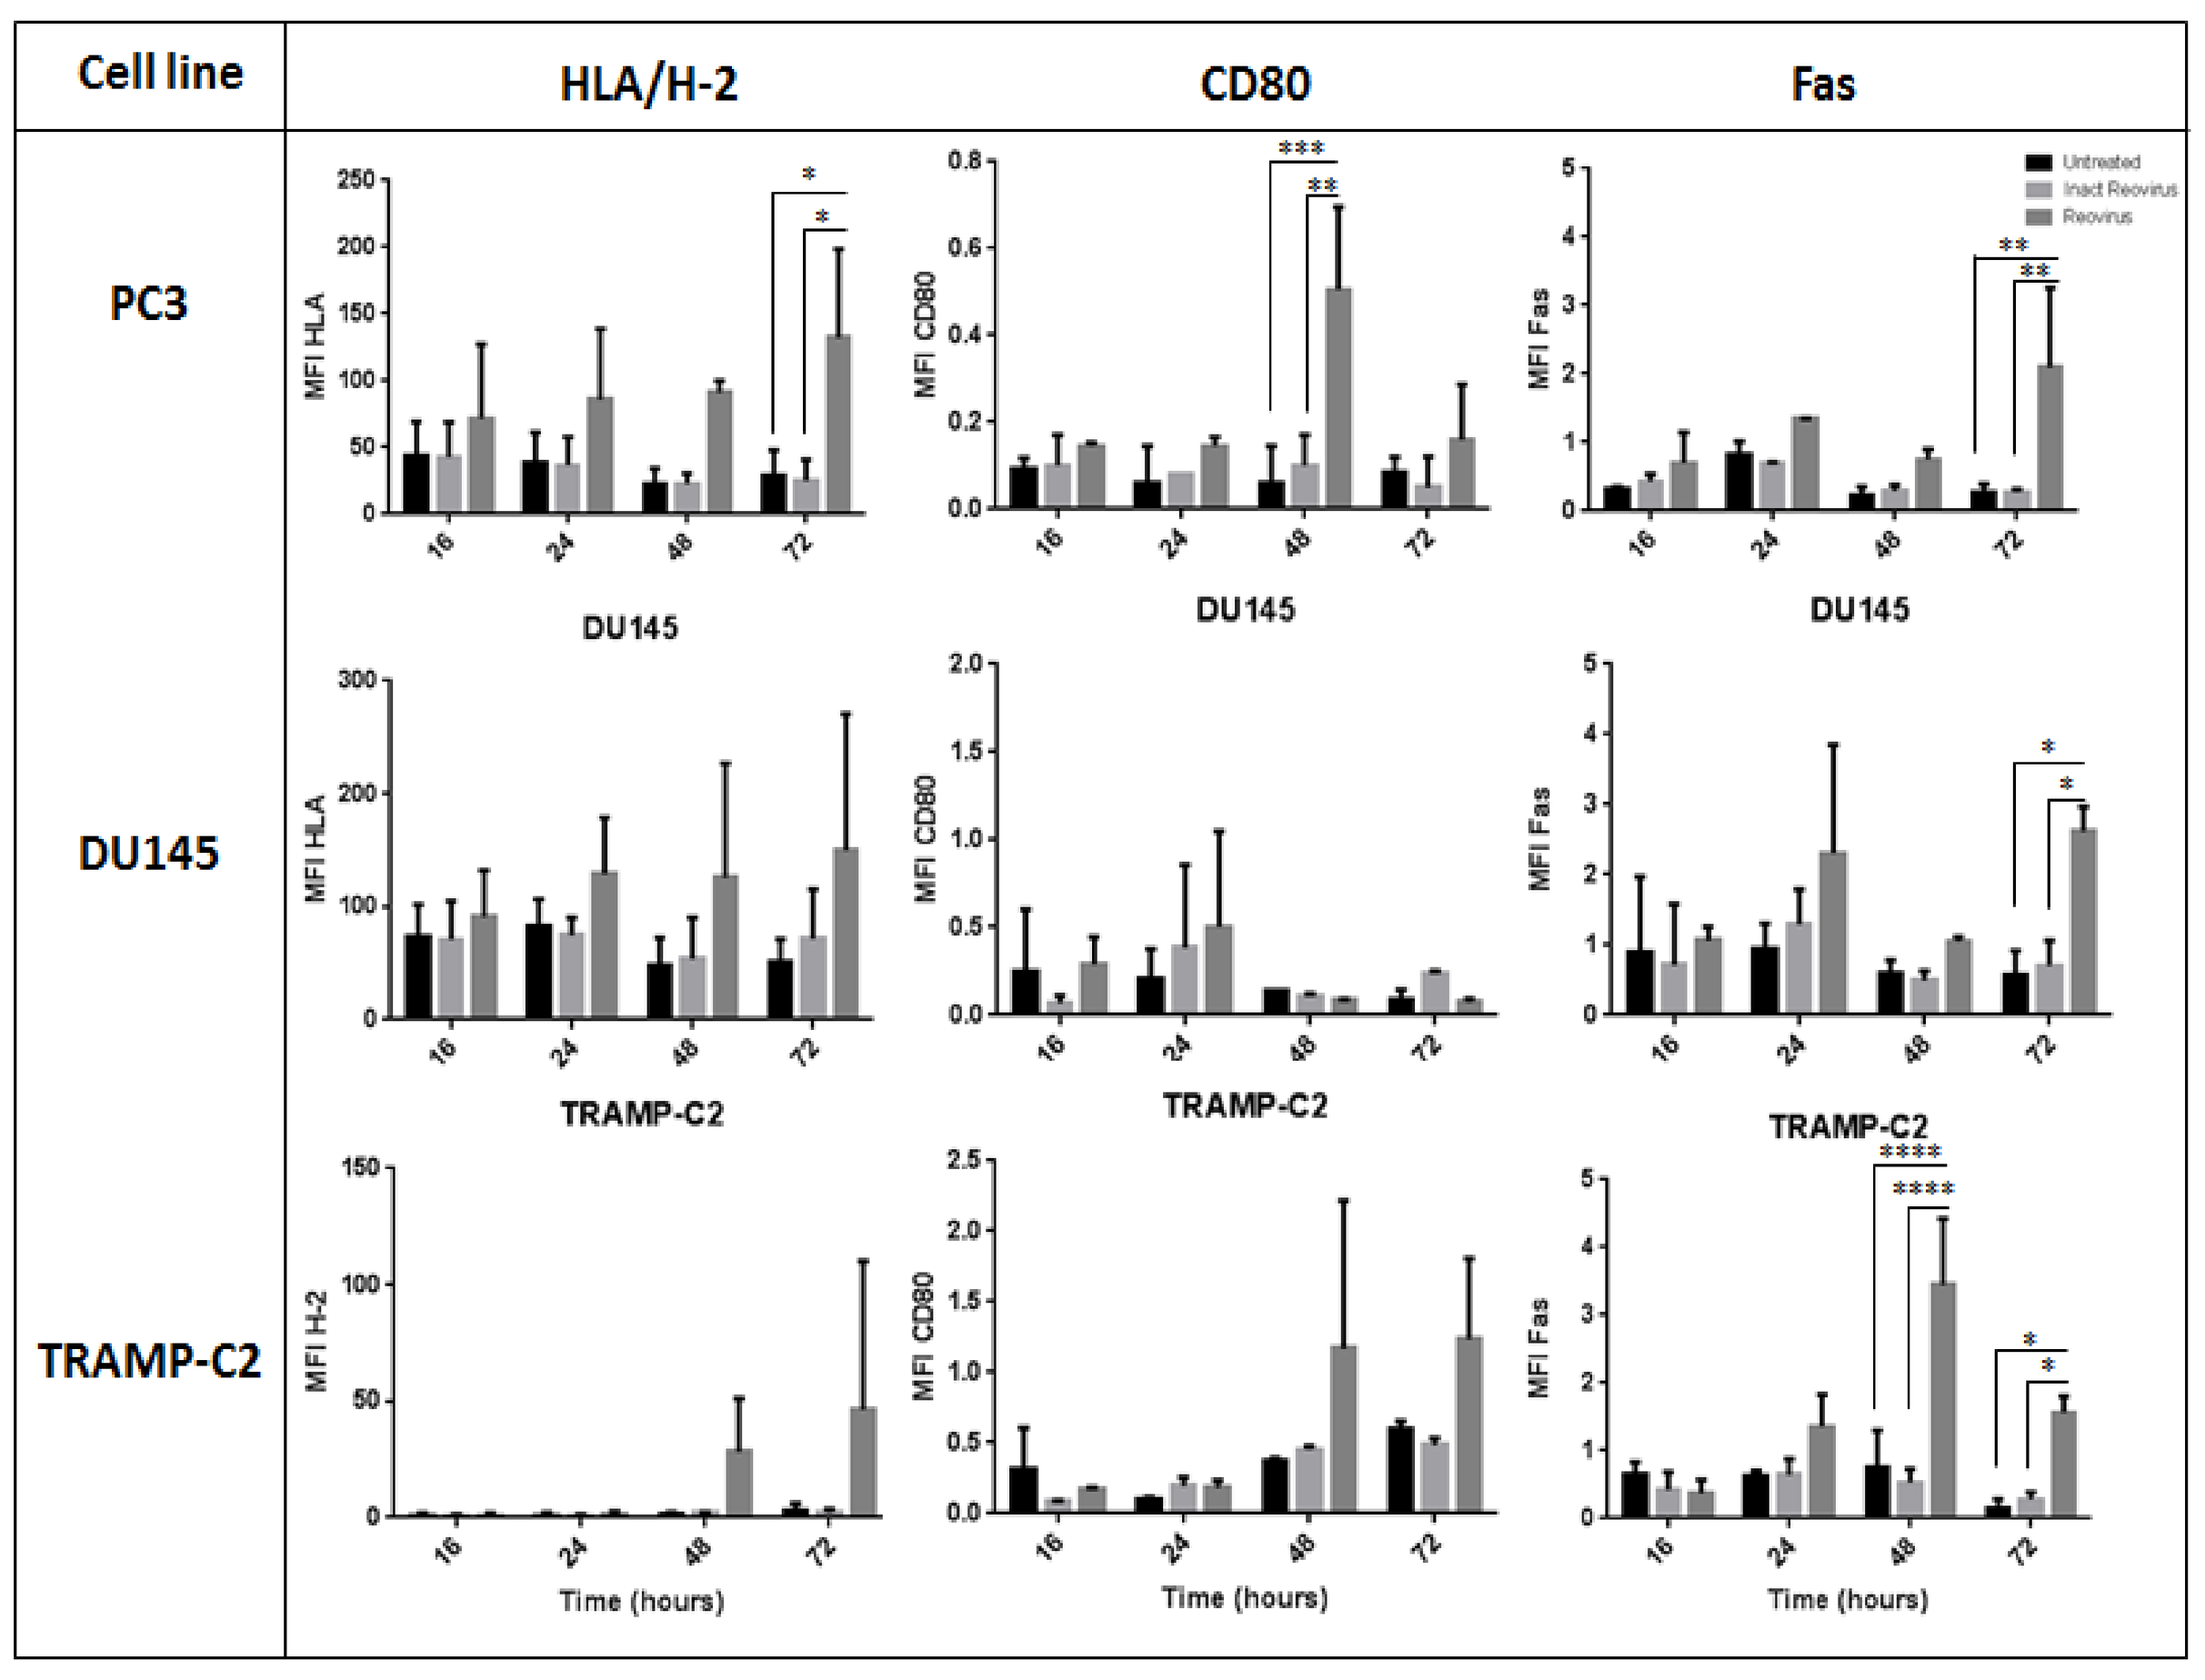
Cell line
HLA/H-2
CD80
Fas
PC3
DU145
TRAMP-C2
MFI HLA
MFI H-2
MFI CD80
MFI Fas
Untreated
Inact Reovirus
Reovirus
Time (hours)

This study was funded by Prostate Cancer UK

Fig 4: Reovirus infection of tumours is needed before a therapeutic effect of immune checkpoint inhibition is seen.

Immune checkpoint blockade (ICB) in the TRAMP-C2 PCa murine model was tested with each agent as a monotherapy and then each ICB antibody or combination of together with reovirus infection. Control mice received 100ug isotype control antibody. Tumour diameters were measured twice weekly.

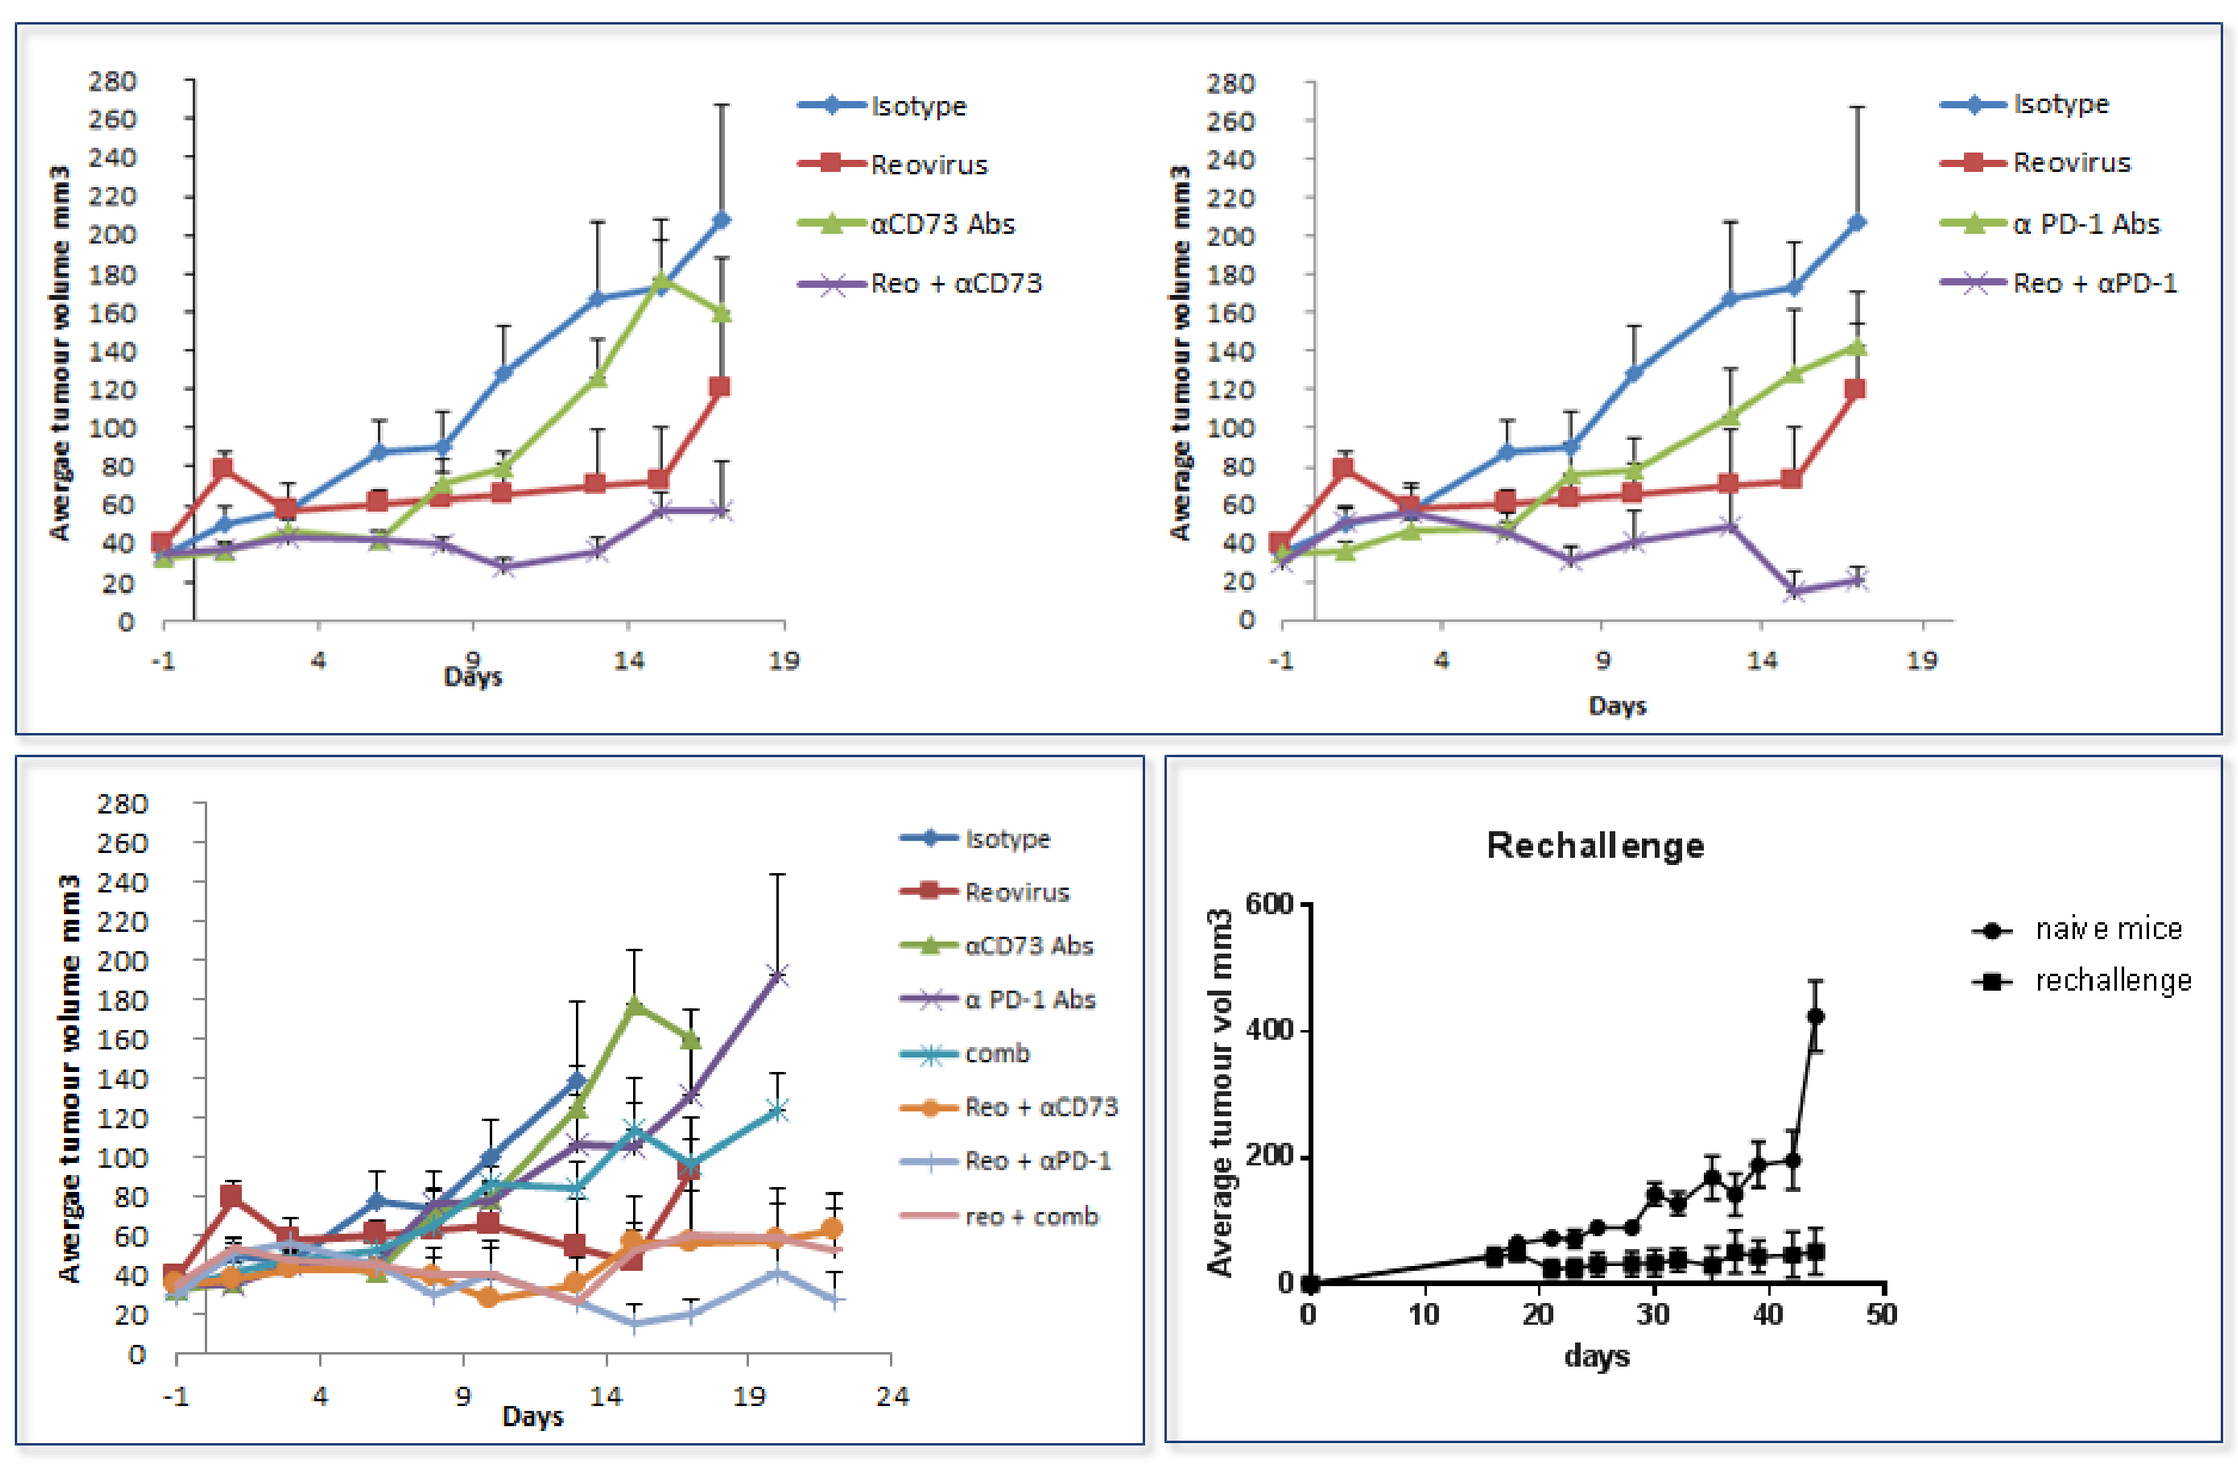
Isotype
Reovirus
αCD73 Abs
Reo + αCD73
α PD-1 Abs
Reo + αPD-1
comb
reo + comb
Avergae tumour volume mm3
Days
Rechallenge
naive mice
rechallenge
Average tumour vol mm3
days

Fig 5. Reovirus infection of tumours induces significant expression of the stimulatory dendritic cell chemoattractants XCL1, CCL5 and FLT3L

Nanostring's Pan Cancer Immune Profiling RNA Panel was used to investigate the differential gene expression of total RNA from untreated or reovirus-treated TRAMP-C2 tumours.
(A) reovirus infection caused an increase in innate (NK cells and DCs) and adaptive immune cell types (T cells and B cells) within the virus-treated TRAMP-C2 tumours as compared to untreated tumours. (B) Of the panel of negative regulators only BTLA and PD-L1 were significantly upregulated in the reovirus treated TRAMP-C2 tumours compared to untreated tumours (C) Significant upregulation of genes encoding XCL5, CCL5 and FLT3LG, known chemoattractants for stimulatory DCs, were observed from reovirus-treated tumours compared to untreated tumours (significant differences between untreated or reovirus-infected cultures as determined by unpaired T-test; * p<0.05, ** p<0.01,*** p<0.001).

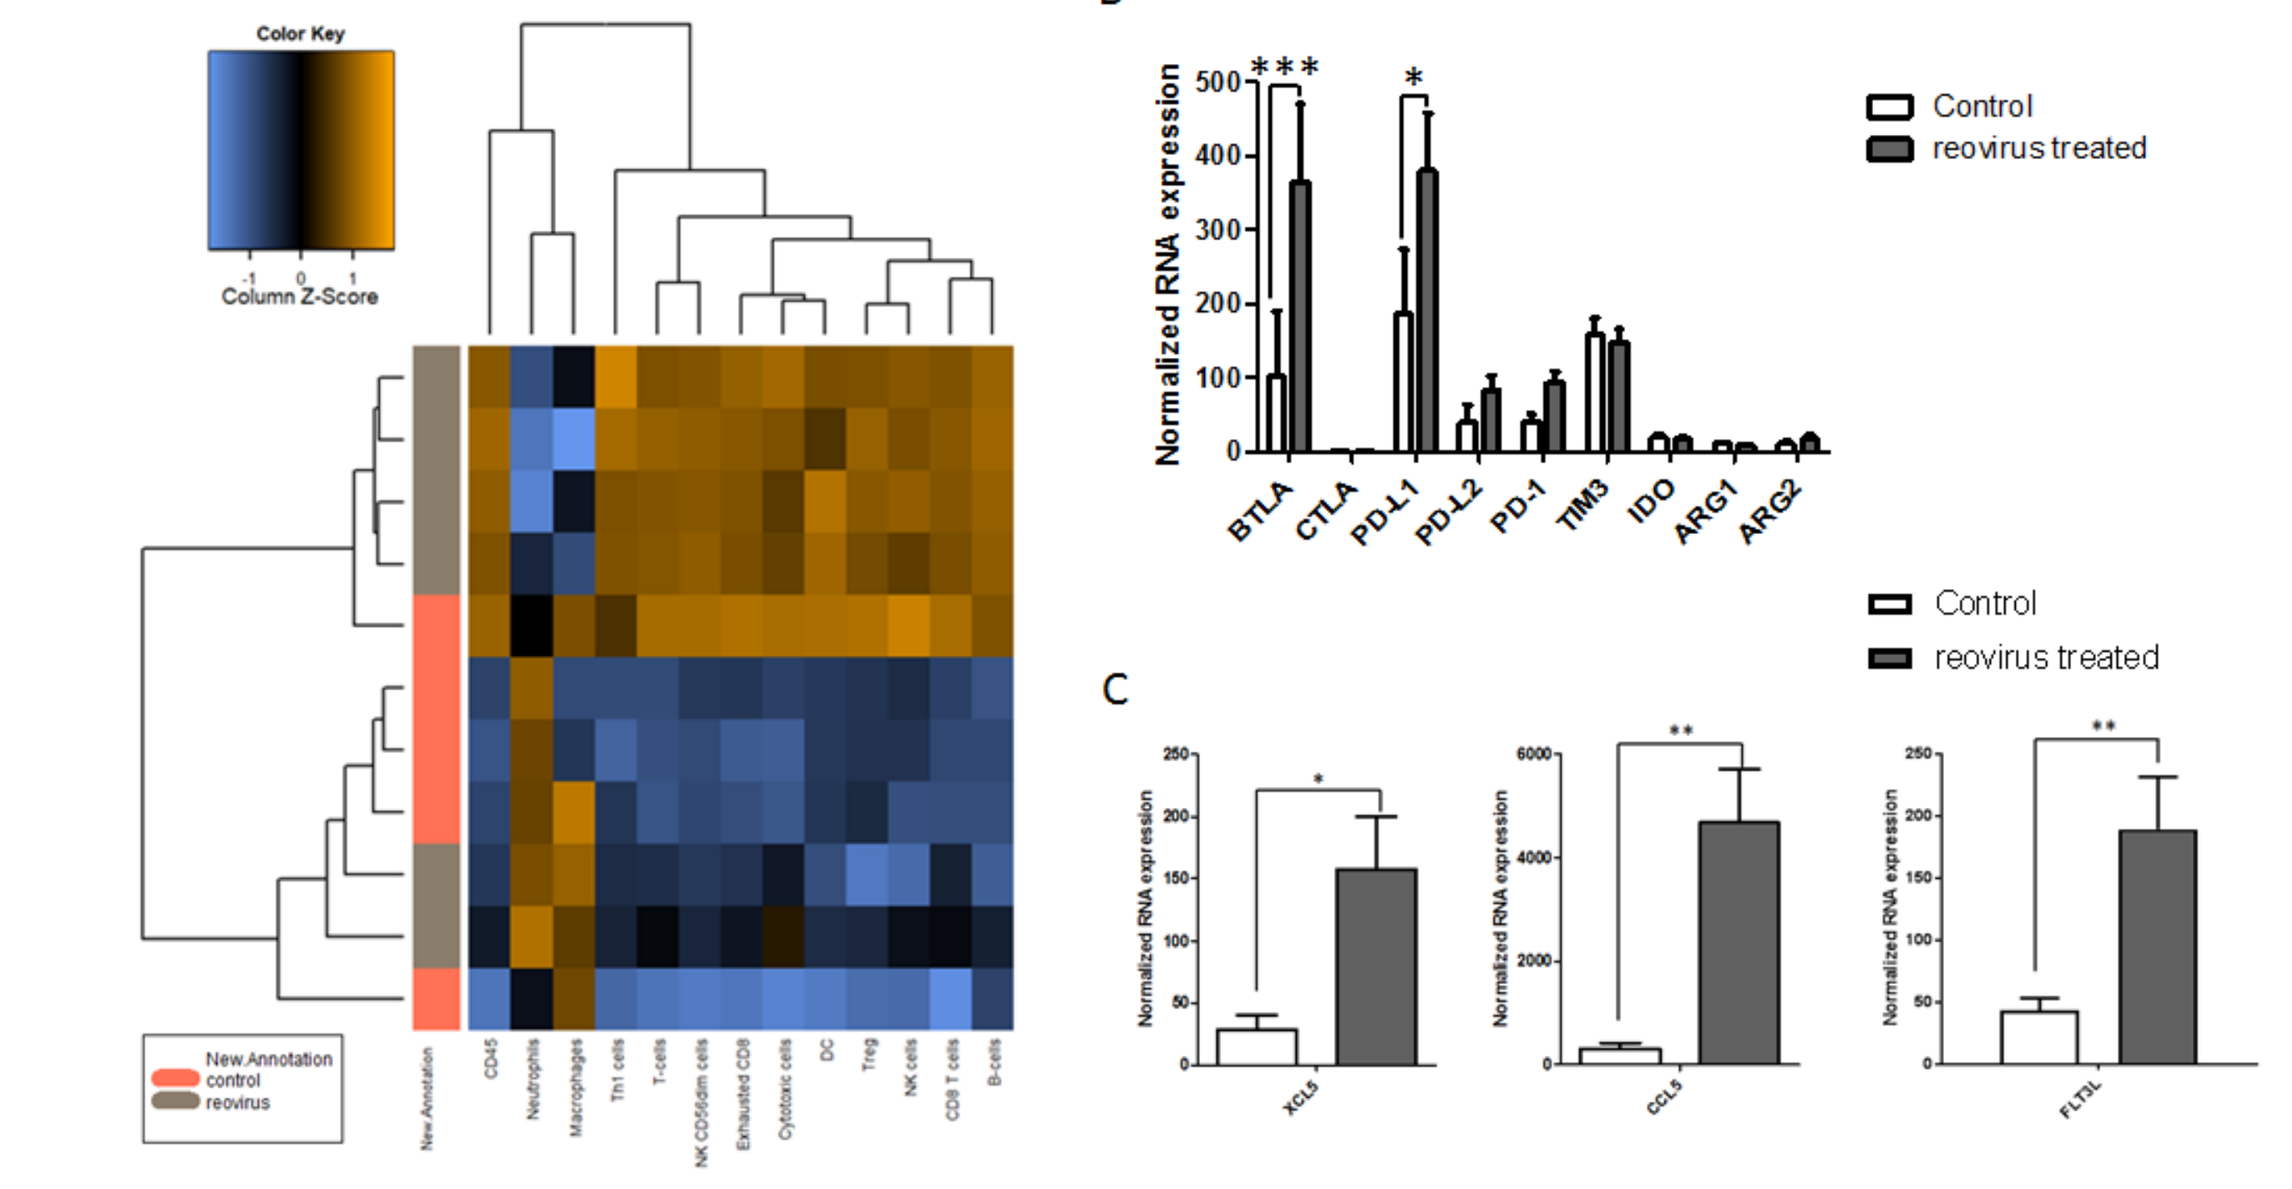
A
B
C
Color Key
Column Z-Score
Normalized RNA expression
Control
reovirus treated
BTLA
CTLA
PD-L1
PD-L2
PD-1
TIM3
IDO
ARG1
ARG2

Summary:

This study has clearly demonstrated that oncolytic virotherapy is able to transform and 'heat up' an immunologically 'cold' prostatic tumour microenvironment, and thereby sensitise it to immune checkpoint blockade. The proinflammatory effects of viral oncolysis may stem from its attraction and activation of NK cells which through the production of chemokines and FLT3LG in the tumour, control the levels of stimulatory DCs and thus priming of effector T cells, increasing the responsiveness of prostate tumours to anti-PD-1 immunotherapy. This combination strategy is feasible for patient treatment.